Abstract
P1197

Axicabtagene Ciloleucel in Combination With Rituximab for the Treatment of Refractory Large B-Cell Lymphoma: Outcomes of the Phase 2 ZUMA-14 Study

Paolo Strati, MD[1]; Lori Leslie, MD[2]; Parveen Shiraz, MD[3]; Elizabeth Budde, MD, PhD[4]; Olalekan O. Oluwole, MBBS, MPH[5]; Matthew Ulrickson, MD[6]; Aravind Ramakrishnan, MD[7]; Jennifer Sun, MS[8]; Rhine Shen, PhD[8]; Justyna Kanska, PhD[8]; Peter McCroskery, MD, MS[8]; Jinghui Dong, PhD[8]; Marco Schupp, MD[8]; Hairong Xu, MD, PhD[8]; and Krish Patel, MD[9]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]John Theurer Cancer Center, Hackensack, NJ, USA; [3]Stanford University, Stanford, CA, USA; [4]City of Hope National Medical Center, Duarte, CA, USA; [5]Vanderbilt University Cancer Center, Nashville, TN, USA; [6]Banner MD Anderson Cancer Center, Gilbert, AZ, USA; [7]Texas Transplant Physician Group, Austin, TX, USA; [8]Kite, a Gilead Company, Santa Monica, CA, USA; and [9]Swedish Cancer Institute, Seattle, WA, USA

BACKGROUND

- Axicabtagene ciloleucel (axi-cel) is an autologous anti-CD19 chimeric antigen receptor (CAR) T-cell therapy (**Figure 1**) approved for the treatment of adult patients with relapsed/refractory (R/R) large B-cell lymphoma (LBCL) after ≥2 lines of systemic therapy and most recently, in the United States, for R/R LBCL within 12 months of first-line chemoimmunotherapy[1]
 - With a median follow-up of 27.1 months in ZUMA-1, the objective response rate (ORR) with axi-cel was 83% (58% complete response [CR] rate) in patients with refractory LBCL[2]
 - Axi-cel has been a successful treatment strategy for many patients; however, approximately 60% of patients have no response or relapse within ~2 years after treatment,[2] highlighting the need for more therapeutic strategies
- In preclinical studies rituximab augmented CD19 CAR T-cell function, increasing tumor reduction and survival in murine models via synergistic targeting with CAR T cells[3]
 - T-cell lines transduced with anti-CD19 CAR elicited strong cytotoxicity against B-cell non-Hodgkin lymphoma cell lines and lymphoma cells isolated from patients[3]
 - The addition of rituximab had a synergistic effect, without causing hematological side effects *in vivo*[3]
- ZUMA-14 is investigating the efficacy and safety of axi-cel plus rituximab in patients with refractory LBCL

Figure 1. Structure of Axi-Cel

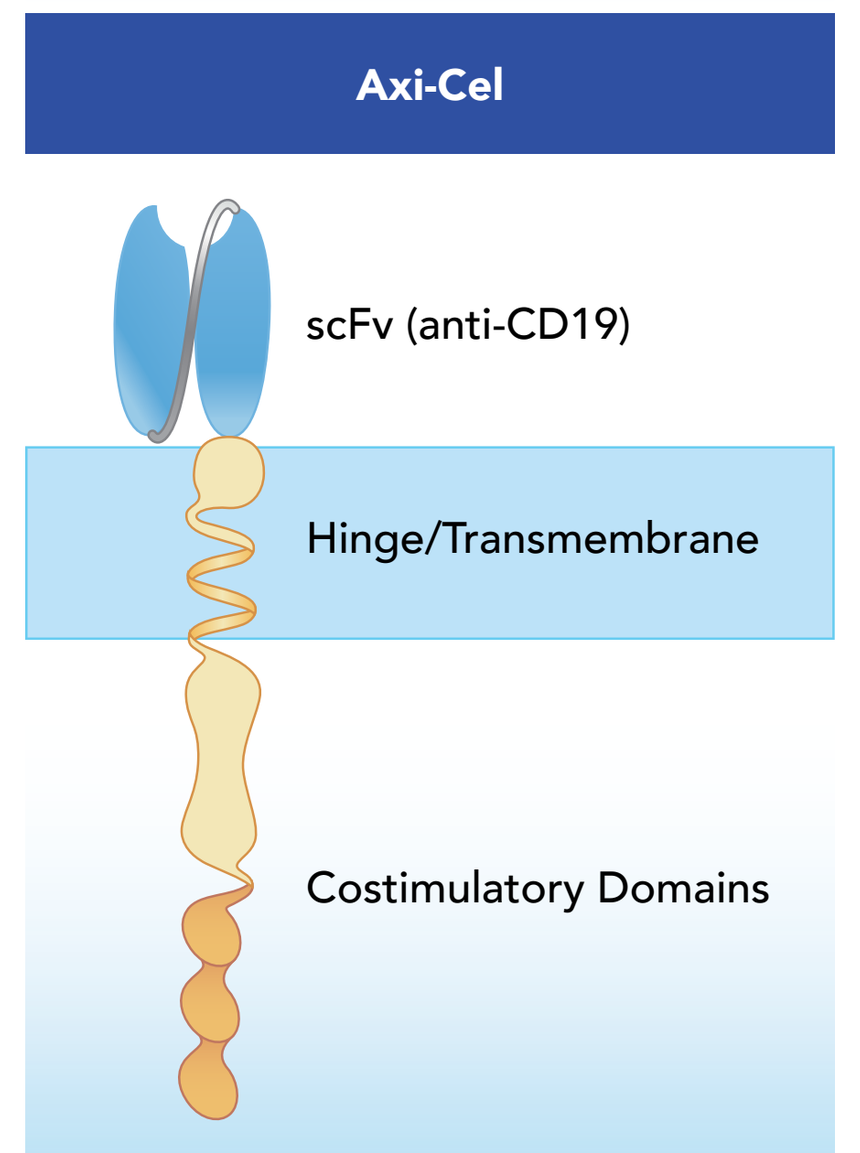
Axi-Cel
scFv (anti-CD19)
Hinge/Transmembrane
Costimulatory Domains

