Abstract
7515

Outcomes in ZUMA-5 With Axicabtagene Ciloleucel (Axi-Cel) in Patients With Relapsed/Refractory Indolent Non-Hodgkin Lymphoma Who Had the High-Risk Feature of Progression Within 24 Months From Initiation of First Anti-CD20–Containing Chemoimmunotherapy (POD24)

Caron A. Jacobson, MD[1]; Julio C. Chavez, MD[2]; Alison R. Sehgal, MD[3]; Basem M. William, MD[4]; Javier Munoz, MD, MS, FACP[5]; Gilles Salles, MD, PhD[6]; Carla Casulo, MD[7]; Pashna N. Munshi, MD[8]; David G. Maloney, MD, PhD[9]; Sven de Vos, MD, PhD[10]; Ran Reshef, MD[11]; Lori A. Leslie, MD[12]; Ibrahim Yakoub-Agha, MD, PhD[13]; Olalekan O. Oluwole, MD, MPH, MBBS[14]; Henry Chi Hang Fung, MD, FACP, FRCPE[15]; Vicki Plaks, LLB, PhD[16]; Yin Yang, MS[16]; Jennifer Lee[16]; Mauro P. Avanzi, MD, PhD[16]; Sattva S. Neelapu, MD[17]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]University of South Florida H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL, USA; [3]UPMC Hillman Cancer Center, Pittsburgh, PA, USA; [4]The Ohio State University Comprehensive Cancer Center, Columbus, OH, USA; [5]Banner MD Anderson Cancer Center, Gilbert, AZ, USA; [6]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [7]University of Rochester Medical Center - James P. Wilmot Cancer Center, Rochester, NY, USA; [8]Georgetown Lombardi Comprehensive Cancer Center, Washington, DC, USA; [9]Fred Hutchinson Cancer Research Center, Seattle, WA, USA; [10]Ronald Reagan University of California Los Angeles Medical Center, Santa Monica, CA, USA; [11]Columbia University Herbert Irving Comprehensive Cancer Center, New York, NY, USA; [12]John Theurer Cancer Center, Hackensack, NJ, USA; [13]CHU de Lille, univ Lille, INSERM U1286, Infinite, 59000 Lille, France; [14]Vanderbilt University Medical Center, Nashville, TN, USA; [15]Fox Chase Cancer Center, Philadelphia, PA, USA; [16]Kite, a Gilead Company, Santa Monica, CA, USA; [17]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

BACKGROUND

- Progression within 24 months from initiating the first anti-CD20–containing chemoimmunotherapy (POD24) is a risk factor for poor survival in patients with indolent non-Hodgkin lymphoma (iNHL)[1,2]
 - Approximately 20% of patients with follicular lymphoma (FL) have POD24[1]
 - In an observational analysis from the National LymphoCare Study, patients with FL who progressed early had a lower 5-year overall survival (OS) rate (50%) than those without early progression (90%)[2]
- Axicabtagene ciloleucel (axi-cel) is an autologous anti-CD19 chimeric antigen receptor (CAR) T-cell therapy (**Figure 1**) approved in the United States (US) for the treatment of adults with relapsed/refractory (R/R) FL after ≥2 lines of systemic therapy, and in the US and European Union for adults with R/R large B-cell lymphoma (LBCL) after ≥2 lines of systemic therapy[3,4]
 - In a long-term follow-up analysis of axi-cel in refractory LBCL, the 4-year OS rate was 44%[5]
- ZUMA-5 is a multicenter, single-arm Phase 2 study of axi-cel in patients with R/R iNHL, including FL and marginal zone lymphoma (MZL)[6]
 - In the primary analysis, overall response rates (ORR) after a 17.5-month median follow-up were similarly high among patients with and without POD24 (93% vs 92%)

Figure 1. Structure of Axi-Cel

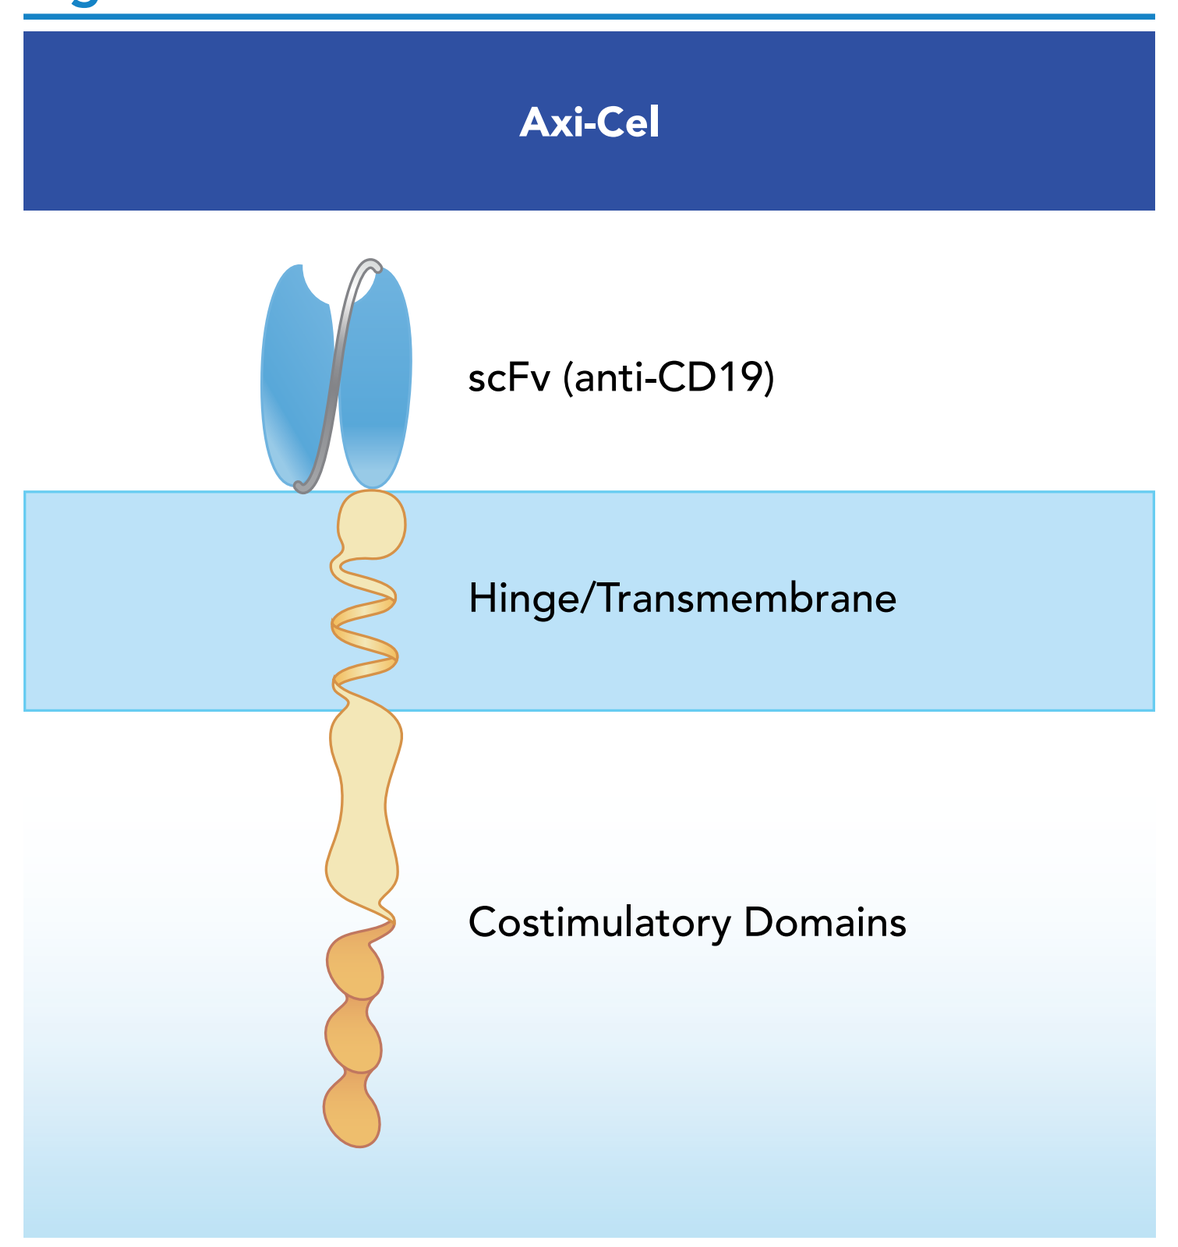

Axi-Cel
scFv (anti-CD19)
Hinge/Transmembrane
Costimulatory Domains

Axi-cel, axicabtagene ciloleucel; scFv, single chain variable fragment

OBJECTIVE

- To report safety and efficacy outcomes and pharmacokinetic/pharmacodynamic profiles with longer follow-up among patients in ZUMA-5 with and without POD24

