Poster
P104

Patient-Reported Outcomes in ZUMA-7, a Phase 3, Randomized, Open-Label Study Evaluating the Efficacy of Axicabtagene Ciloleucel (Axi-Cel) Versus Standard-of-Care Therapy in Relapsed/Refractory Large B-Cell Lymphoma

Mahmoud Elsawy, MD, MSc[1]; Julio C. Chavez, MD[2]; Irit Avivi, MD[3]; Jean-François Larouche, MD[4]; Luciano Wannesson, MD[5]; Kate Cwynarski, FCRPATH, PhD[6]; Keren Osman, MD[7]; Kelly Davison, MD, PhD[8]; Jakob D. Rudzki, MD[9]; Saurabh Dahiya, MD, FACP[10]; Kathleen Dorritie, MD[11]; Samantha Jaglowski, MD[12]; John Radford, MD, FRCP, FMedSci[13]; Franck Morschhauser, MD, PhD[14]; David Cunningham, MD, FRCP, FMedSci[15]; Alejandro Martin Garcia-Sancho, MD[16]; Dimitrios Tzachanis, MD, PhD[17]; Matthew L. Ulrickson, MD[18]; Reem Karmali, MD[19]; Natasha Kekre, BSc, MD, MPH, FRCPC[20]; Catherine Thieblemont, MD, PhD[21]; Gunilla Enblad, MD, PhD[22]; Peter Dreger, MD, PhD[23]; Ram Malladi, MD[24]; Namita Joshi, PhD[25]; Wei-Jhih Wang, PhD[25]; Caitlyn T. Solem, PhD[25]; Julia Thornton Snider, PhD[26]; Christina To, MD[26]; and Marie José Kersten, MD, PhD[27]

[1]Queen Elizabeth II Health Sciences Centre and Dalhousie University, Halifax, NS, Canada; [2]Moffitt Cancer Center, Tampa, FL, USA; [3]Tel Aviv Sourasky Medical Center and Sackler Faculty of Medicine, Tel Aviv University, Tel Aviv, Israel; [4]CHU de Québec, Hôpital de l'Enfant-Jésus, Québec, QC, Canada; [5]Istituto Oncologico della Svizzera Italiana, Switzerland; [6]University College London Hospitals NHS Foundation Trust, London, United Kingdom; [7]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [8]McGill University Health Centre, Montreal, QC, Canada; [9]The Medical University of Innsbruck, University Clinic for Internal Medicine, Innsbruck, Austria; [10]University of Maryland Medical Center, Baltimore, MD, USA; [11]UPMC Hillman Cancer Center, Pittsburgh, PA, USA; [12]The Ohio State University, Comprehensive Cancer Center, Columbus, OH, USA; [13]The Christie NHS Foundation Trust and the University of Manchester, Manchester, United Kingdom; [14]University of Lille, CHU Lille, France; [15]The Royal Marsden NHS Foundation Trust, London, United Kingdom; [16]Salamanca University Hospital, IBSAL, CIBERONC, Salamanca, Spain; [17]University of California, San Diego, La Jolla, CA, USA; [18]Banner MD Anderson Cancer Center, Gilbert, AZ, USA; [19]Northwestern University, Chicago, IL, USA; [20]The Ottawa Hospital, Ottawa, ON, Canada; [21]University of Paris, Hôpital Saint-Louis, Hemato-oncology, DMU DHI, Paris, France; [22]Uppsala University Hospital, Uppsala, Sweden; [23]University of Heidelberg, Heidelberg, Germany; [24]Cambridge University Hospitals NHS Foundation Trust, Cambridge, United Kingdom; [25]OPEN Health, Bethesda, MD, USA; [26]Kite, a Gilead Company, Santa Monica, CA, USA; and [27]Amsterdam UMC, University of Amsterdam, Cancer Center Amsterdam, Amsterdam, Netherlands, on behalf of HOVON/LLPC

BACKGROUND

- Outcomes are poor for patients with large B-cell lymphoma (LBCL) who relapse early or are refractory to first-line therapy. Furthermore, patients receiving second-line standard-of-care (SOC) therapy often report poor health-related quality of life (QoL)[1]
- ZUMA-7 (NCT03391466) is a pivotal Phase 3, randomized, open-label, multicenter study of axi-cel (an autologous anti-CD19 chimeric antigen receptor [CAR] T-cell therapy) versus SOC in second-line relapsed/refractory (R/R) LBCL[2]
 - Primary analysis results were presented at ASH 2021 (plenary, December 12)
- Axi-cel is an approved therapy for patients with R/R LBCL after 2 or more lines of therapy
 - ZUMA-1 (NCT02348216) investigated the safety and efficacy of axi-cel in patients with refractory LBCL
 - A long-term follow-up analysis recently presented at ASH 2021 demonstrated a 5-year overall survival rate of 43% after a median follow-up of 63 months[3]

OBJECTIVE

- Here, we report the first comparative analysis of patient-reported outcomes (PROs) with CAR T-cell therapy versus SOC as second-line treatment in R/R LBCL in ZUMA-7