Axi-cel, axicabtagene ciloleucel; scFv, single-chain variable fragment.

OBJECTIVE

- To report outcomes of ZUMA-14, a Phase 2, multicenter study of axi-cel in combination with rituximab in patients with refractory LBCL after ≥2 lines of systemic therapy

METHODS

Figure 2. ZUMA-14 Treatment Schema

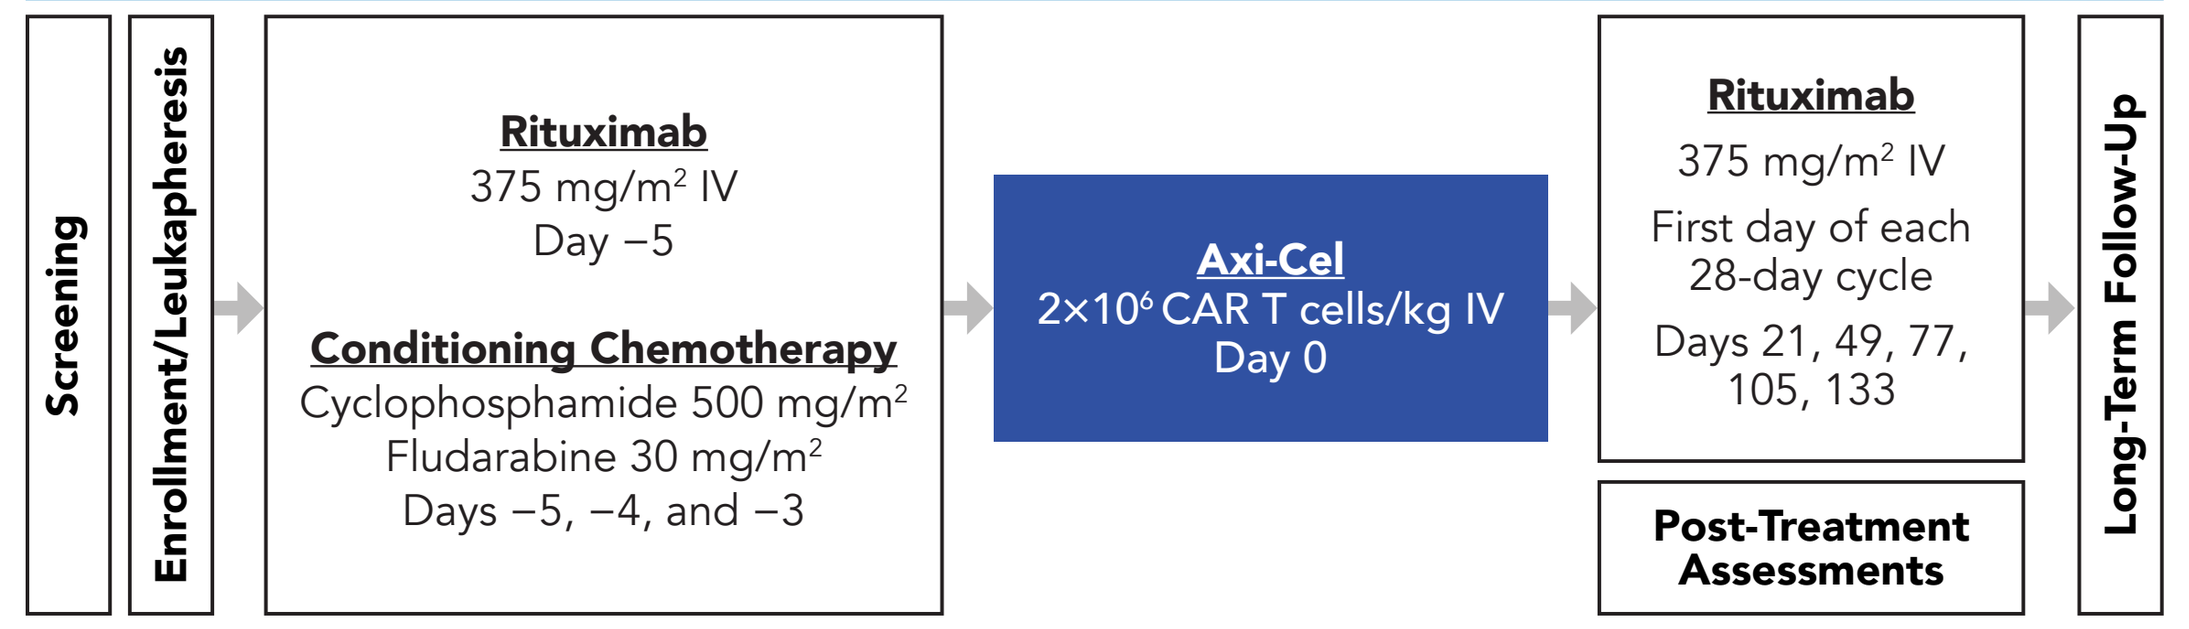
Screening
Enrollment/Leukapheresis
Rituximab
375 mg/m2 IV
Day −5
Conditioning Chemotherapy
Cyclophosphamide 500 mg/m2
Fludarabine 30 mg/m2
Days −5, −4, and −3
Axi-Cel
2×10^6 CAR T cells/kg IV
Day 0
Rituximab
375 mg/m2 IV
First day of each 28-day cycle
Days 21, 49, 77, 105, 133
Post-Treatment Assessments
Long-Term Follow-Up