METHODS

Figure 2. ZUMA-5 Study Design

R/R iNHL (N=148) → Leukapheresis → Conditioning Chemotherapy: Fludarabine 30 mg/m² IV, cyclophosphamide 500 mg/m² IV on Days −5, −4, −3 → Axi-Cel Infusion: 2×10⁶ CAR+ cells/kg on Day 0 → Post-treatment Assessment and Long-term Follow-Up Periods

Key ZUMA-5 Eligibility Criteria

- R/R FL (Grades 1–3a) or MZL (nodal or extranodal)[a]
- ≥2 Prior lines of therapy that must have included an anti-CD20 mAb combined with an alkylating agent[b]

Patients and Analysis

- The updated efficacy analysis occurred when ≥80 treated patients with FL had ≥18 months of follow-up[c]
- Data cutoff date: September 14, 2020
- Axi-cel–treated patients with FL or MZL and available data on progression after an anti-CD20 mAb + alkylating agent were included in the POD24 analysis (N=129)

[a] Patients with stable disease (without relapse) >1 year from completion of last therapy were not eligible. [b] Single-agent anti-CD20 antibody did not count as line of therapy for eligibility. [c] Efficacy-evaluable patients included ≥80 treated patients with FL who had ≥18 months of follow-up after axi-cel infusion and treated patients with MZL who had ≥4 weeks of follow-up after axi-cel infusion as of the data cutoff date.
Axi-cel, axicabtagene ciloleucel; CAR, chimeric antigen receptor; FL, follicular lymphoma; iNHL, indolent non-Hodgkin lymphoma; IV, intravenous; mAb, monoclonal antibody; MZL, marginal zone lymphoma; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; R/R, relapsed/refractory.

RESULTS

Table 1. Baseline Disease Characteristics

Characteristic	With POD24 (n=81)	Without POD24 (n=48)
Disease type, n (%)		
FL	68 (84)	40 (83)
MZL	13 (16)	8 (17)
Median age (range), years	60 (34–78)	62 (42–79)
≥65 years, n (%)	26 (32)	18 (38)
Male, n (%)	42 (52)	32 (67)
Stage III-IV disease, n (%)	67 (83)	45 (94)
≥3 FLIPI, n/n (%)	30/68 (44)	17/40 (43)
High tumor bulk (GELF criteria), n (%)[a]	41 (51)	21 (44)
Median no. of prior therapies (range)	3 (1–10)[b]	3.5 (2–8)
≥3, n (%)	49 (60)	36 (75)
Prior PI3Ki therapy, n (%)	22 (27)	17 (35)
Prior lenalidomide, n (%)	25 (31)	19 (40)
Prior autologous SCT, n (%)	16 (20)	11 (23)
Refractory disease, n (%)[c]	62 (77)	30 (63)

[a] Disease burden, as defined by GELF criteria: involvement of ≥3 nodal sites (≥3 cm diameter each); any nodal or extranodal tumor mass with ≥7 cm diameter; B symptoms; splenomegaly; pleural effusions or peritoneal ascites; cytopenias; or leukemia. [b] Enrollment of 3 patients with FL who had 1 prior line of therapy occurred in ZUMA-5 before a protocol amendment requiring ≥2 prior lines of therapy. [c] Patients with iNHL who progressed within 6 months of completion of the most recent prior treatment.
FL, follicular lymphoma; FLIPI, Follicular Lymphoma International Prognostic Index; GELF, Groupe d'Etude des Lymphomes Folliculaires; MZL, marginal zone lymphoma; PI3Ki, phosphoinositide 3-kinase inhibitor; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; SCT, stem cell transplantation.

- Baseline characteristics were generally similar among patients with and without POD24 (**Table 1**)
 - Among evaluable patients with FL, median tumor burden by sum of product diameters was numerically similar in those with and without POD24 (2303 mm² vs 2839 mm²)
 - Among evaluable patients with MZL, median SPD appeared higher among those with POD24 than without POD24 (2028 mm² vs 954 mm²)