METHODS

Figure 1. ZUMA-7 Study Schema and Endpoints

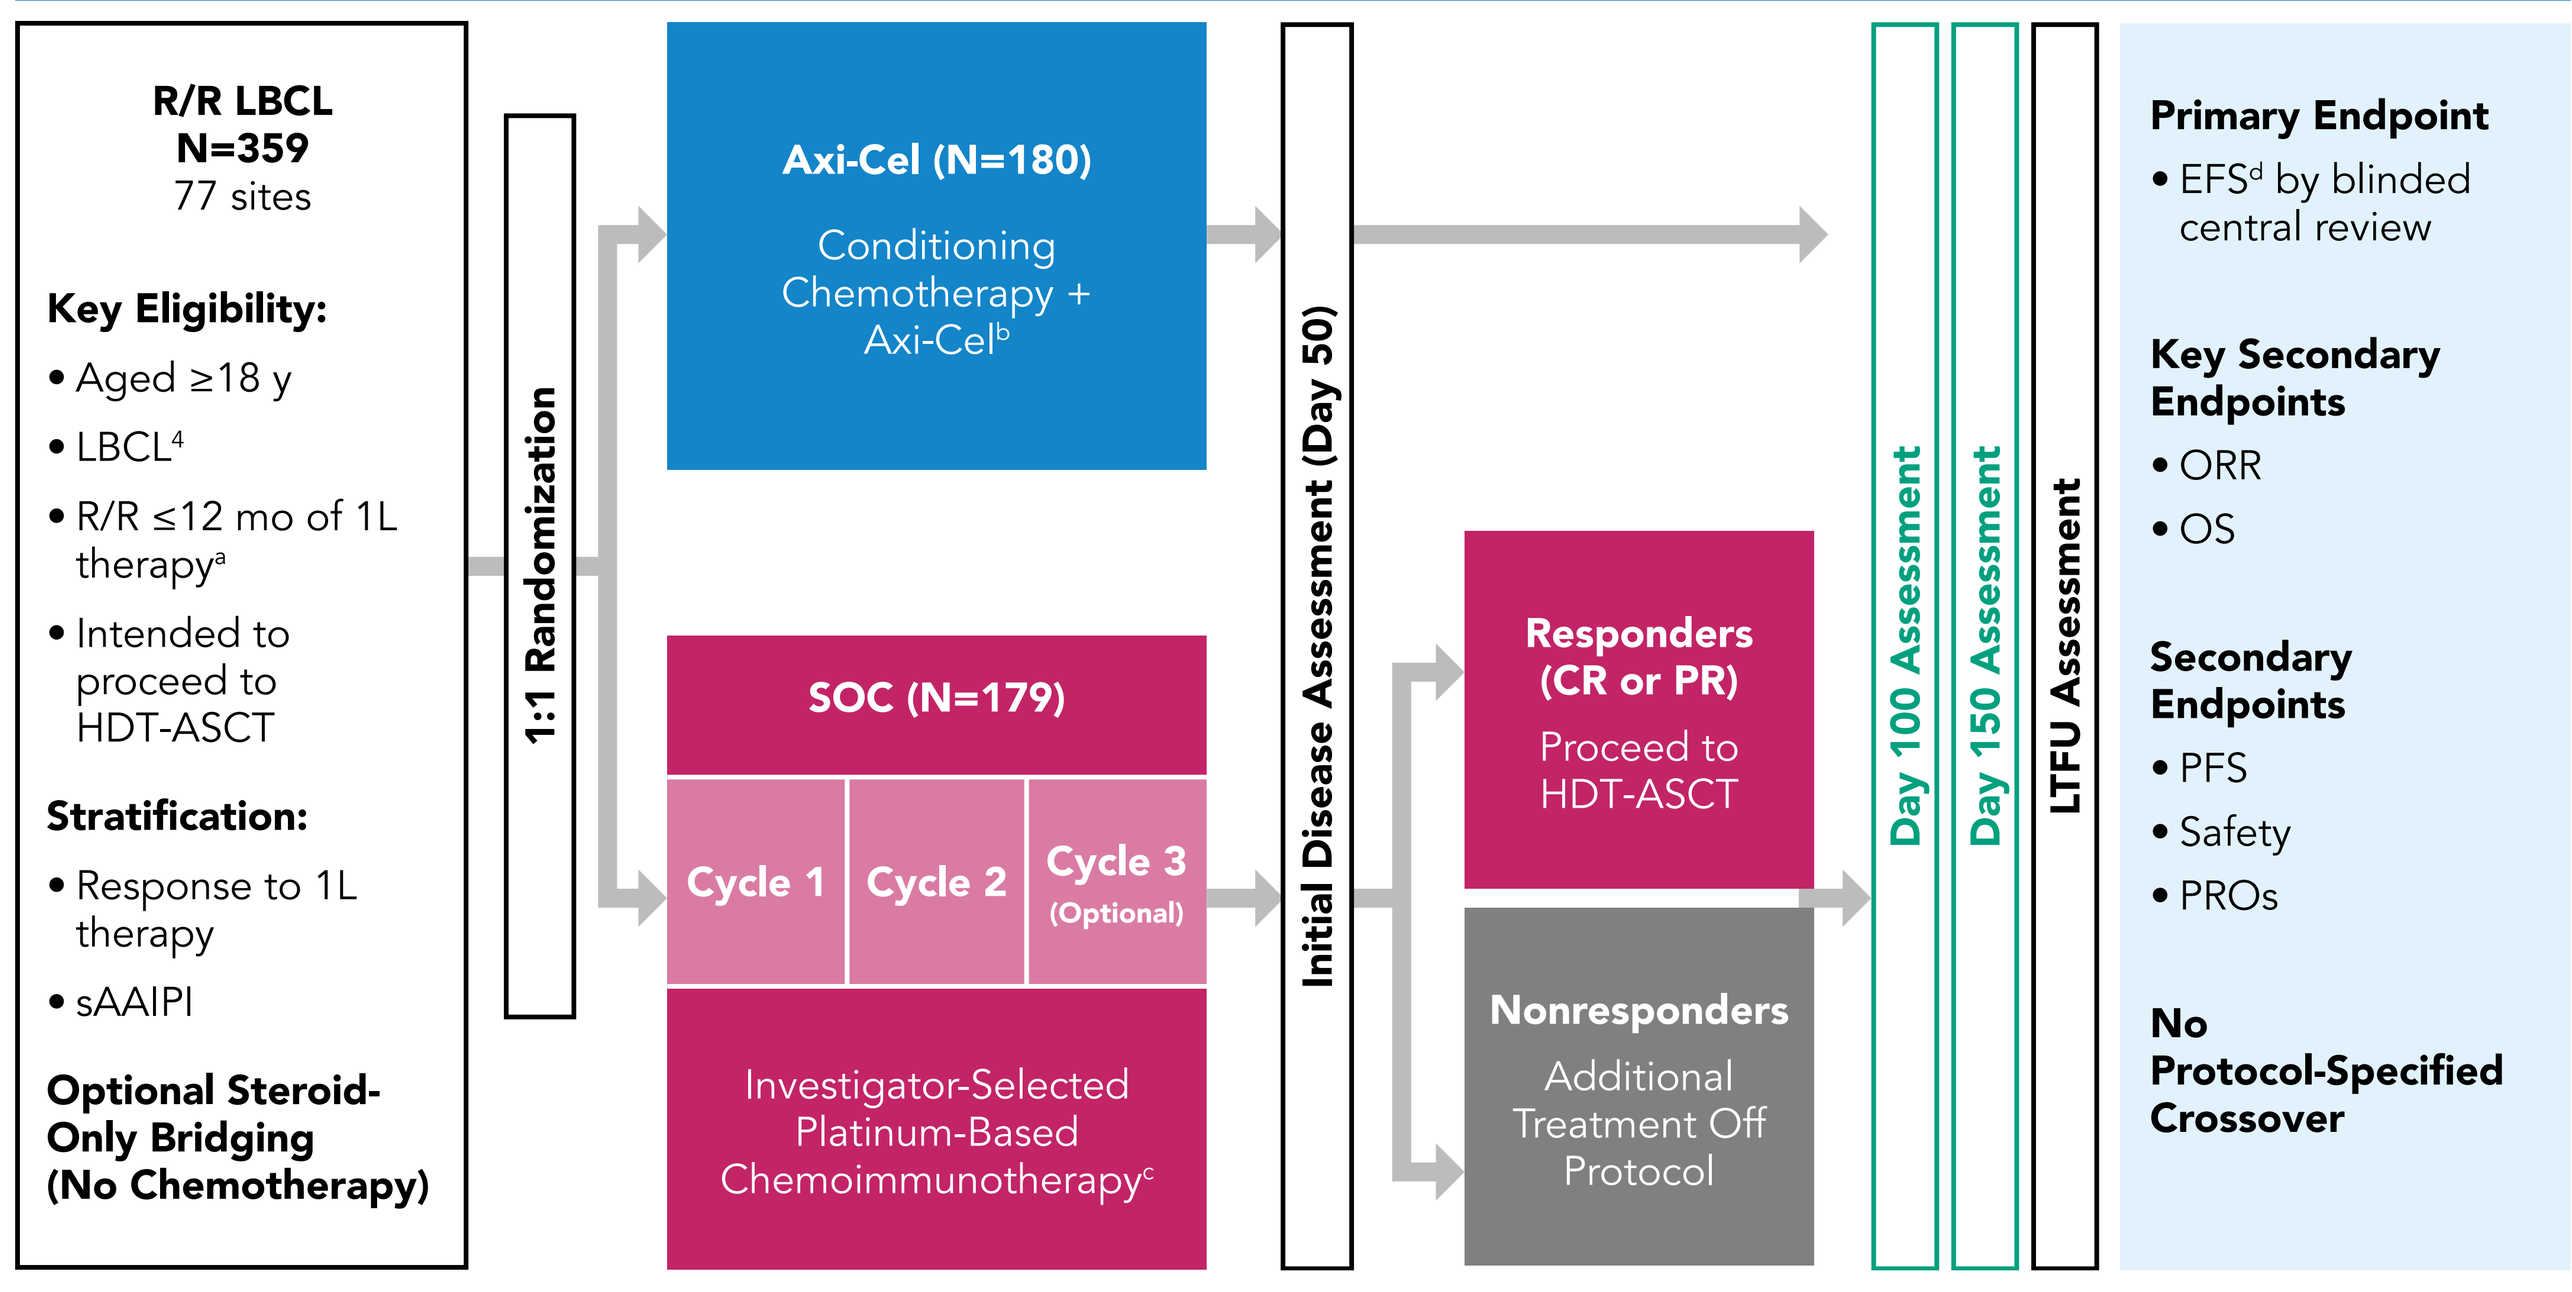
R/R LBCL
N=359
77 sites
Key Eligibility:
• Aged ≥18 y
• LBCL[4]
• R/R ≤12 mo of 1L therapy[a]
• Intended to proceed to HDT-ASCT
Stratification:
• Response to 1L therapy
• sAAIPI
Optional Steroid-Only Bridging (No Chemotherapy)
1:1 Randomization
Axi-Cel (N=180)
Conditioning Chemotherapy + Axi-Cel[b]
SOC (N=179)
Cycle 1
Cycle 2
Cycle 3 (Optional)
Investigator-Selected Platinum-Based Chemoimmunotherapy[c]
Initial Disease Assessment (Day 50)
Responders (CR or PR)
Proceed to HDT-ASCT
Nonresponders
Additional Treatment Off Protocol
Day 100 Assessment
Day 150 Assessment
LTFU Assessment
Primary Endpoint
• EFS[d] by blinded central review
Key Secondary Endpoints
• ORR
• OS
Secondary Endpoints
• PFS
• Safety
• PROs
No Protocol-Specified Crossover