Key Eligibility Criteria
- Histologically proven LBCL
- Prior anti-CD20 monoclonal antibody and anthracycline
- Chemotherapy-refractory disease (primary refractory, no response to ≥2L therapy, or refractory post-ASCT)

Primary Endpoint
- Investigator-assessed CR rate

Key Additional Endpoints
- ORR, DOR, PFS, OS, safety, and biomarker assessments

2L, second line; ASCT, autologous stem cell transplantation; axi-cel, axicabtagene ciloleucel; CAR, chimeric antigen receptor; CR, complete response; DOR, duration of response; IV, intravenous; LBCL, large B-cell lymphoma; ORR, objective response rate; OS, overall survival; PFS, progression-free survival.

RESULTS

Figure 3. Patient Disposition

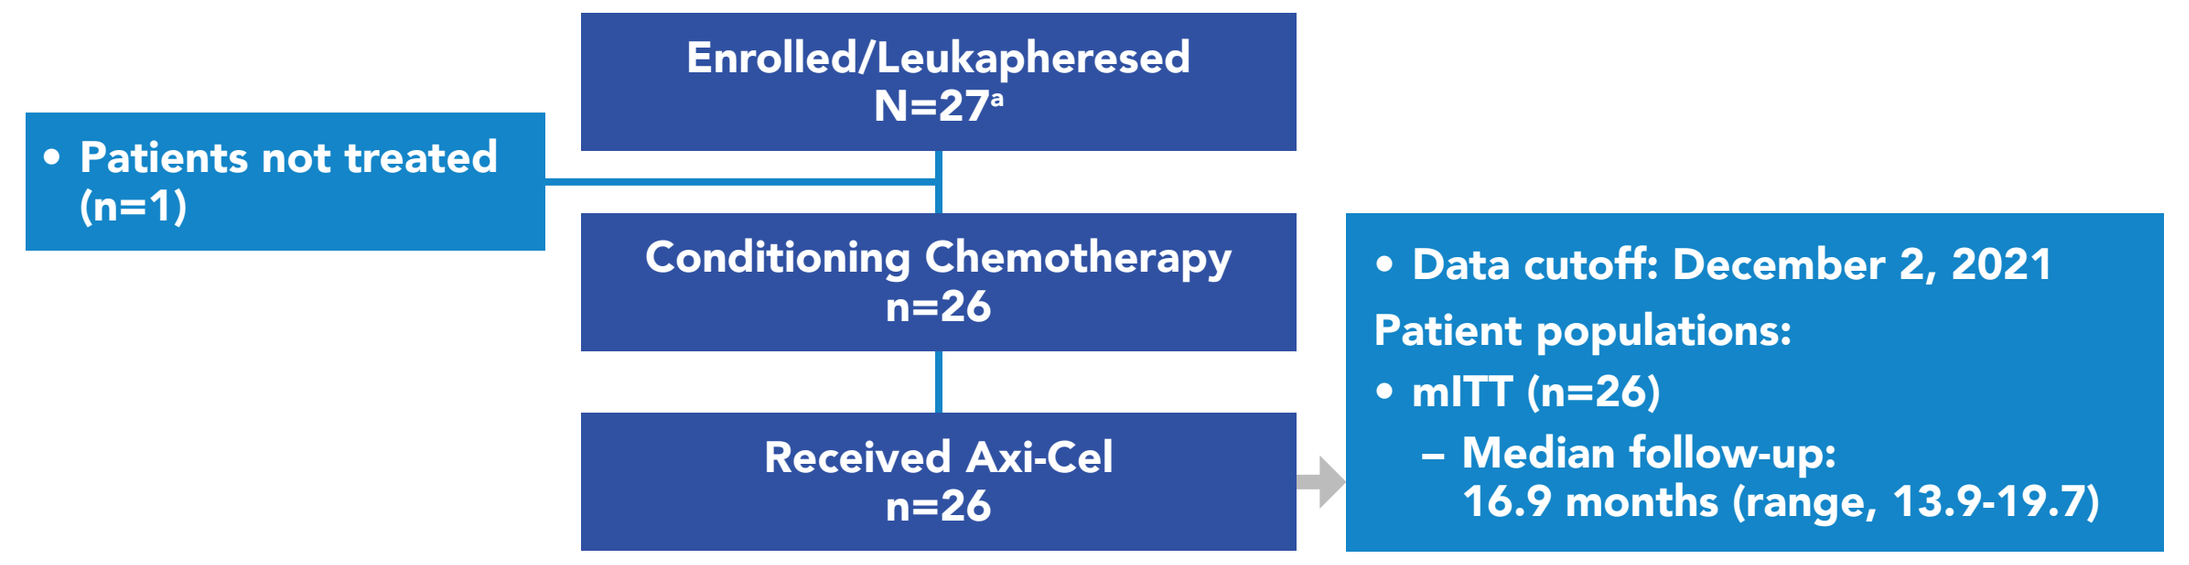
Enrolled/Leukapheresed
N=27a
Patients not treated (n=1)
Conditioning Chemotherapy
n=26
Received Axi-Cel
n=26
Data cutoff: December 2, 2021
Patient populations:
mITT (n=26)
– Median follow-up: 16.9 months (range, 13.9-19.7)

[a] Patients were considered enrolled in the study once the leukapheresis procedure was initiated.
Axi-cel, axicabtagene ciloleucel; mITT, modified intent-to-treat.