RESULTS *(continued)*

Figure 3. ORR by IRRC Assessment in Patients With iNHL by POD24 Status

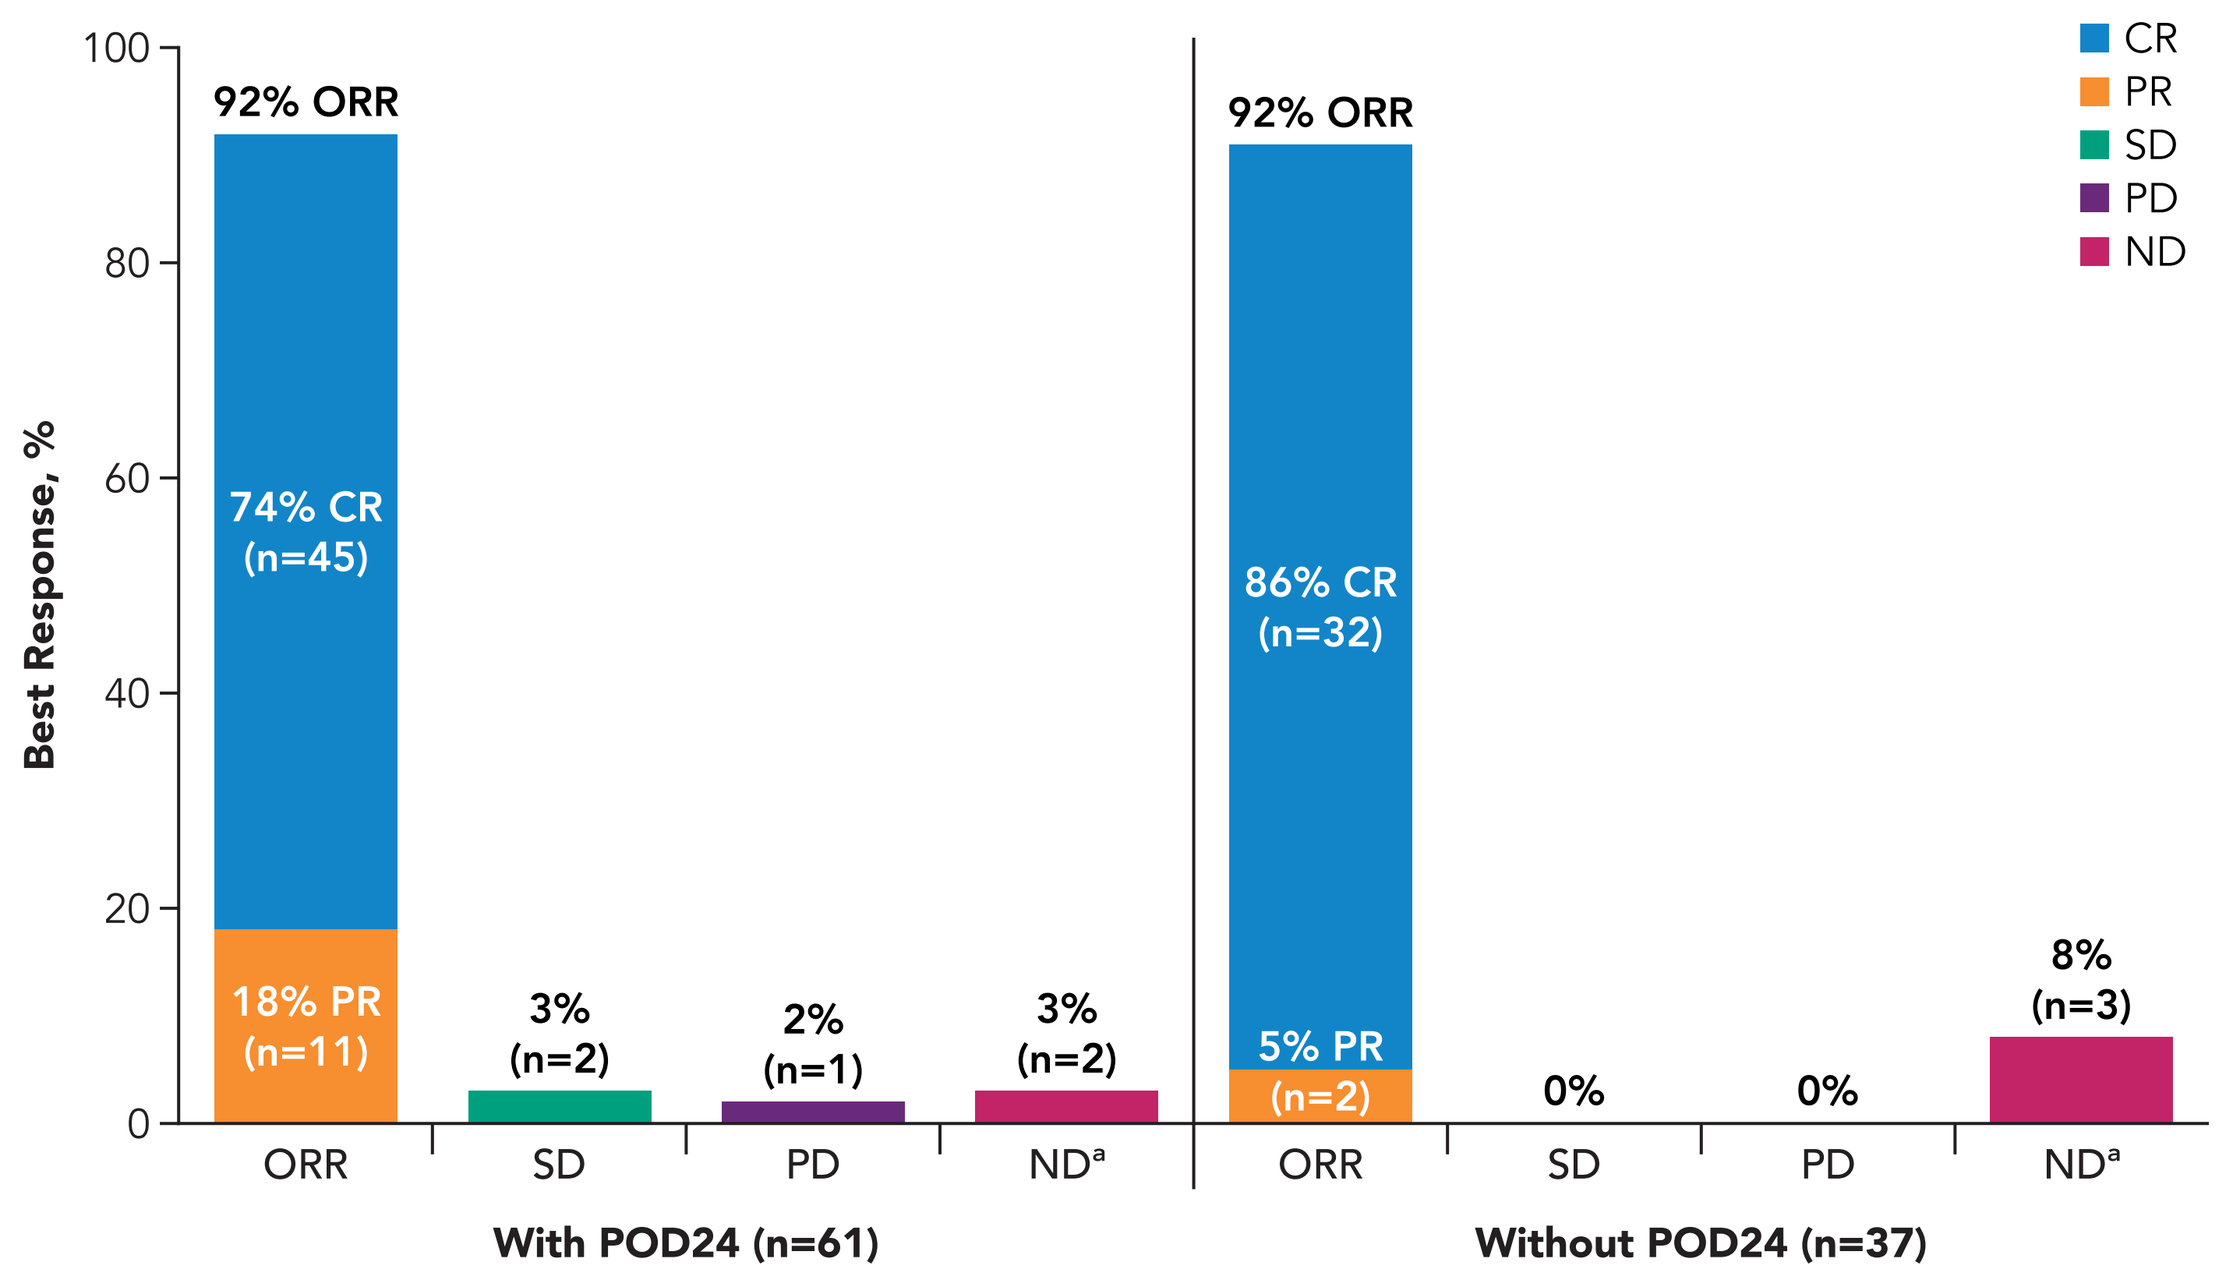

CR
PR
SD
PD
ND
Best Response, %
92% ORR
74% CR (n=45)
18% PR (n=11)
3% (n=2)
2% (n=1)
3% (n=2)
ORR
SD
PD
ND[a]
With POD24 (n=61)
92% ORR
86% CR (n=32)
5% PR (n=2)
0%
0%
8% (n=3)
Without POD24 (n=37)

Assessed by an IRRC according to the Lugano Classification.[7]
[a] Among the 5 patients reported as ND, 4 (1 FL without POD24; 3 MZL) had no disease at baseline and post-baseline per IRRC but were considered with disease by the investigator; 1 patient with FL and POD24 died before the first disease assessment.
CR, complete response; FL, follicular lymphoma; IRRC, Independent Radiology Review Committee; MZL, marginal zone lymphoma; ND, not done/undefined; ORR, overall response rate; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; PD, progressive disease; PR, partial response; SD, stable disease.