[a] Refractory disease was defined as no CR to 1L therapy; relapsed disease was defined as CR followed by biopsy-proven disease relapse ≤12 months from completion of 1L therapy. [b] Axi-cel patients underwent leukapheresis followed by conditioning chemotherapy with cyclophosphamide (500 mg/m²/day) and fludarabine (30 mg/m²/day) 5, 4, and 3 days before receiving a single axi-cel infusion (target intravenous dose, 2×10⁶ CAR T cells/kg). [c] Protocol-defined SOC regimens included R-GDP, R-DHAP, R-ICE, or R-ESHAP. [d] EFS was defined as time from randomization to the earliest date of disease progression per Lugano Classification,[5] commencement of new lymphoma therapy, or death from any cause.
1L, first-line; axi-cel, axicabtagene ciloleucel; CAR, chimeric antigen receptor; CR, complete response; EFS, event-free survival; HDT-ASCT, high-dose therapy with autologous stem cell rescue; LBCL, large B-cell lymphoma; LTFU, long-term follow-up; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PR, partial response; PRO, patient-reported outcome; R-DHAP, rituximab, dexamethasone, cytarabine, and cisplatin; R-ESHAP, rituximab, etoposide, methylprednisolone, cytarabine, and cisplatin; R-GDP, rituximab, gemcitabine, cisplatin, and dexamethasone; R-ICE, rituximab, ifosfamide, carboplatin, and etoposide; R/R, relapsed/refractory; sAAIPI, second-line age-adjusted International Prognostic Index; SOC, standard of care.

Table 1. PRO Instruments

Instrument	Description	Scales/Domains
EORTC QLC-C30	Cancer-specific 30-item questionnaire, including global health status, functional, and symptom scales[6-7]	• Functional scales: physical, role, emotional, cognitive, and social functioning • Symptom scales: fatigue, nausea and vomiting, pain, dyspnea, insomnia, appetite loss, constipation, diarrhea, and financial difficulties
EQ-5D-5L	General questionnaire with 5 QoL domains plus a global assessment[8-9]	• Mobility, self-care, usual activities, pain/discomfort, and anxiety/depression • VAS rating of global assessment of their current (day of assessment) state of health
WPAI: General Health	Measure of work productivity and activity impairment[10]	• Absenteeism, presenteeism, overall work impairment, and activity impairment

EORTC, European Organization for Research and Treatment of Cancer; PRO, patient-reported outcome; QLQ-C30, Quality of Life Questionnaire-Core 30; QoL, quality of life; VAS, visual analog scale; WPAI, Work Productivity and Activity Impairment.

- Assessments (**Figure 1**, **Table 1**) were taken at baseline (prior to treatment), Day 50, Day 100, Day 150, Month 9, and every 3 months thereafter from randomization up to 24 months or time of event-free survival event (disease progression, death from any cause, or new lymphoma therapy), whichever occurred first
- The analysis population (QoL analysis set) included all patients who had a baseline PRO and ≥1 measure completed at Day 50, Day 100, or Day 150
- Prespecified hypotheses for 3 PRO domains (European Organization for Research and Treatment of Cancer [EORTC] Quality of Life Questionnaire-Core 30 [QLQ-30] Physical Functioning, EORTC QLQ-C30 Global Health Status/QoL, and EQ-5D-5L visual analog scale [VAS]) were tested using a mixed-effect model with repeated measures at Day 100 and subsequent time points if previous time points were statistically significant
- A clinically meaningful change was defined as 10 points for each EORTC QLQ-C30 score, 7 points for EQ-5D-5L VAS score, and 0.06 for the EQ-5D-5L index[11,12]
- Exploratory analyses on other domains of EORTC QLQ-C30 and EQ-5D-5L were also performed[11-13]

RESULTS

Table 2. Baseline Characteristics: QoL Analysis Set

Characteristic, n (%)	Axi-Cel N=165	SOC N=131	Overall N=296
Age ≥65 years	46 (28)	42 (32)	88 (30)
sAAIPI of 2-3	69 (42)	56 (43)	125 (42)
Response to 1L therapy at randomization			
Primary refractory	119 (72)	89 (68)	208 (70)
Relapse ≤12 mo of 1L therapy	46 (28)	42 (32)	88 (30)
Double-/triple-hit status per investigator			
HGBL (double-/triple-hit)	35 (21)	22 (17)	57 (19)
Negative	102 (62)	76 (58)	178 (60)
Not tested	28 (17)	33 (25)	61 (21)

1L, first-line; axi-cel, axicabtagene ciloleucel; HGBL, high-grade B-cell lymphoma; sAAIPI, second-line age-adjusted International Prognostic Index; SOC, standard of care.

- Of 359 patients enrolled in the ZUMA-7 study, 296 patients (82%) had baseline PROs and ≥1 follow-up measure and were included for analysis (QoL analysis set)
- Overall, 70% of patients had primary refractory disease, 42% had high second-line age-adjusted International Prognostic Index (2-3), and 30% were ≥65 years old (**Table 2**)
- Using Global Health Status/QoL as a representative measure of the EORTC QLQ-C30, 208 patients (70%) completed the Day 100 assessment (88% axi-cel; 47% SOC)