- Of the 26 patients treated with rituximab after axi-cel, 15 patients completed treatment while 11 patients did not, 1 due to an adverse event and 10 due to progressive disease
 - The median number of cycles of rituximab was 6 cycles

RESULTS *(Continued)*

Table 1. ZUMA-14 Baseline Characteristics

Characteristics	Axi-Cel + Rituximab (n=26)
Age, median (range), years ≥65 years	62.5 (38-82) 12 (46)
Male, n (%)	14 (54)
ECOG PS of 1, n (%)	14 (54)
Disease stage, n (%) I/II III/IV	 2 (8)/3 (12) 5 (19)/16 (62)
Number of prior therapies, n (%) 1 2 ≥3	 5 (19) 19 (73) 2 (8)
Prior transplant, n (%)	2 (8)
Extranodal disease, n (%)	16 (62)
Elevated LDH,[a] n (%)	10 (38)
aaIPI, n (%) 0/1/2	 4 (15)/13 (50)/9 (35)
Primary refractory disease, n (%)	5 (19)

[a] LDH>ULN per local laboratory reference range.
aaIPI, age-adjusted International Prognostic Index; axi-cel, axicabtagene ciloleucel; ECOG PS, Eastern Cooperative Oncology Group performance status; LDH, lactate dehydrogenase; ULN upper limit of normal.

Table 2. Safety Summary

Parameter, n (%)	Axi-Cel + Rituximab (n=26)
Serious AEs	15 (58)
Worst Grade ≥3 cytopenias	22 (85)
Worst Grade ≥3 infections	5 (19)
Deaths Progressive disease	6 (23) 6 (23)

AE, adverse event; axi-cel, axicabtagene ciloleucel.