Figure 4. DOR, PFS, and OS in Patients With iNHL by POD24 Status

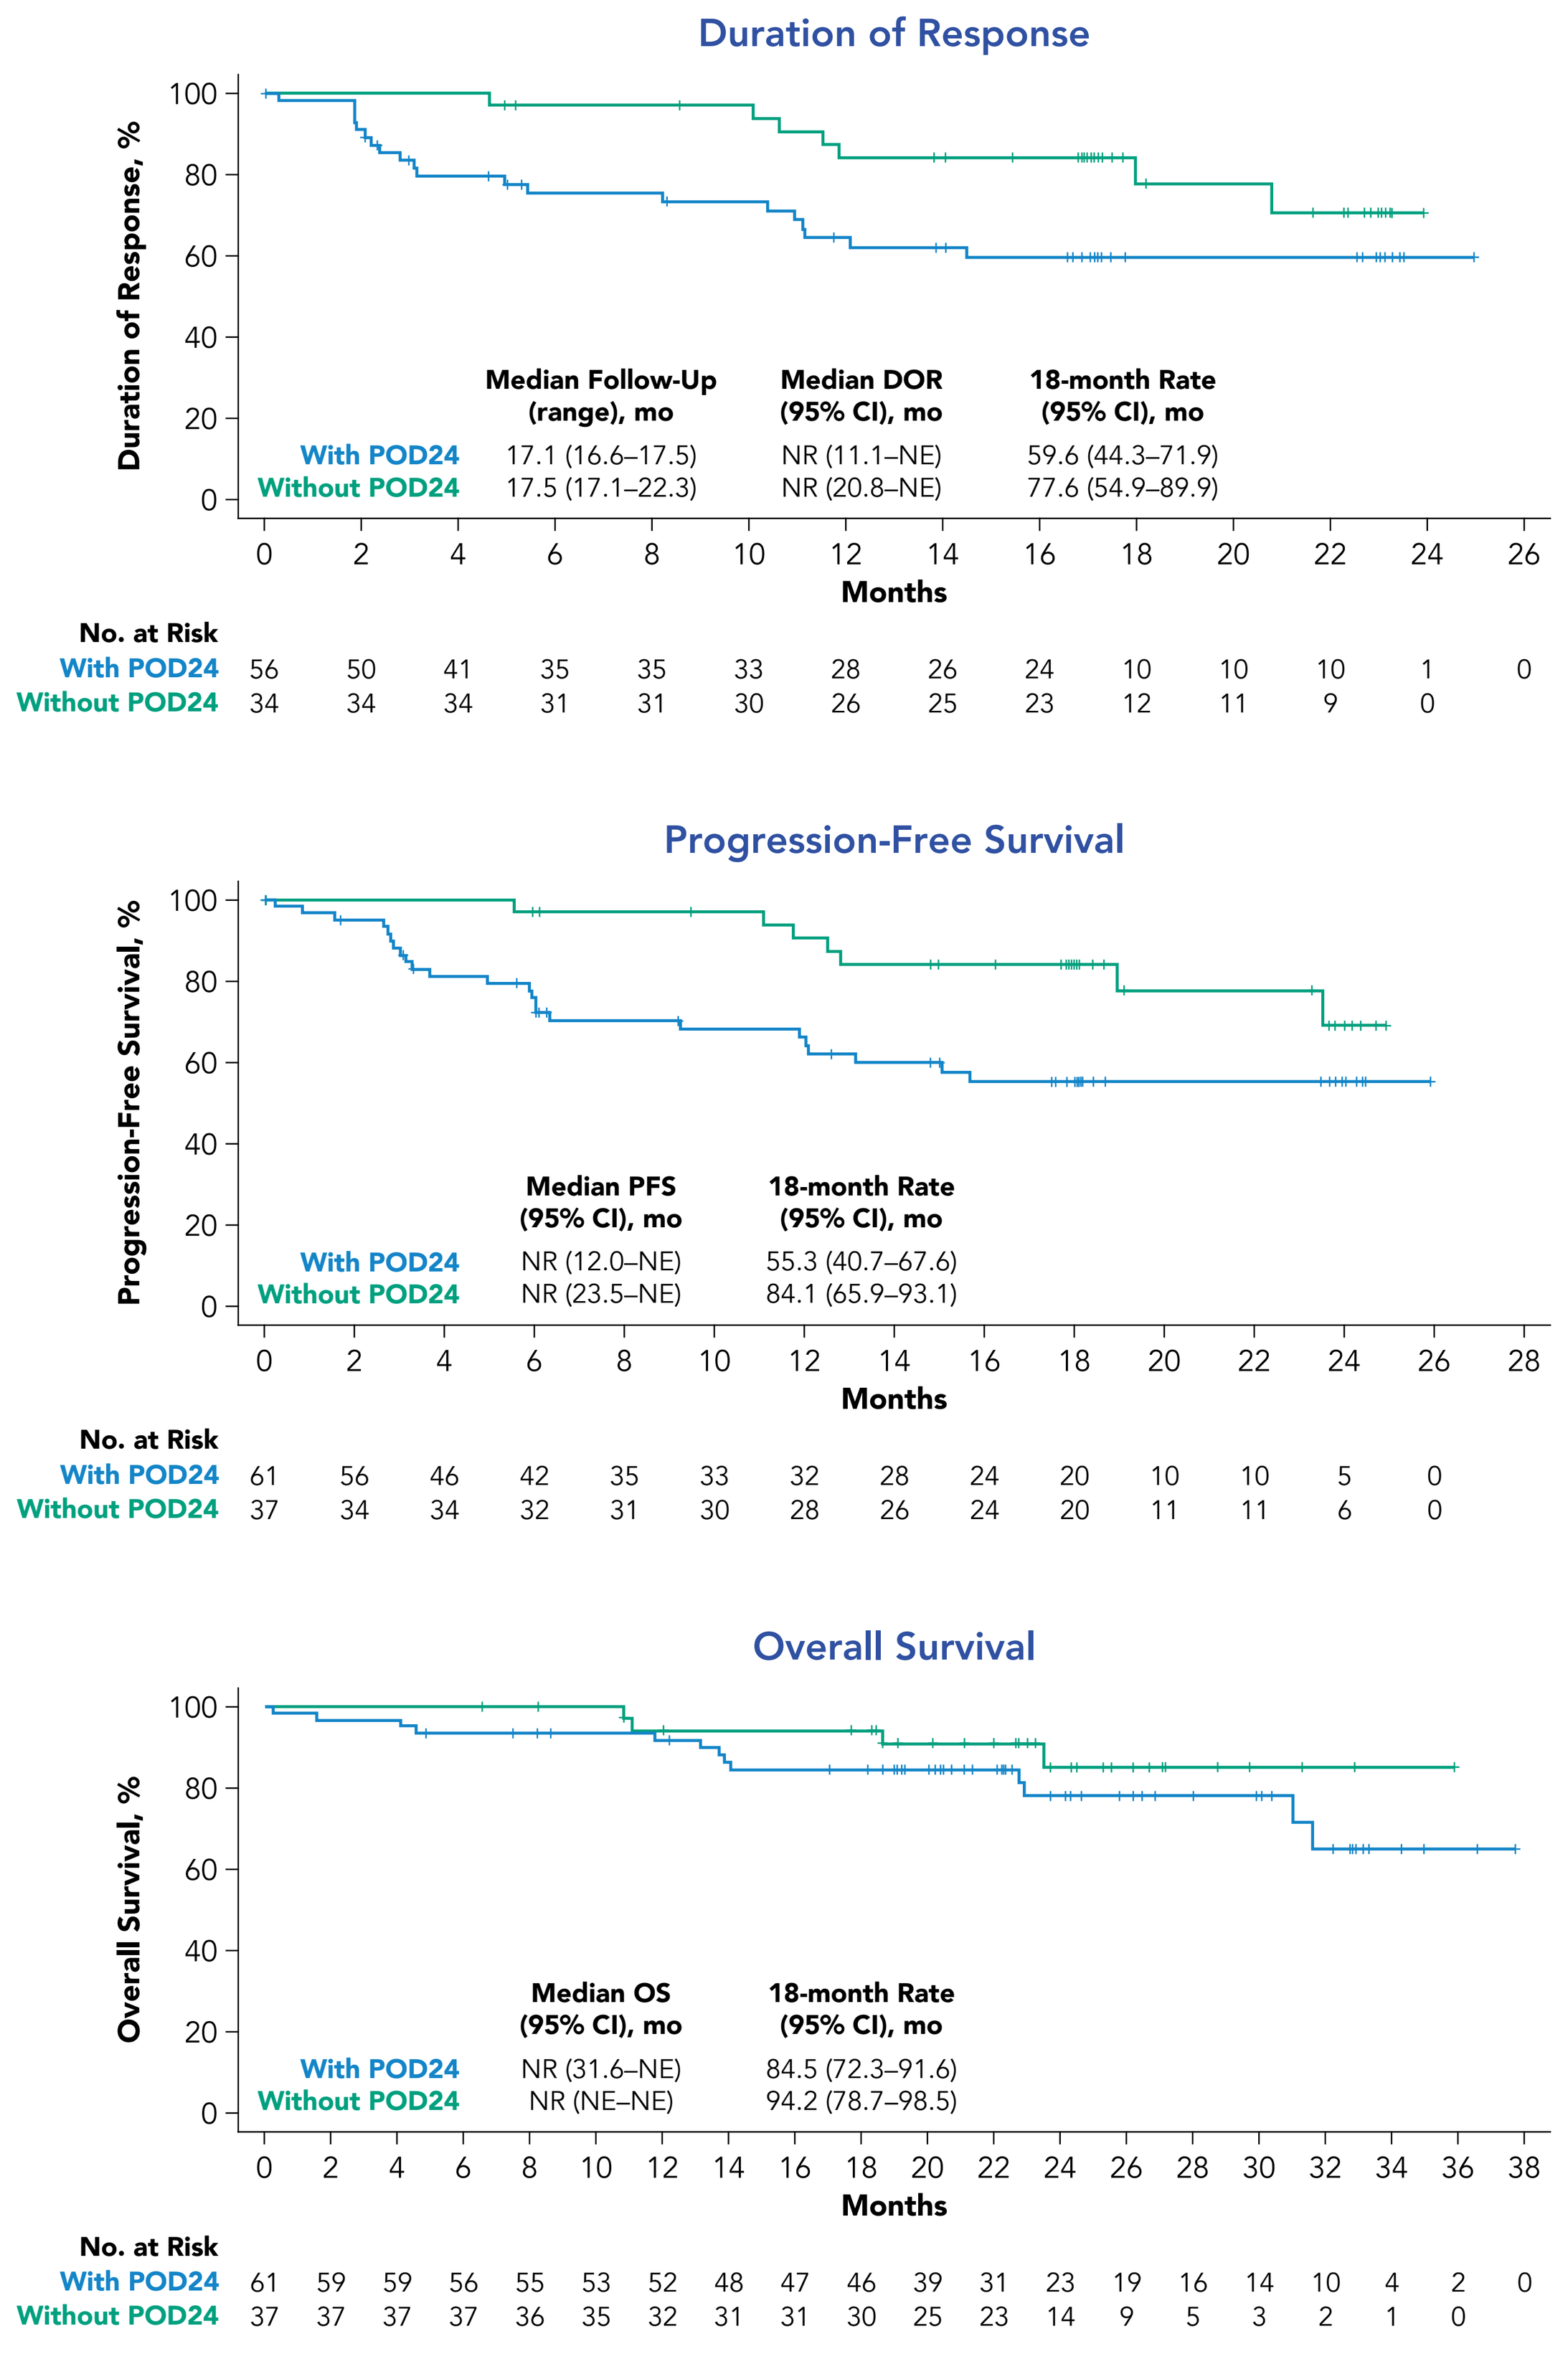

Duration of Response
Duration of Response, %
Median Follow-Up (range), mo | Median DOR (95% CI), mo | 18-month Rate (95% CI), mo
With POD24 17.1 (16.6–17.5) NR (11.1–NE) 59.6 (44.3–71.9)
Without POD24 17.5 (17.1–22.3) NR (20.8–NE) 77.6 (54.9–89.9)
Months
No. at Risk
With POD24 56 50 41 35 35 33 28 26 24 10 10 10 1 0
Without POD24 34 34 34 31 31 30 26 25 23 12 11 9 0
Progression-Free Survival
Progression-Free Survival, %
Median PFS (95% CI), mo | 18-month Rate (95% CI), mo
With POD24 NR (12.0–NE) 55.3 (40.7–67.6)
Without POD24 NR (23.5–NE) 84.1 (65.9–93.1)
Months
No. at Risk
With POD24 61 56 46 42 35 33 32 28 24 20 10 10 5 0
Without POD24 37 34 34 32 31 30 28 26 24 20 11 11 6 0
Overall Survival
Overall Survival, %
Median OS (95% CI), mo | 18-month Rate (95% CI), mo
With POD24 NR (31.6–NE) 84.5 (72.3–91.6)
Without POD24 NR (NE–NE) 94.2 (78.7–98.5)
Months
No. at Risk
With POD24 61 59 59 56 55 53 52 48 47 46 39 31 23 19 16 14 10 4 2 0
Without POD24 37 37 37 37 36 35 32 31 31 30 25 23 14 9 5 3 2 1 0

DOR, duration of response; FL, follicular lymphoma; mo, month; MZL, marginal zone lymphoma; NE, not estimable; NR, not reached; OS, overall survival; PFS, progression-free survival; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy.

Table 2. Efficacy Outcomes Among Patients With FL and MZL by POD24 Status

	Follicular Lymphoma		Marginal Zone Lymphoma	
Parameter	**With POD24 (n=49)**	**Without POD24 (n=29)**	**With POD24 (n=12)**	**Without POD24 (n=8)**
ORR, n (%)	46 (94)	28 (97)	10 (83)	6 (75)
CR	38 (78)	26 (90)	7 (58)	6 (75)
PR	8 (16)	2 (7)	3 (25)	0
Median DOR (95% CI), months	NR (14.5–NE)	NR (20.8–NE)	11.1 (1.9–NE)	NR (10.6–NE)
18-mo rate (95% CI), %	63.9 (47.2–76.6)	78.2 (53.3–90.8)	NR (NE–NE)	75.0 (12.8–96.1)
Median PFS (95% CI), months	NR (13.1–NE)	NR (23.5–NE)	9.2 (2.8–NE)	NR (11.8–NE)
18-mo rate (95% CI), %	59.8 (43.7–72.6)	85.3 (65.4–94.2)	30.7 (5.1–62.6)	75.0 (12.8–96.1)
Median OS (95% CI), months	NR (31.6–NE)	NR (NE–NE)	NR (13.7–NE)	NR (18.7–NE)
18-mo rate (95% CI), %	85.7 (72.4–92.9)	93.1 (75.1–98.2)	76.4 (30.9–94.0)	100.0 (NE–NE)