RESULTS *(Continued)*

Figure 2. Change From Baseline for Prespecified PRO Endpoints

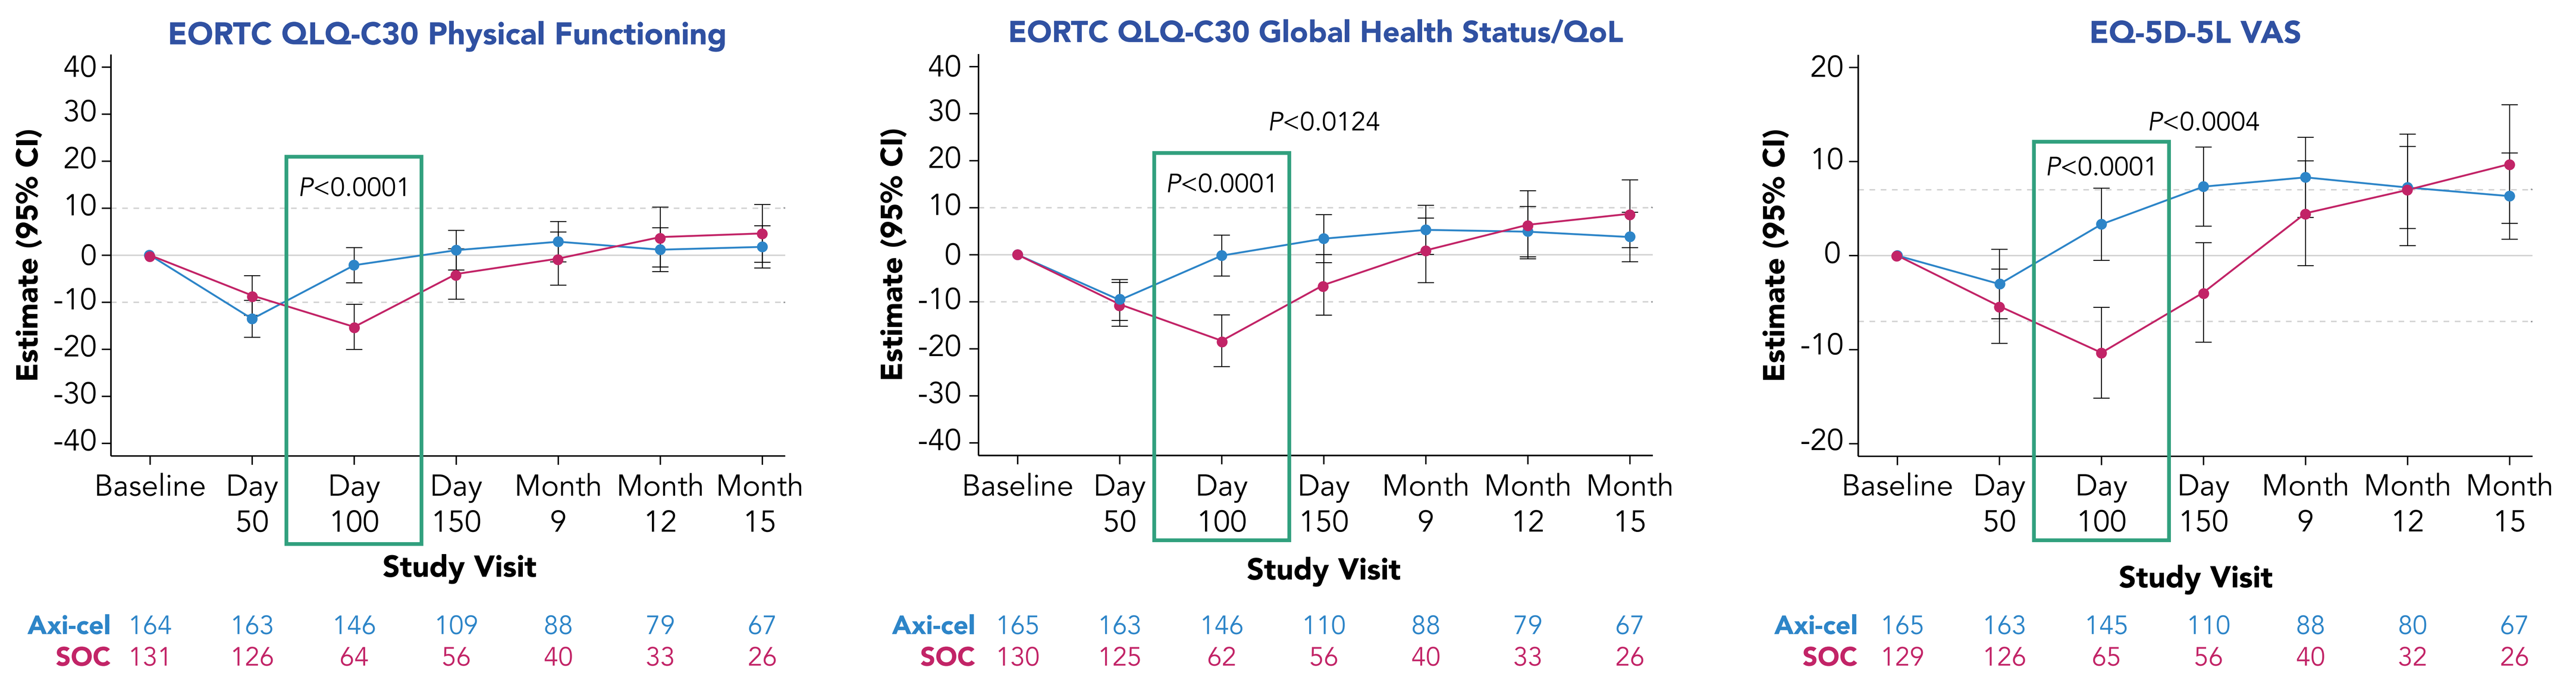
EORTC QLQ-C30 Physical Functioning
P<0.0001
Estimate (95% CI)
Baseline
Day 50
Day 100
Day 150
Month 9
Month 12
Month 15
Study Visit
Axi-cel 164 163 146 109 88 79 67
SOC 131 126 64 56 40 33 26
EORTC QLQ-C30 Global Health Status/QoL
P<0.0001
P<0.0124
Axi-cel 165 163 146 110 88 79 67
SOC 130 125 62 56 40 33 26
EQ-5D-5L VAS
P<0.0001
P<0.0004
Axi-cel 165 163 145 110 88 80 67
SOC 129 126 65 56 40 32 26

Axi-cel, axicabtagene ciloleucel; EORTC, European Organization for Research and Treatment of Cancer; PRO, patient-reported outcome; QLQ-30, Quality of Life Questionnaire-Core 30; QoL, quality of life; SOC, standard of care; VAS, visual analog scale.
Evaluated via mixed-effect model with repeated measures. Statistical significance and clinical meaningfulness coincide for all except for EORTC QLQ-C30 Global Health Status/QoL at Day 150, which was less than a 10-point change (9.8).

- For patients in the QoL analysis set treated with axi-cel versus SOC, there was a statistically significant and clinically meaningful difference in mean change of scores from baseline at Day 100 in favor of axi-cel on all prespecified PRO domains (**Figure 2**)
 - Sensitivity analyses controlling for covariates and patterns of missingness showed similar results with retained significance at Day 100