Table 3. Treatment-Emergent Adverse Events

	Axi-Cel + Rituximab (n=26)	
AE, n (%)	**Any Grade**	**Grade ≥3**
Any AE[a,b]	**26 (100)**	**24 (92)**
Pyrexia	25 (96)	0 (0)
Neutrophil count decreased	17 (65)	16 (62)
Hypotension	16 (62)	1 (4)
Nausea	16 (62)	0 (0)
Anemia	13 (50)	12 (46)
Headache	13 (50)	0 (0)
Confusional state	10 (38)	2 (8)
Decreased appetite	10 (38)	1 (4)
Fatigue	10 (38)	0 (0)
Diarrhea	8 (31)	1 (4)
Hypokalemia	8 (31)	1 (4)
Tachycardia	8 (31)	1 (4)
Prolonged cytopenias[c]	**14 (54)**	**10 (38)**
Prolonged anemia	5 (19)	4 (15)
Prolonged neutropenia	12 (46)	9 (35)
Prolonged thrombocytopenia	3 (12)	3 (12)

[a] Any grade treatment-emergent AEs that occurred in >30% of patients. [b] AEs were coded using MedDRA version 24.1 and graded per National Cancer Institute CTCAE version 5.0. [c] Events present on or after Day 30 post-infusion.
AE, adverse event; axi-cel, axicabtagene ciloleucel; CTCAE, Common Terminology Criteria for Adverse Events; MedDRA, Medical Dictionary for Regulatory Activities.

- Six patients (23%) died during the study, all due to progressive disease (**Table 2**)
- Most patients (92%) experienced Grade ≥3 adverse events (AEs), and serious AEs were experienced by 58% of patients (**Table 3**)
 - One patient experienced a Grade 1 bacterial infection of furunculosis

Table 4. Cytokine Release Syndrome

Parameter	Axi-Cel + Rituximab (n=26)
Any grade CRS, n (%)[a] Grade ≥3	25 (96) 0
Most common any grade symptoms of CRS, n (%)[b] Pyrexia Hypotension Sinus tachycardia	 25 (100) 10 (40) 3 (12)
AE management for CRS, n (%) Tocilizumab Steroids[c]	 20 (77) 8 (31)
Median time to onset (range), days	4 (1-7)
Median duration of events (range), days	5 (2-15)
Patients with resolved events by data cutoff, n/n (%)	25/25 (100)

[a] CRS was graded per Lee DW, et al. *Blood.* 2014;124:188-195. Individual symptoms of CRS were graded per National Cancer Institute CTCAE version 5.0. [b] Any grade treatment-emergent AEs of interest, CRS, that occurred in >10% of patients. [c] No corticosteroids were used prophylactically.
AE, adverse event; axi-cel, axicabtagene ciloleucel; CRS, cytokine release syndrome; CTCAE, Common Terminology Criteria for Adverse Events.

- No patients experienced Grade ≥3 cytokine release syndrome (CRS; **Table 4**)
- Median time to onset of CRS was 4 days (range, 1-7), with a median duration of 5 days (range, 2-15)

Table 5. Neurologic Events

Parameter	Axi-Cel + Rituximab (n=26)
Any grade NE, n (%)[a] Grade ≥3	16 (62) 4 (15)
Most common any grade symptoms of NE, n (%)[b] Confusional state Tremor Agitation Aphasia Somnolence	 10 (38) 7 (27) 5 (19) 3 (12) 3 (12)
AE management for NE, n (%) Tocilizumab Steroids	 3 (12) 9 (35)
Median time to onset (range), days	6 (2-12)
Median duration of events (range), days	7 (1-39)
Patients with resolved events by data cutoff, n/n (%)	16/16 (100)

[a] AEs were coded using MedDRA version 24.1 and graded per National Cancer Institute CTCAE version 5.0.
[b] Any grade treatment-emergent AEs of interest, NEs, that occurred in >10% of patients.
AE, adverse event; axi-cel, axicabtagene ciloleucel; CTCAE, Common Terminology Criteria for Adverse Events; MedDRA, Medical Dictionary for Regulatory Activities; NE, neurologic event.

- Grade ≥3 neurologic events (NEs) occurred in 4 patients (15%), all were Grade 3 NEs (**Table 5**)
- Median time to onset of NEs was 6 days (range, 2-12), with a median duration of 7 days (range, 1-39)