CR, complete response; DOR, duration of response; FL, follicular lymphoma; MZL, marginal zone lymphoma; ND, not done/undefined; NE, not estimable; NR, not reached; OS, overall survival; PFS, progression-free survival; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; PR, partial response.

- The ORR was similar among efficacy-evaluable patients with and without POD24 (**Figure 3**; **Table 2**)
- Estimated median duration of response (DOR) was not reached for patients with and without POD24, after a median follow-up of 17.1 months and 17.5 months, respectively (**Figure 4**; **Table 2**)
 - Responses were ongoing in 52% of efficacy-evaluable patients with POD24 and 70% of those without POD24 at data cutoff
 - The 18-month DOR rates in patients with and without POD24 were 60% and 78%, respectively
- Median progression-free survival (PFS) and median OS were not reached in patients with and without POD24 (**Figure 4**; **Table 2**)
 - The 18-month PFS rates in patients with and without POD24 were 55% and 84%, respectively
 - The 18-month OS rates were 85% and 94%, respectively
- Incidences of Grade ≥3 adverse events were generally similar in patients with and without POD24 (**Table 3**)
 - Grade 5 events occurred in 3 patients with POD24, including 1 event in the context of cytokine release syndrome (CRS); no Grade 5 events occurred in patients without POD24
 - Grade 4 CRS occurred in 1 patient with POD24
 - Grade 4 neurologic events occurred in 2 patients with POD24
 - In patients without POD24, no Grade 4 CRS or neurologic events occurred

Table 3. Summary of Adverse Events in Patients With and Without POD24

AE[a]	With POD24 (n=81)	Without POD24 (n=48)
Any AE, n (%)	80 (99)	48 (100)
Grade ≥3	68 (84)	42 (88)
Grade 5	3 (4)	0
Any serious AE, n (%)	37 (46)	26 (54)
Grade ≥3 cytopenias, n (%)	56 (69)	31 (65)
Grade ≥3 infections, n (%)	12 (15)	10 (21)
CRS, n (%)	66 (81)	42 (88)
Grade ≥3	7 (9)	1 (2)
Median time to onset, days	4	4
Median duration, days	7	5
Neurologic events, n (%)	46 (57)	31 (65)
Grade ≥3	14 (17)	8 (17)
Median time to onset, days	8	7
Median duration, days	11	13

[a] CRS was graded per Lee, et al. 2014.[8] Symptoms of CRS and any other AEs were graded per National Cancer Institute's Common Terminology Criteria for Adverse Events version 4.03.
AE, adverse event; CRS, cytokine release syndrome; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy.