Figure 3. Return to Baseline for Prespecified PRO Endpoints

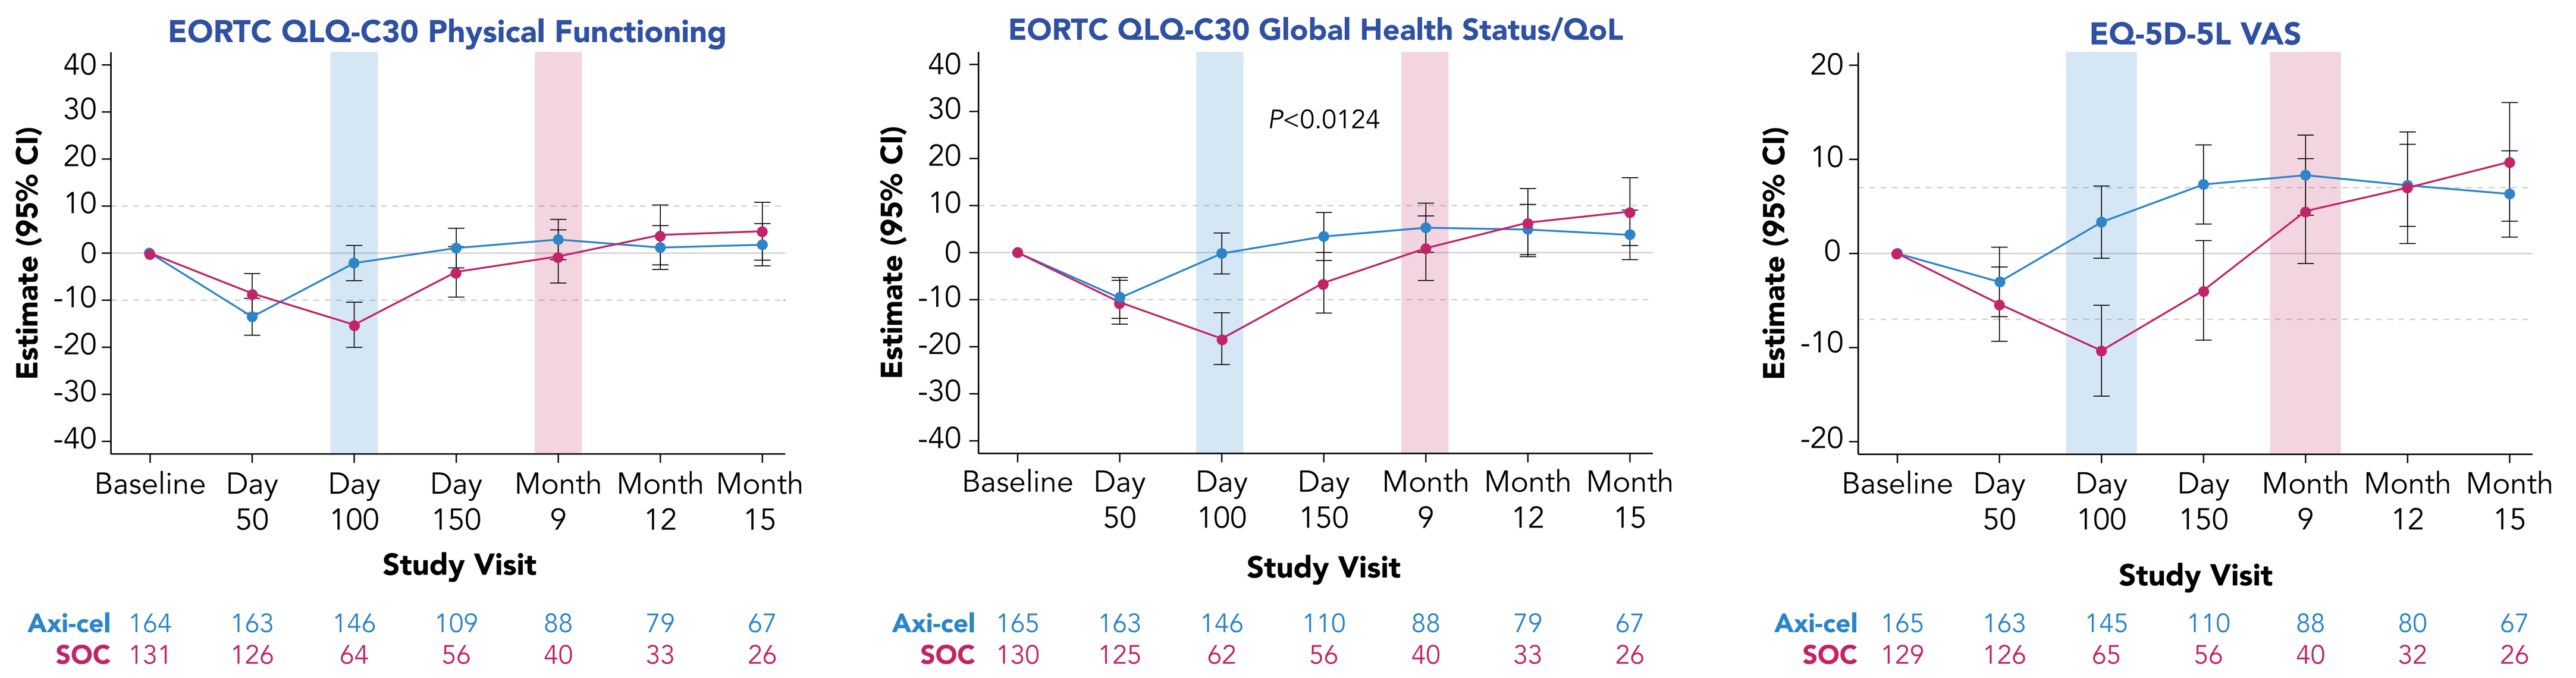
EORTC QLQ-C30 Physical Functioning
Estimate (95% CI)
Baseline
Day 50
Day 100
Day 150
Month 9
Month 12
Month 15
Study Visit
Axi-cel 164 163 146 109 88 79 67
SOC 131 126 64 56 40 33 26
EORTC QLQ-C30 Global Health Status/QoL
P<0.0124
Axi-cel 165 163 146 110 88 79 67
SOC 130 125 62 56 40 33 26
EQ-5D-5L VAS
Axi-cel 165 163 145 110 88 80 67
SOC 129 126 65 56 40 32 26

Axi-cel, axicabtagene ciloleucel; EORTC, European Organization for Research and Treatment of Cancer; PRO, patient-reported outcome; QLQ-30, Quality of Life Questionnaire-Core 30; QoL, quality of life; SOC, standard of care; VAS, visual analog scale.
Evaluated via mixed-effect model with repeated measures. Statistical significance and clinical meaningfulness coincide for all except for EORTC QLQ-C30 Global Health Status/QoL at Day 150, which was less than a 10-point change (9.8). Shading indicates when the QoL measure is not statistically different from baseline.

- The mean estimated scores for the axi-cel arm returned to or exceeded scores at baseline by Day 100 – Day 150 (Months 3 – 5) versus Month 9 or later for the SOC arm (**Figure 3**)