Figure 4. ORR Was 88% With a CR Rate of 73%

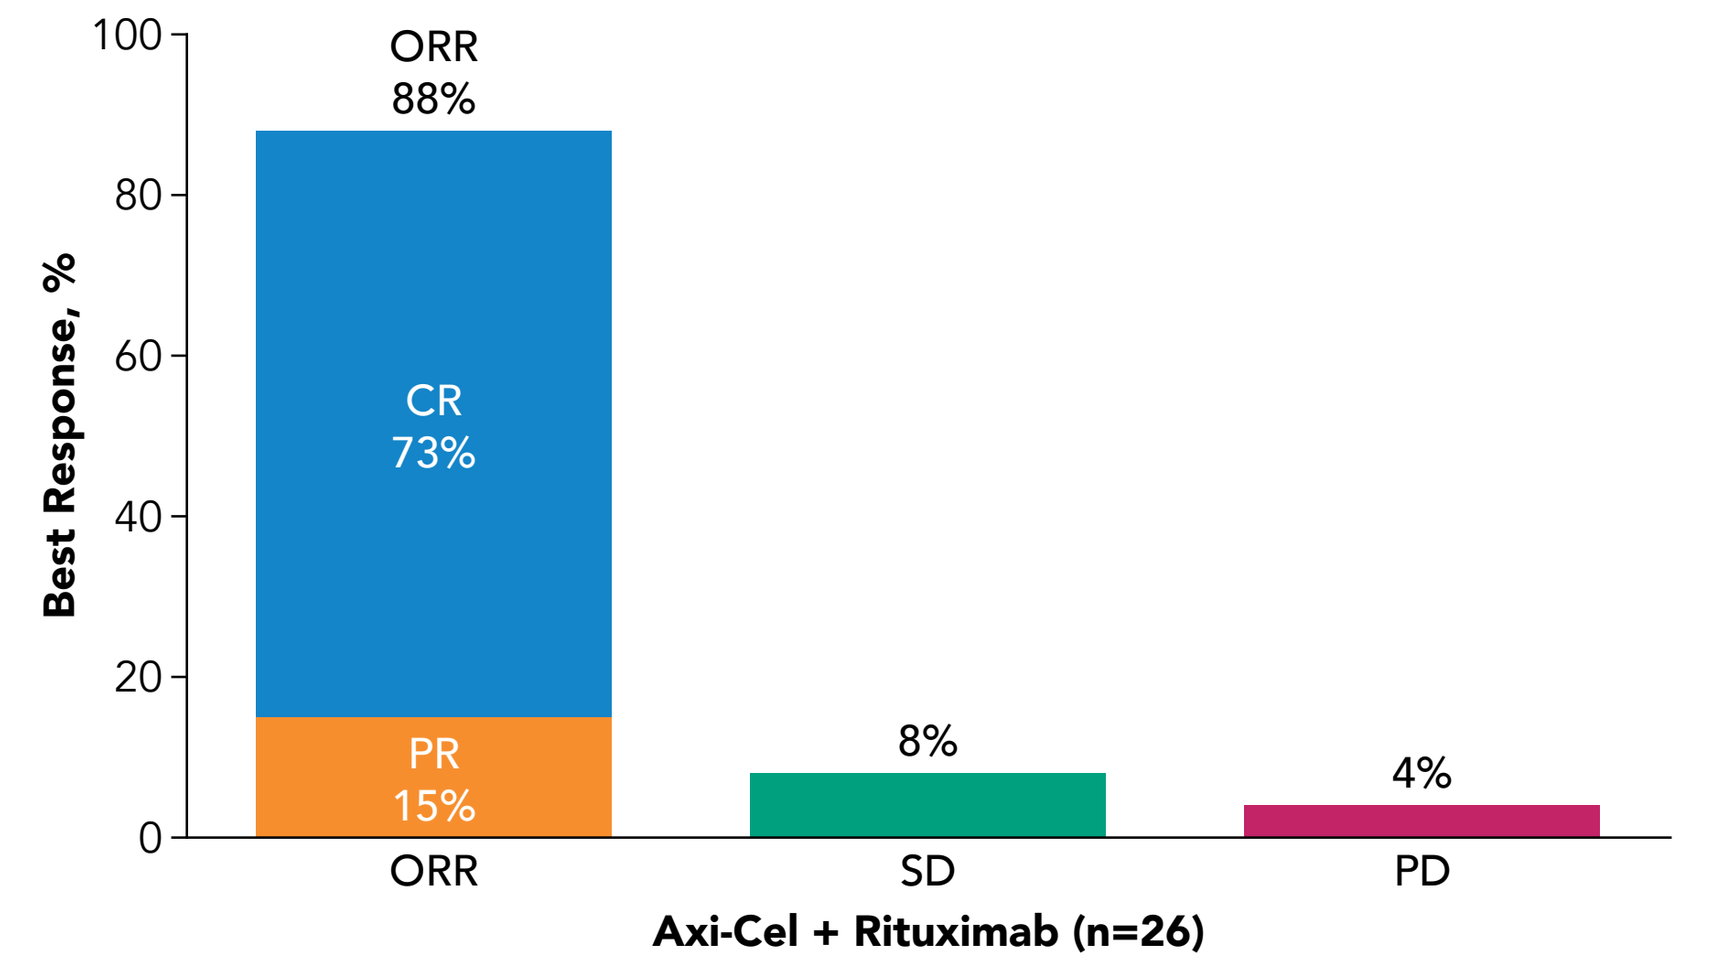
Best Response, %
100
80
60
40
20
0
ORR
88%
CR
73%
PR
15%
8%
4%
ORR
SD
PD
Axi-Cel + Rituximab (n=26)

Axi-cel, axicabtagene ciloleucel; CR, complete response; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease.