Figure 5. CAR T-Cell Expansion and Key Pretreatment Serum Analytes in Patients With FL by POD24 Status

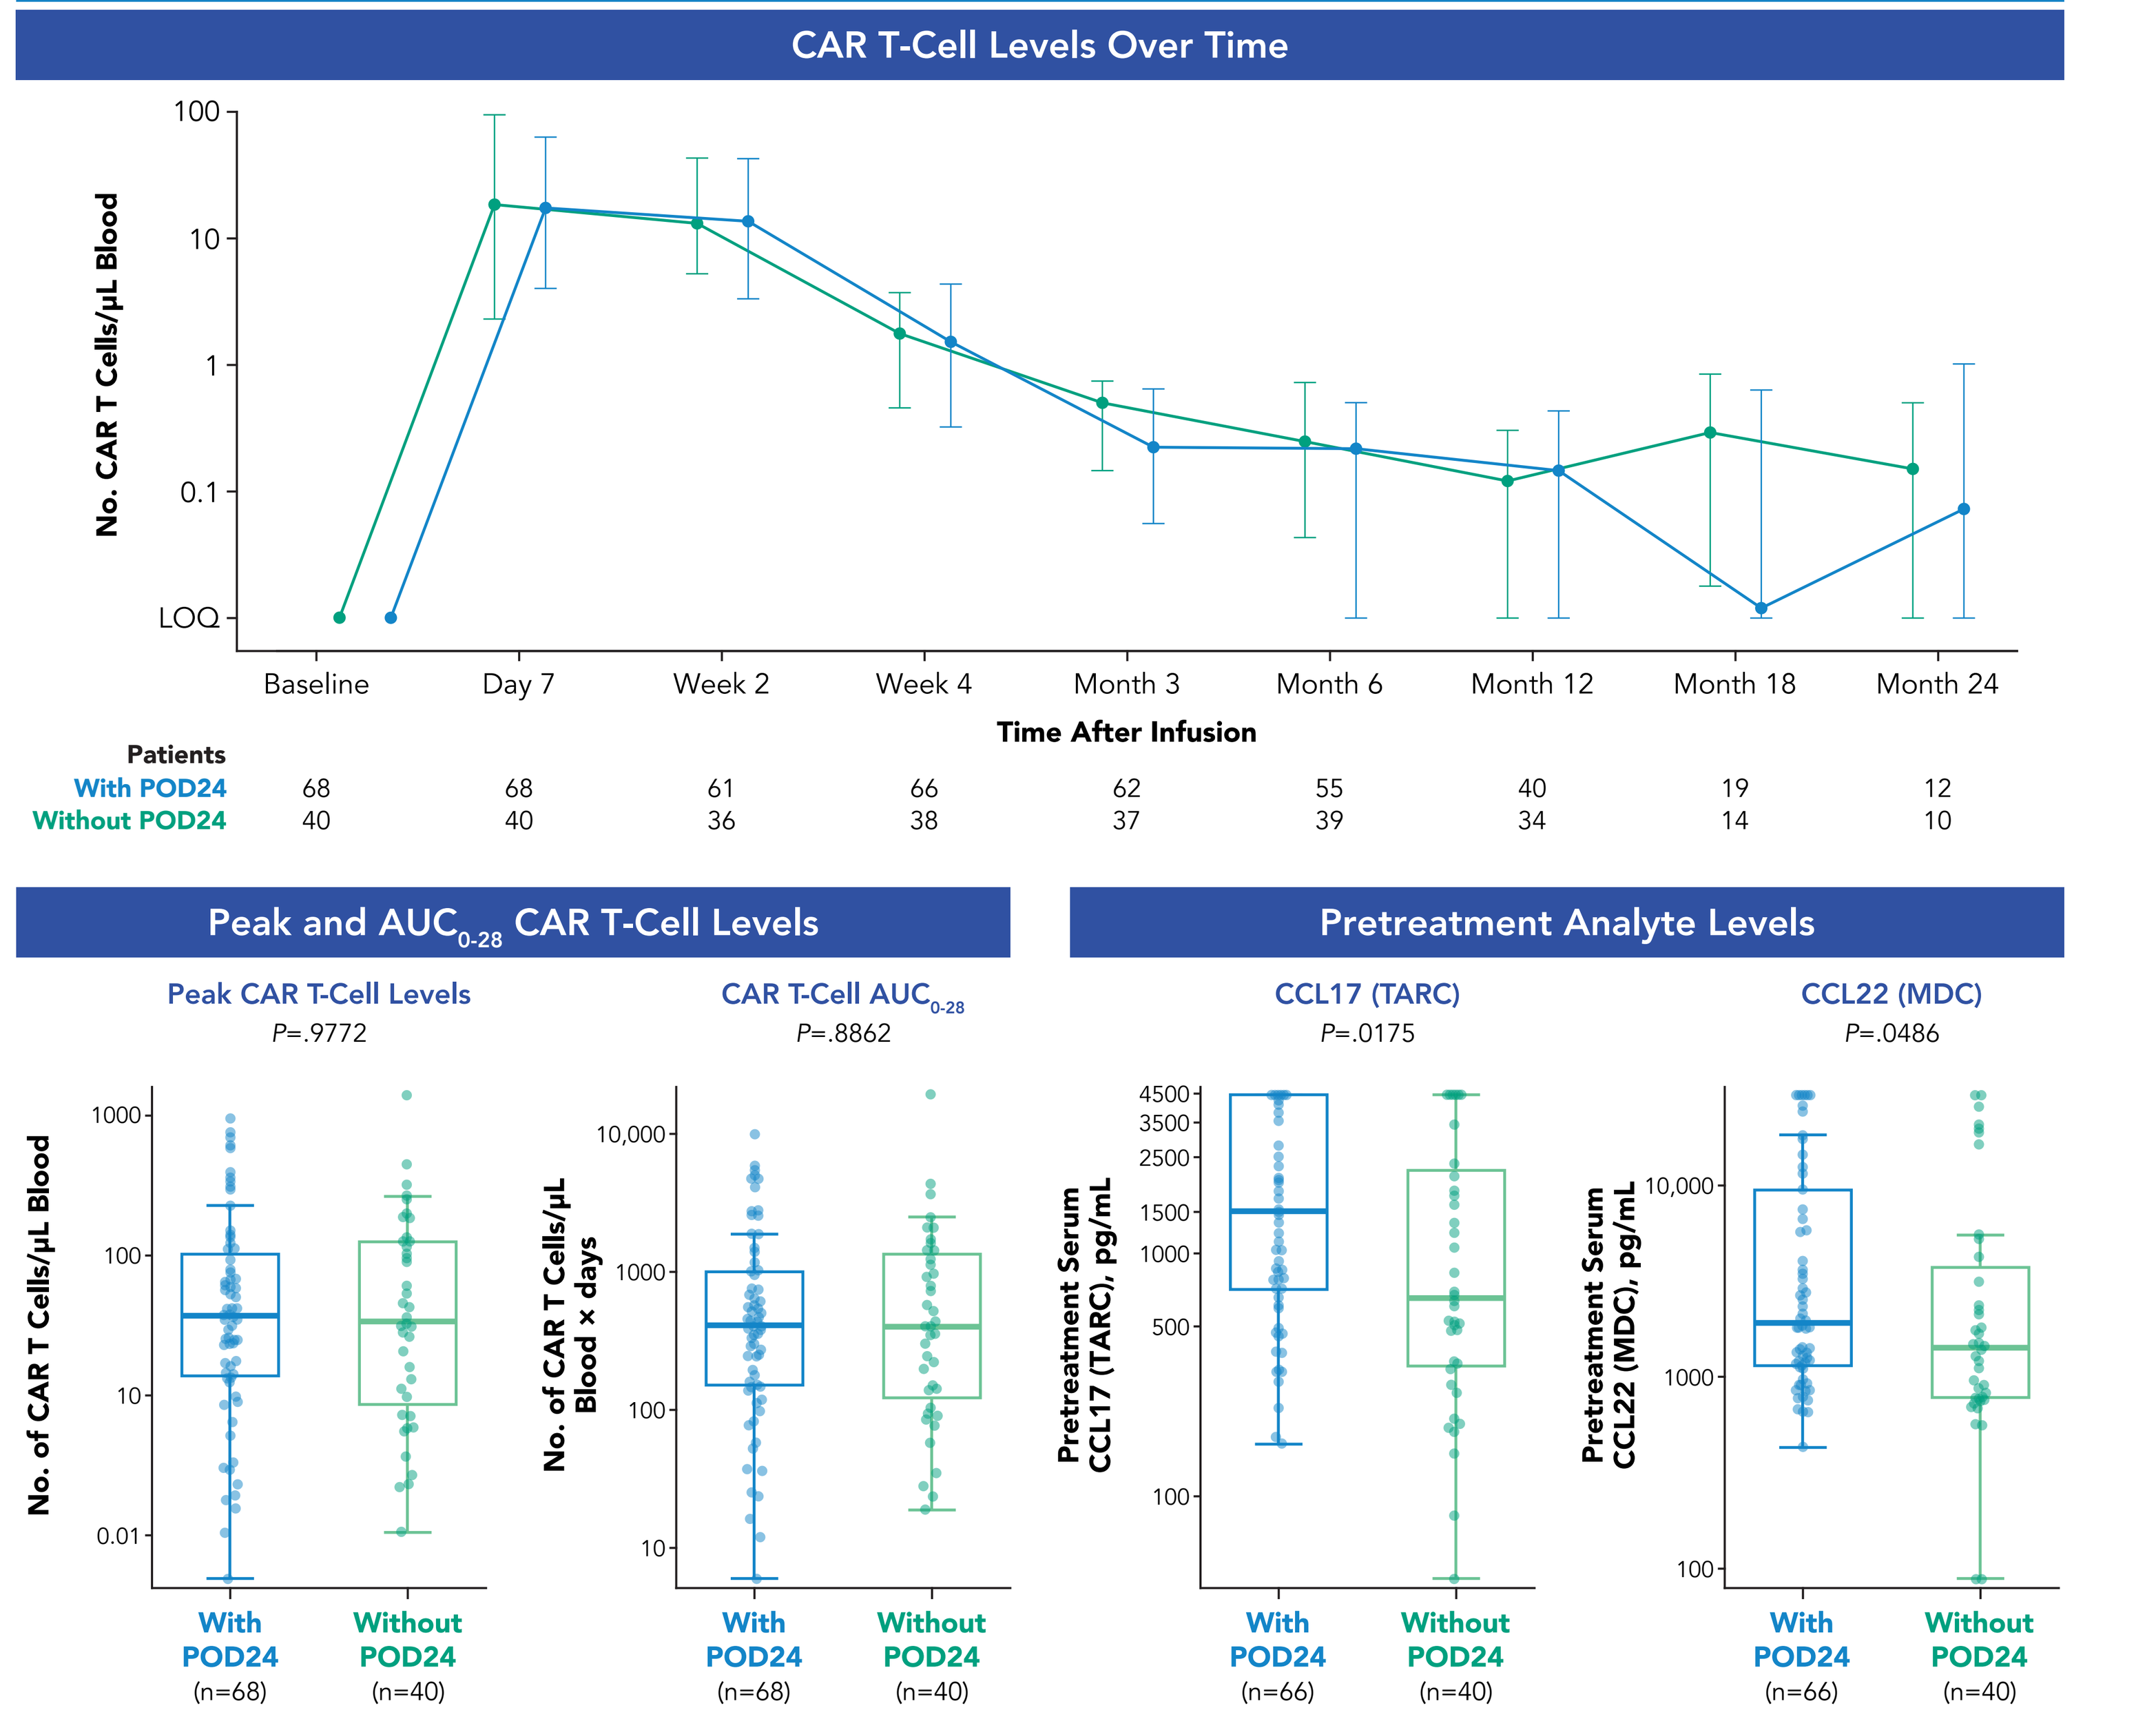

CAR T-Cell Levels Over Time
No. CAR T Cells/µL Blood
LOQ
Baseline
Day 7
Week 2
Week 4
Month 3
Month 6
Month 12
Month 18
Month 24
Time After Infusion
Patients
With POD24 68 68 61 66 62 55 40 19 12
Without POD24 40 40 36 38 37 39 34 14 10
Peak and AUC0-28 CAR T-Cell Levels
Peak CAR T-Cell Levels
P=.9772
No. of CAR T Cells/µL Blood
CAR T-Cell AUC0-28
P=.8862
No. of CAR T Cells/µL Blood × days
With POD24 (n=68)
Without POD24 (n=40)
Pretreatment Analyte Levels
CCL17 (TARC)
P=.0175
Pretreatment Serum CCL17 (TARC), pg/mL
CCL22 (MDC)
P=.0486
Pretreatment Serum CCL22 (MDC), pg/mL
With POD24 (n=66)
Without POD24 (n=40)

P values were calculated using the Wilcoxon rank sum test.
[a] Data were not available for 2 patients with FL before retreatment.
AUC0-28, area under the curve between Day 0 and Day 28; CAR, chimeric antigen receptor; CCL, chemokine (C-C motif) ligand; FL, follicular lymphoma; LOQ, limit of quantification; MDC, macrophage-derived chemokine; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; TARC; thymus- and activation-regulated chemokine.

Table 4. Peak Cytokine Levels in Patients With FL by POD24 Status

Peak Cytokine Levels (Range)	With POD24 (n=68)	Without POD24 (n=40)	*P* value
IL-6, pg/mL	15.0 (1.6[a]–976.0[b])	13.0 (1.6[a]–976.0[b])	.7574
IL-2, pg/mL	3.2 (0.9[a]–90.6)	2.7 (0.9[a]–63.7)	.6181
IFN-γ, pg/mL	122.8 (7.5[a]–1876.0[b])	67.8 (7.5[a]–1545.1)	.0789
Granzyme B, pg/mL	7.8 (1.0[a]–1062.7)	8.2 (1.0[a]–449.4)[c]	.9447
CXCL10, pg/mL	1001.9 (264.4–2000.0[b])	1093.2 (165.9–2000.0[b])	.5385
IL-10, pg/mL	10.7 (0.7[a]–331.5)	7.4 (0.7[a]–66.5)	.2255
TNF-α, pg/mL	4.6 (1.6–62.7)	4.1 (0.7[a]–11.0)	.1644
IL-1RA, pg/mL	1202.0 (221.0–9000.0[b])	982.5 (239.0–9000.0[b])	.2983
GM-CSF, pg/mL	1.9[a] (1.9[a]–23.2)	1.9[a] (1.9[a]–34.1)	.7177
CCL2 (MCP-1), pg/mL	790.1 (249.3–1500.0[b])	829.2 (249.4–1500.0[b])	.9975
IL-15, pg/mL	33.8 (9.3–93.3)	34.5 (12.0–104.3)	.5908
Ferritin, ng/mL	720.2 (86.4–5237.8)	658.9 (91.3–3459.9)	.4900
SAA, pg/mL	1.5×10⁸ (1.5×10⁶–1.4×10⁹[b])	1.7×10⁸ (4.9×10⁶–1.4×10⁹[b])	.9695
CRP, mg/L	68.3 (3.9–496.0[b])	71.2 (2.9–377.8)	.8164