Figure 4. Change From Baseline for EORTC QLQ-C30 Functional Scales

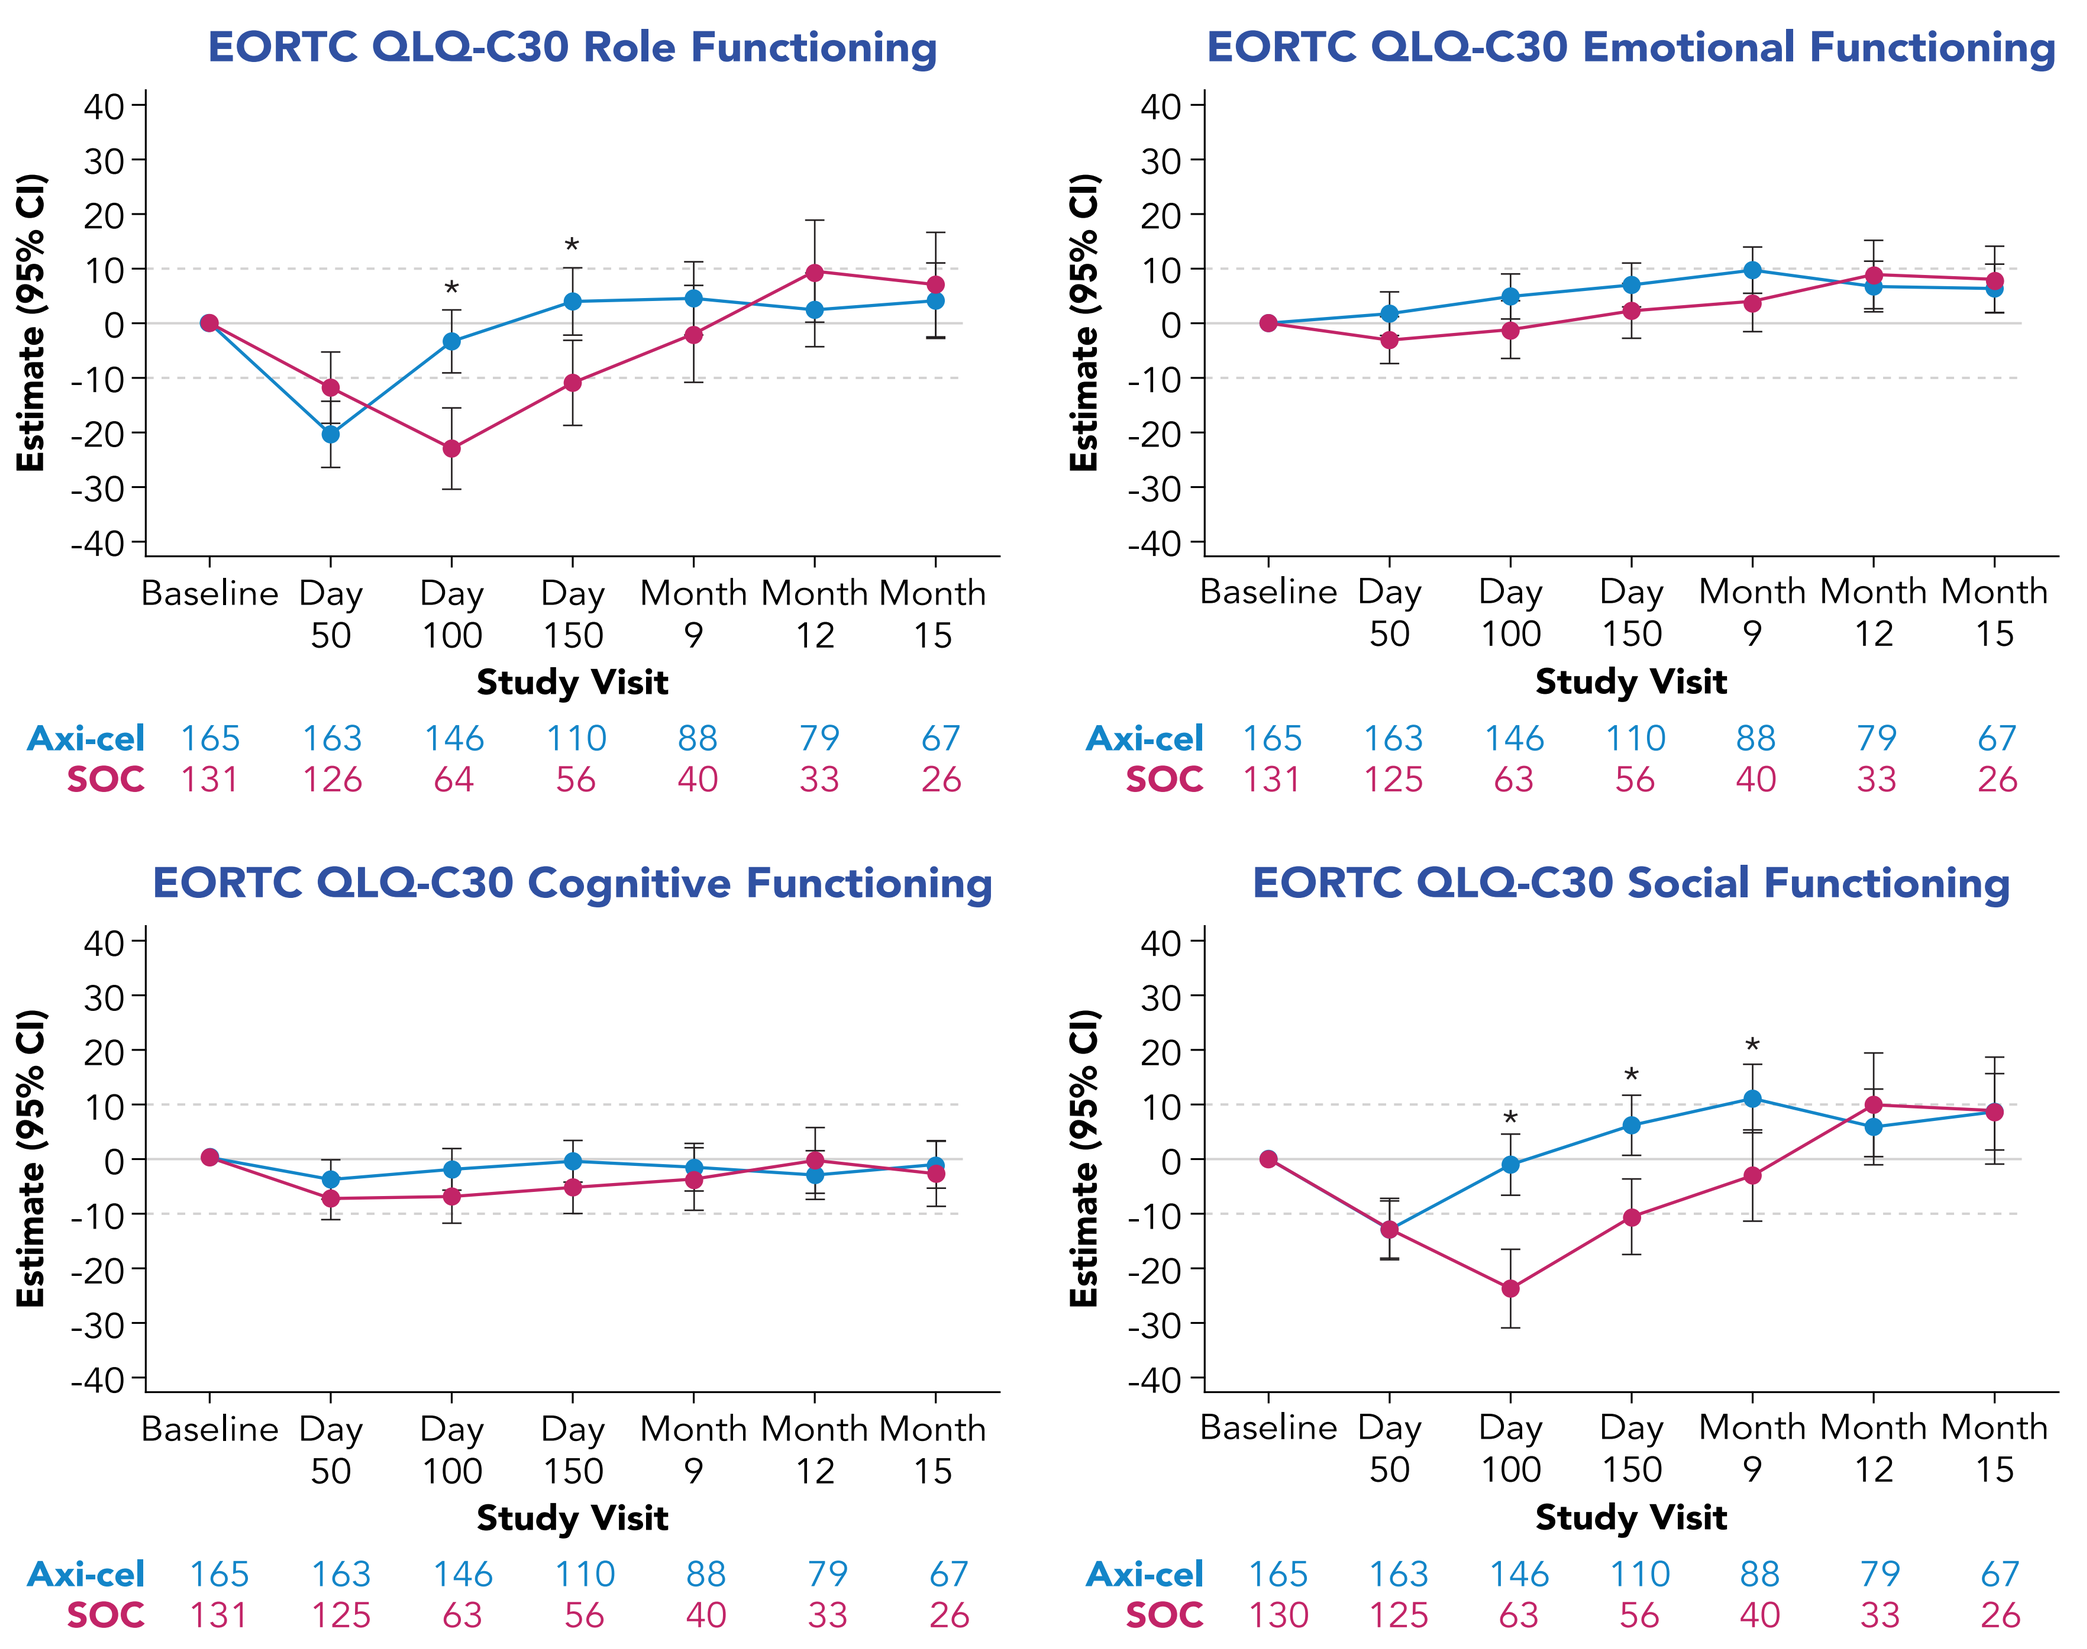
EORTC QLQ-C30 Role Functioning
Estimate (95% CI)
Baseline
Day 50
Day 100
Day 150
Month 9
Month 12
Month 15
Study Visit
Axi-cel 165 163 146 110 88 79 67
SOC 131 126 64 56 40 33 26
EORTC QLQ-C30 Emotional Functioning
Axi-cel 165 163 146 110 88 79 67
SOC 131 125 63 56 40 33 26
EORTC QLQ-C30 Cognitive Functioning
Axi-cel 165 163 146 110 88 79 67
SOC 131 125 63 56 40 33 26
EORTC QLQ-C30 Social Functioning
Axi-cel 165 163 146 110 88 79 67
SOC 130 125 63 56 40 33 26

*P<0.05.
Axi-cel, axicabtagene ciloleucel; EORTC, European Organization for Research and Treatment of Cancer; QLQ-C30, Quality of Life Questionnaire-Core 30; SOC, standard of care.
Evaluated via mixed-effect model with repeated measures. Symptom scales not shown.