P values were calculated using the Wilcoxon rank sum test.
[a] Lower limit of quantification in assay used. [b] Upper limit of quantification in assay used. [c] Data were not available for 2 patients with FL at peak.
CCL, chemokine (C-C motif) ligand; CRP, C-reactive protein; CXCL, C-X-C motif chemokine ligand; FL, follicular lymphoma; GM-CSF, granulocyte-macrophage colony-stimulating factor; IFN, interferon; IL, interleukin; MCP-1, monocyte chemoattractant protein 1; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy; RA, receptor agonist; SAA, serum amyloid A; TNF, tumor necrosis factor.

- In efficacy-evaluable patients with FL, median peak CAR T-cell levels were similar in patients with and without POD24 (36.9 cells/µL and 34.5 cells/µL, respectively; **Figure 5**)
 - Median AUCs were also similar among patients with and without POD24 (422.5 cells/µL × days and 407.6 cells/µL × days, respectively)
- Pretreatment levels of CCL17 (TARC) and CCL22 (MDC) appeared higher in patients with POD24 than without POD24 (**Figure 5**)
- Peak levels of key biomarkers associated with axi-cel toxicity appeared generally similar in all treated patients with and without POD24 (**Table 4**)
 - Pharmacokinetic/pharmacodynamic findings between groups were similar in patients with MZL
- Of the 14 patients (13 FL; 1 MZL) in broader ZUMA-5 population with available data at relapse after axi-cel, 100% had detectable CD19
 - Detectable CD19 was confirmed in all evaluable biopsies from patients with and without POD24

Table 5. Axi-Cel Product Characteristics in Patients with FL by POD24 Status

Characteristic (range)	With POD24 (n=68)	Without POD24 (n=40)
No. CCR7+CD45RA+ T cells, 10⁶	38.5 (6.4–268.6)[a]	46.6 (1.1–296.6)[a]
CD4/CD8 ratio	0.7 (0.1–31.3)[a]	0.8 (0.1–13.1)[a]
Transduction rate, %	60.5 (18.0–86.0)	60.5 (26.0–77.0)
IFN-γ in coculture, pg/mL	5511.0 (753.0–1.9×10⁴)	6315.0 (1267.0–1.8×10⁴)

[a] Based on available data: with POD24, n=57; without POD24, n=36 (CCR7+CD45RA+ cells) and n=35 (CD4/CD8 ratio).
Axi-cel, axicabtagene ciloleucel; FL, follicular lymphoma; IFN, interferon; POD24, progression of disease <24 months from initiating the first anti-CD20–containing chemoimmunotherapy.