- Additional exploratory analyses of PRO endpoints also showed improvements with axi-cel over SOC (**Figure 4**)
- The differences in change from baseline were statistically significant (P<0.05) in favor of axi-cel for
 - Nausea and vomiting, diarrhea, insomnia, and appetite loss measures at Day 100
 - Role functioning at Day 100 and Day 150
 - Social functioning, fatigue, and dyspnea measures at Day 100, Day 150, and Month 9

Figure 5. Change From Baseline for Work Productivity and Activity Impairment

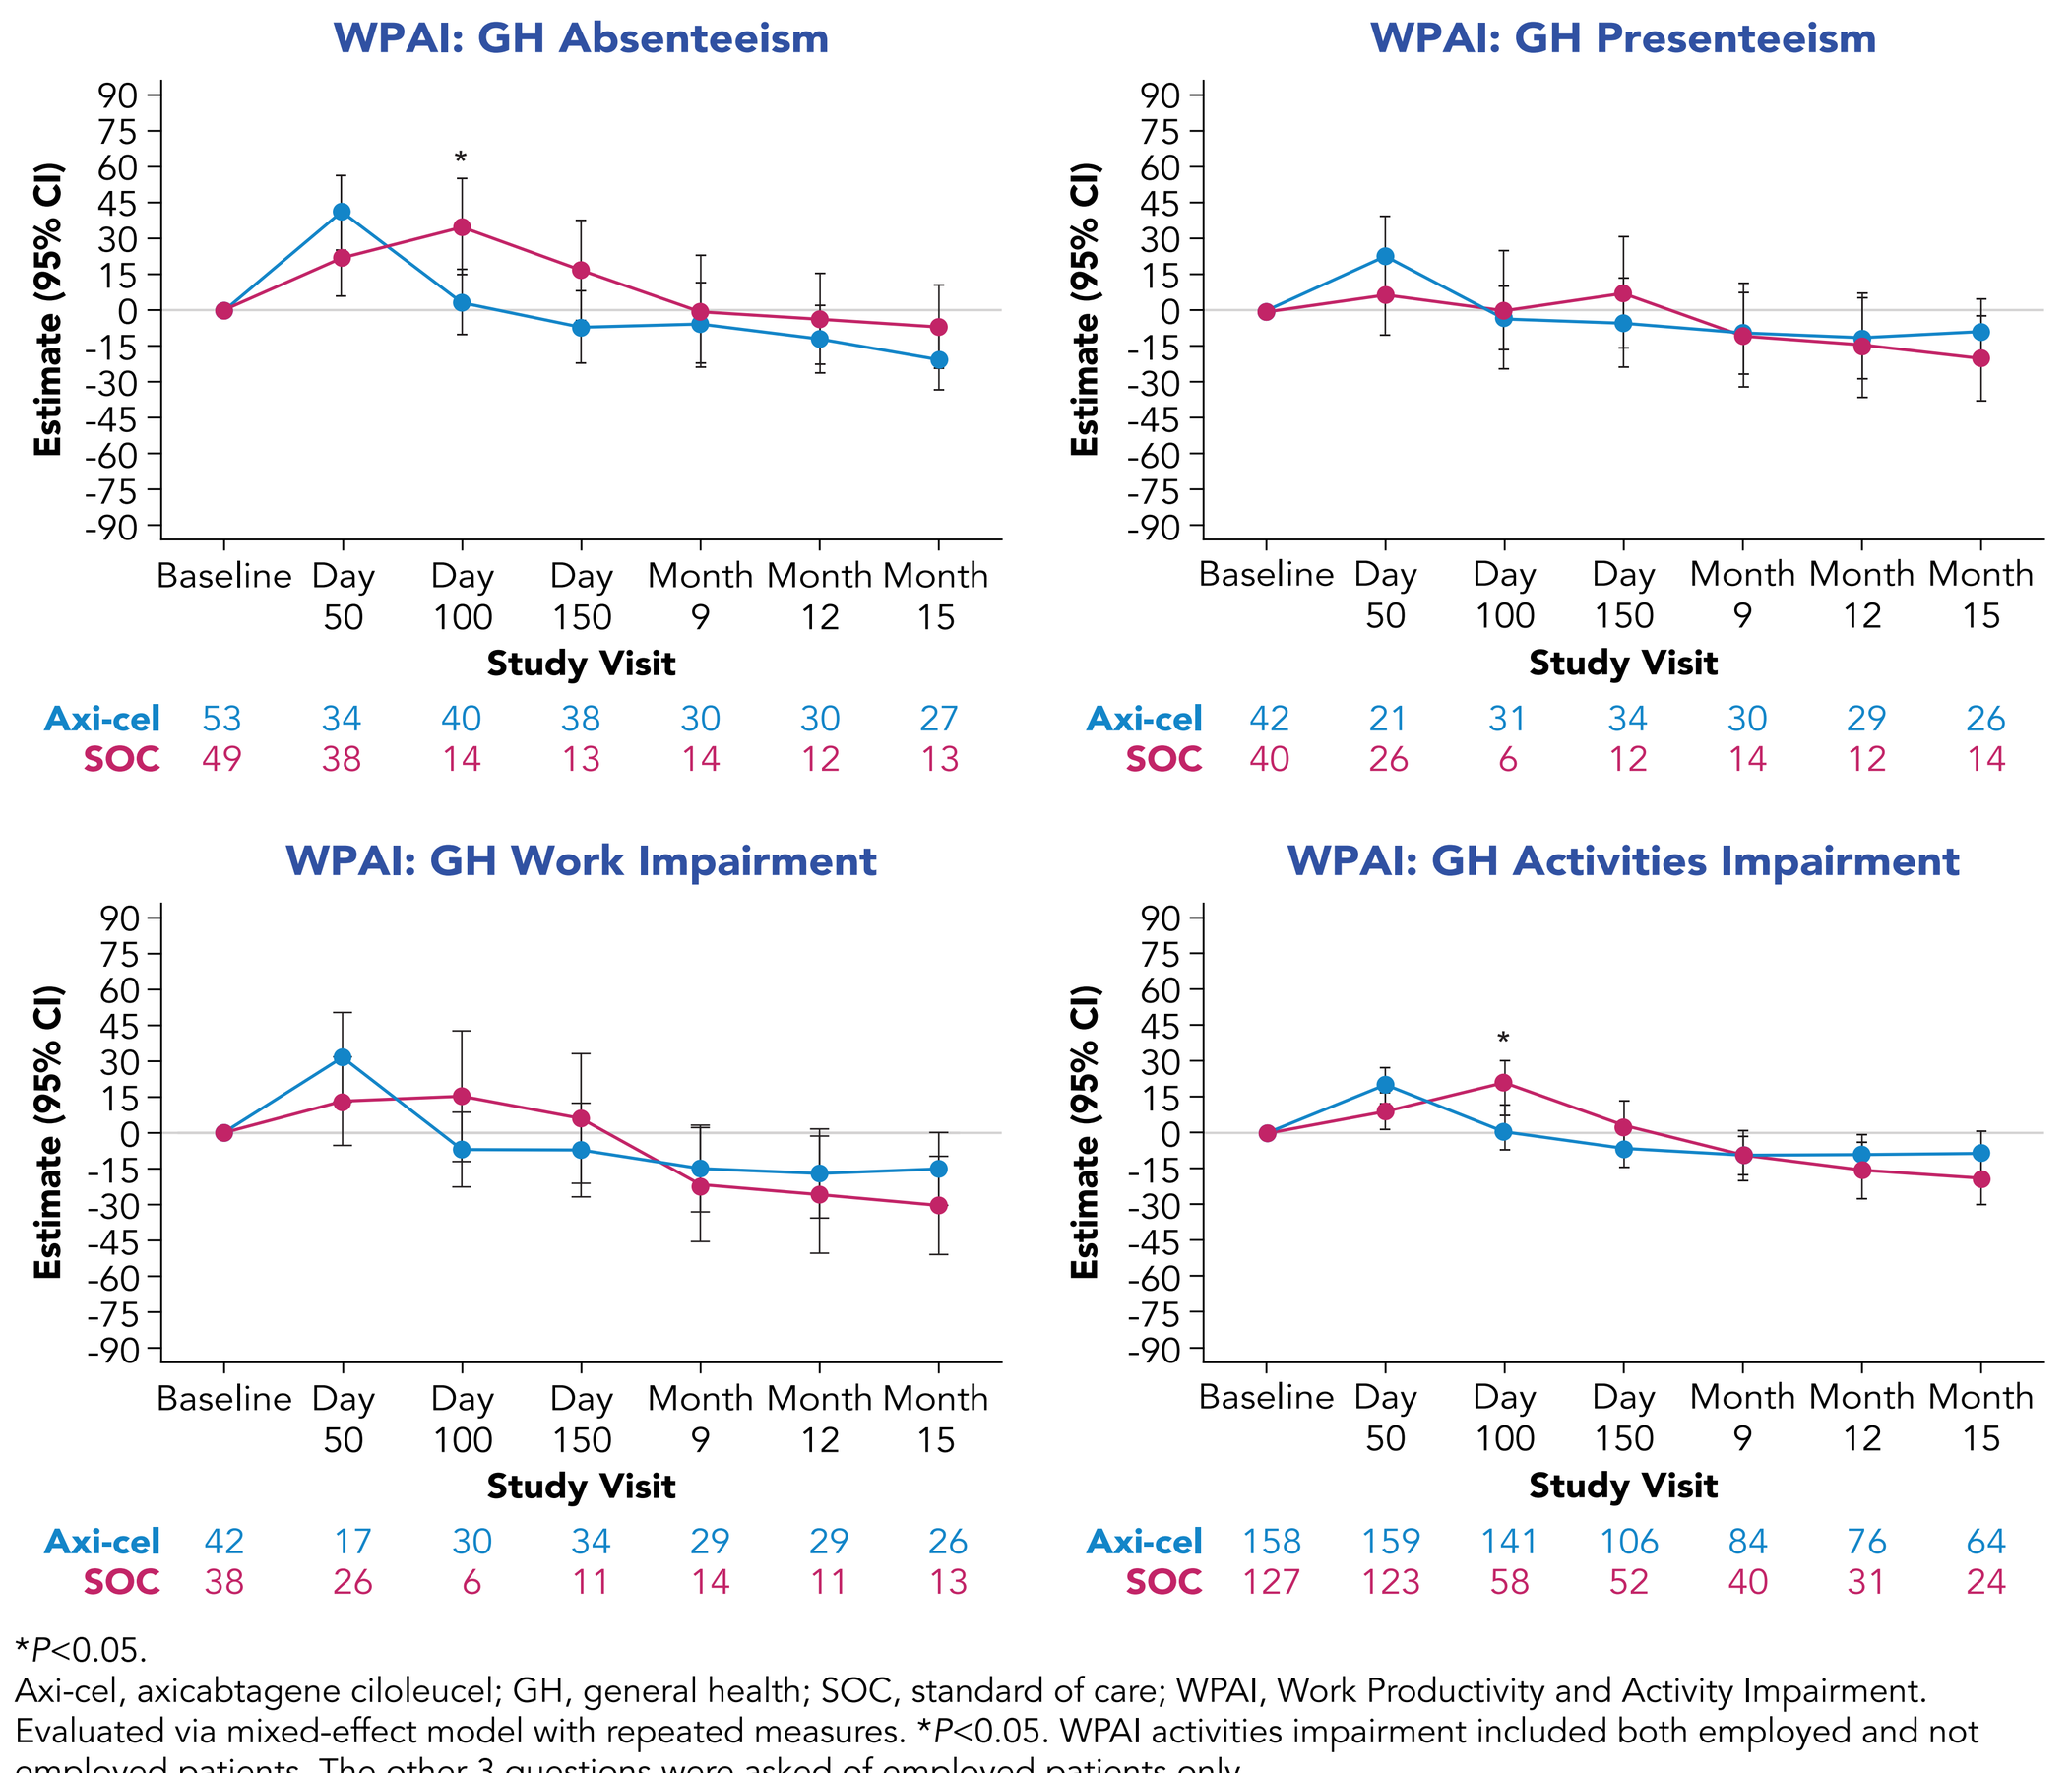
WPAI: GH Absenteeism
Estimate (95% CI)
Baseline
Day 50
Day 100
Day 150
Month 9
Month 12
Month 15
Study Visit
Axi-cel 53 34 40 38 30 30 27
SOC 49 38 14 13 14 12 13
WPAI: GH Presenteeism
Axi-cel 42 21 31 34 30 29 26
SOC 40 26 6 12 14 12 14
WPAI: GH Work Impairment
Axi-cel 42 17 30 34 29 29 26
SOC 38 26 6 11 14 11 13
WPAI: GH Activities Impairment
Axi-cel 158 159 141 106 84 76 64
SOC 127 123 58 52 40 31 24

*P<0.05.
Axi-cel, axicabtagene ciloleucel; GH, general health; SOC, standard of care; WPAI, Work Productivity and Activity Impairment.
Evaluated via mixed-effect model with repeated measures. *P<0.05. WPAI activities impairment included both employed and not employed patients. The other 3 questions were asked of employed patients only.

- Patients treated with axi-cel had statistically significant (P<0.05) lower mean absenteeism and lower mean activities impairment at Day 100 (**Figure 5**)

CONCLUSIONS

- ZUMA-7, the first randomized, global, multicenter Phase 3 study of axi-cel versus SOC in second-line R/R LBCL, demonstrates that treatment with axi-cel results in clinically meaningful improvement in QoL over SOC at Day 100 as measured by multiple validated PRO instruments
 - Score comparisons at later time points warrant cautious interpretation because attrition due to disease progression, new lymphoma therapy, or death was disproportionately higher on the SOC arm and may select patients with the best outcomes
- The data also suggest faster recovery to pretreatment QoL with axi-cel compared with SOC
- The superior clinical outcomes and patient experience with axi-cel over SOC should help inform treatment choices in second-line R/R LBCL

REFERENCES

1. Lin V, et al. *J Clin Oncol.* 2020;38:e20070.
2. Locke FL, et al. *N Engl J Med.* 2022;386:640-654.
3. Jacobson CA, et al. ASH 2021. Abstract #1764.
4. Swerdlow SH, et al. *Blood.* 2016;127:2375-2390.
5. Cheson BD, et al. *J Clin Oncol.* 2014;32:3059-3068.
6. Aaronson NK, et al. *J Natl Cancer Inst.* 1993;85:365-376.
7. Fayers P, et al. The EORTC QLQ-C30 Scoring Manual (3rd Edition). 2001.
8. Herdman M, et al. *Qual Life Res.* 2011;20:1727-1736.
9. Pickard AS, et al. *Value Health.* 2019;22:931-941.
10. Reilly MC, et al. *Pharmacoeconomics.* 1993;4:353-365.
11. Maringwa JT, et al. *Support Care Cancer.* 2011;19:1753-1760.
12. Pickard AS, et al. *Health Qual Life Outcomes.* 2007;5:70.
13. Thieblemont C, et al. *Br J Haematol.* 2020;189:84-96.
14. Elsawy M, et al. ASH 2021. Abstract #430.

ACKNOWLEDGMENTS

The authors would like to thank and acknowledge all ZUMA-7 investigators and contributing Kite members as well as ZUMA-7 patients, families, friends, and caregivers for their contributions to this research

This study was funded by Kite

Medical writing support was provided by Hildebrando Neto, PhD, and Skye Geherin, PhD, of Nexus Global Group Science LLC, funded by Kite

These data were previously presented at the 2021 Annual Meeting of the American Society of Hematology[14]

DISCLOSURES

ME: honoraria from and consulting or advisory role for Bristol Myers Squibb, Kite, Janssen, Novartis, and Pfizer.

Full author disclosures are available at the following Quick Response (QR) code:

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from the author of this poster.

2022 Annual Meeting of the European Society for Blood and Marrow Transplantation