- Axi-cel product attributes were generally similar among patients with and without POD24 (**Table 5**)

CONCLUSIONS

- Axi-cel demonstrated a high rate of durable responses in patients with POD24 iNHL
 - Although medians for PFS were not reached in either group, estimated PFS rates at 18 months appeared lower in patients with POD24 than those without POD24
 - Among patients with FL, higher median pretreatment levels of analytes previously associated with relapse (CC17 [TARC] and CCL22 [MDC])[9] were observed in patients with POD24 than without POD24, potentially contributing to differences in the 18-month PFS rate
- Safety profiles were similarly manageable in patients with and without POD24
- Among patients with FL, posttreatment pharmacokinetic and pharmacodynamic profiles appeared largely comparable in patients with and without POD24
- Axi-cel may be a promising option for patients with POD24 iNHL, a population with particularly high-risk disease[1]

REFERENCES

1. Casulo C and Barr P. *Blood*. 2019; 133(14):1540-1547
2. Casulo C, et al. *J Clin Oncol*. 2015; 33(23): 2516–2522.
3. YESCARTA® (axicabtagene ciloleucel) Prescribing information. Kite Pharma, Inc; 2021.
4. YESCARTA® (axicabtagene ciloleucel) [summary of product characteristics]. Amsterdam, The Netherlands: Kite Pharma EU B.V.; 2018.
5. Jacobson CA, et al. ASH 2020. #1187.
6. Jacobson CA, et al. ASH 2020. #700.
7. Cheson BD, et al. *J Clin Oncol*. 2014;32:3059-68.
8. Lee DW, et al. *Blood*. 2014;124:188-195.
9. Plaks V, et al. AACR 2021. #CT036

ACKNOWLEDGMENTS

- The patients, families, friends, and caregivers
- The study investigators, coordinators, and health care staff at each study site
- The authors thank Lisa Johnson, PhD; Justin Chou, PhD; Emily Marsh; Marika Sherman, MS; and Akshay Sudhindra, MD, of Kite, a Gilead Company, for their contributions to this analysis
- Medical writing support was provided by Danielle Luebke, PhD, of Nexus Global Group Science, funded by Kite, a Gilead Company
- This study was funded by Kite, a Gilead Company

DISCLOSURES

CAJ: honoraria from Kite, a Gilead Company, Bristol Myers Squibb, Celgene, Novartis, Humanigen, Precision BioSciences, Bluebird Bio, Nkarta, Lonza, and AbbVie; consultancy or advisory role for Kite, a Gilead Company, Celgene, Novartis, Bristol Myers Squibb, Precision BioSciences, Nkarta, Lonza, Pfizer, Humanigen, AbbVie, and Bluebird Bio; speakers' bureau participation for Axis and Clinical Care Options; research funding from Kite, a Gilead Company, and Pfizer; and travel support from Kite, a Gilead Company, Celgene, Novartis, Bristol Myers Squibb, Precision Biosciences, Lonza, Pfizer, and Humanigen. **JCC:** consultancy or advisory role for MorphoSys, Bayer, Karyopharm, Kite, a Gilead Company, Novartis, Janssen, AbbVie, TeneoBio, and Pfizer; speakers' bureau participation for MorphoSys, AstraZeneca, BeiGene, Genentech, Kite, a Gilead Company, and Epizyme; and research funding from Merck. **BMW:** consulting or advisory role with Celgene, Kyowa Kirin, and Guidepoint Global; research funding from Incyte, Dova, Merck, and Seattle Genetics. **JM:** honoraria from Kyowa Kirin and Seattle Genetics; consultancy or advisory role for Pharmacyclics, Bayer, Kite, a Gilead Company, Pfizer, Janssen, Juno/Celgene, Bristol Myers Squibb, Kyowa Kirin, Alexion, Fosun Kite, Innovent, Seattle Genetics, and BeiGene; speakers' bureau participation for Kite, a Gilead Company, Kyowa Kirin, Bayer, Pharmacyclics/Janssen, Seattle Genetics, Acrotech/Aurobindo, BeiGene, Verastem, AstraZeneca, Juno/Celgene/Bristol Myers Squibb, Genentech/Roche, and AbbVie; research funding from Bayer, Kite, a Gilead Company, Celgene, Merck, Portola Pharmaceuticals, Incyte, Genentech, Pharmacyclics, Seattle Genetics, Janssen, and Millennium. **GS:** honoraria from Amgen; consultancy or advisory role for AbbVie, Autolus, Allogene, BeiGene, Celgene/Bristol Myers Squibb, Debiopharm, Genmab, Kite, a Gilead Company, Epizyme, Janssen, Karyopharm, Miltenyi, MorphoSys, Novartis, Roche, Takeda, VelosBio, BeiGene, and Inctye; and participation in educational events for AbbVie, Celgene, Gilead, Janssen, Kite, a Gilead Company, MorphoSys, Novartis, and Roche. **PNM:** honoraria from Kite, a Gilead Company, and Incyte; speakers' bureau participation for Kite, a Gilead Company, and Incyte. **CC:** no relevant financial relationship(s) to disclose. **DGM:** stock or other ownership in A2 Biotherapeutics; honoraria from Bioline RX, Juno Therapeutics, Celgene, Kite, a Gilead Company, Gilead Sciences, Novartis, and Pharmacyclics; consultancy or advisory role for A2 Biotherapeutics; research funding from Kite, a Gilead Company, Juno Therapeutics, and Celgene; and patents, royalties, or other intellectual property from Juno Therapeutics. **SdV:** consultancy or advisory role for Bayer and Verastem. **RR:** honoraria from Gilead and Novartis; consultancy or advisory role for Magenta, Atara, Celgene and Gilead; research funding from Gilead, Bristol Myers Squibb, Takeda, Incyte, Pharmacyclics, Immatics, Shire, Bluebird, Atara and Kiadis; expert testimony provided for Monsanto; and travel support from Gilead. **LAL:** honoraria from Kite, a Gilead Company, AstraZeneca, Seattle Genetics, Epizyme, Karyopharm, Celgene, Bristol Myers Squibb, PCYC, and BeiGene; consulting or advisory role with TG Therapeutics, AbbVie, PCYC, Kite, a Gilead Company, Celgene, Bristol Myers Squibb, and ADC Therapeutics; speakers' bureau with Kite, a Gilead Company, Celgene, Bristol Myers Squibb, BeiGene, PCYC, Janssen, AstraZeneca, Seattle Genetics, Karyopharm, and Epizyme. **IY-A:** honoraria from Kite, a Gilead Company, Novartis, and Celgene. **OOO:** consultancy or advisory role for Kite, a Gilead Company, Pfizer, Spectrum Pharmaceuticals, Legend and Bayer. **HCHF:** personal fees from Janssen Oncology, Takeda, Incyte, Amgen, Celgene/Bristol Myers Squibb, Pfizer, Sanofi, AbbVie, Karyopharm, AstraZeneca and Kite, a Gilead Company. **VP and YY:** employment with Kite, a Gilead Company. **JL:** employment with Kite, a Gilead Company; stock or other ownership in Gilead Sciences. **MPA:** previous employment with Kite, a Gilead Company; stock or other ownership in Gilead Sciences; patents, royalties, or other intellectual property from MSKCC and NYBC; and travel support from Kite, a Gilead Company. **SSN:** personal fees from Kite, a Gilead Company, Merck, Bristol Myers Squibb, Novartis, Celgene, Pfizer, Allogene Therapeutics, Cell Medica/Kuur, Incyte, Precision Biosciences, Legend Biotech, Adicet Bio, Calibr, and Unum Therapeutics; research support from Kite, a Gilead Company, Bristol Myers Squibb, Merck, Poseida, Cellectis, Celgene, Karus Therapeutics, Unum Therapeutics, Allogene Therapeutics, Precision Biosciences, and Acerta; and royalties from Takeda Pharmaceuticals; intellectual property related to cell therapy.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.