- The ORR was 88% (95% CI, 70-98), and the CR rate was 73% (95% CI, 52-88) (**Figure 4**)
- With a median follow-up of 16.9 months, 61% of the patients had ongoing response, all ongoing in CR

Figure 5. Swimmer Plot of Responses, Survival Status, and MRD by Time

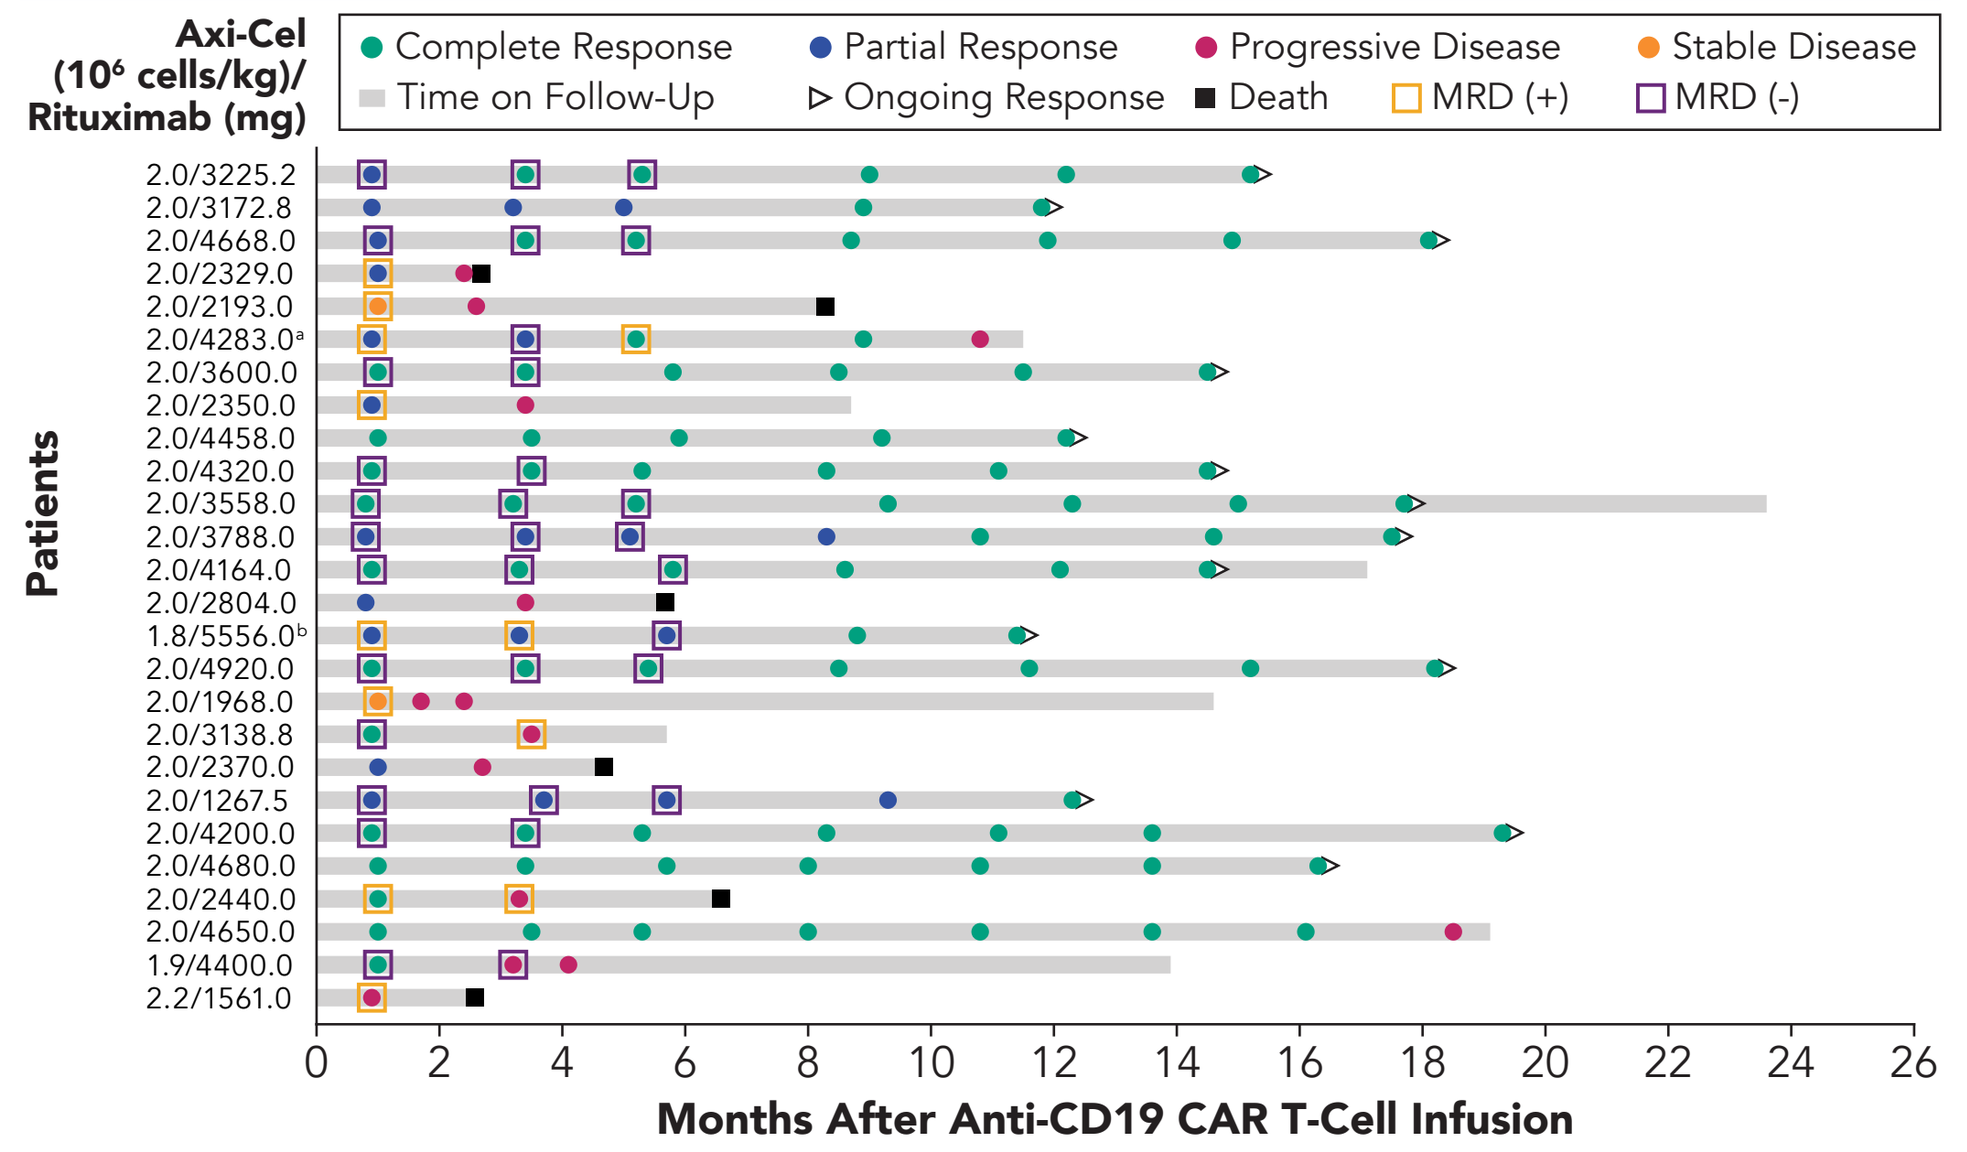
Axi-Cel (10^6 cells/kg)/ Rituximab (mg)
Complete Response
Partial Response
Progressive Disease
Stable Disease
Time on Follow-Up
Ongoing Response
Death
MRD (+)
MRD (-)
Patients
2.0/3225.2
2.0/3172.8
2.0/4668.0
2.0/2329.0
2.0/2193.0
2.0/4283.0a
2.0/3600.0
2.0/2350.0
2.0/4458.0
2.0/4320.0
2.0/3558.0
2.0/3788.0
2.0/4164.0
2.0/2804.0
1.8/5556.0b
2.0/4920.0
2.0/1968.0
2.0/3138.8
2.0/2370.0
2.0/1267.5
2.0/4200.0
2.0/4680.0
2.0/2440.0
2.0/4650.0
1.9/4400.0
2.2/1561.0
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Months After Anti-CD19 CAR T-Cell Infusion

[a] One partial response patient who converted to a complete response at Month 5 and later progressed at Month 11 had detectable MRD at Month 5.
[b] MRD negativity at Month 6 in 1 partial response patient predicted a complete response conversion at Month 9.
Axi-cel, axicabtagene ciloleucel; CAR, chimeric antigen receptor; MRD, minimal residual disease.

- Seven patients converted from a partial response/stable disease to CR ≥3 months post–axi-cel infusion

Figure 6. Duration of Response, Progression-Free Survival, and Overall Survival

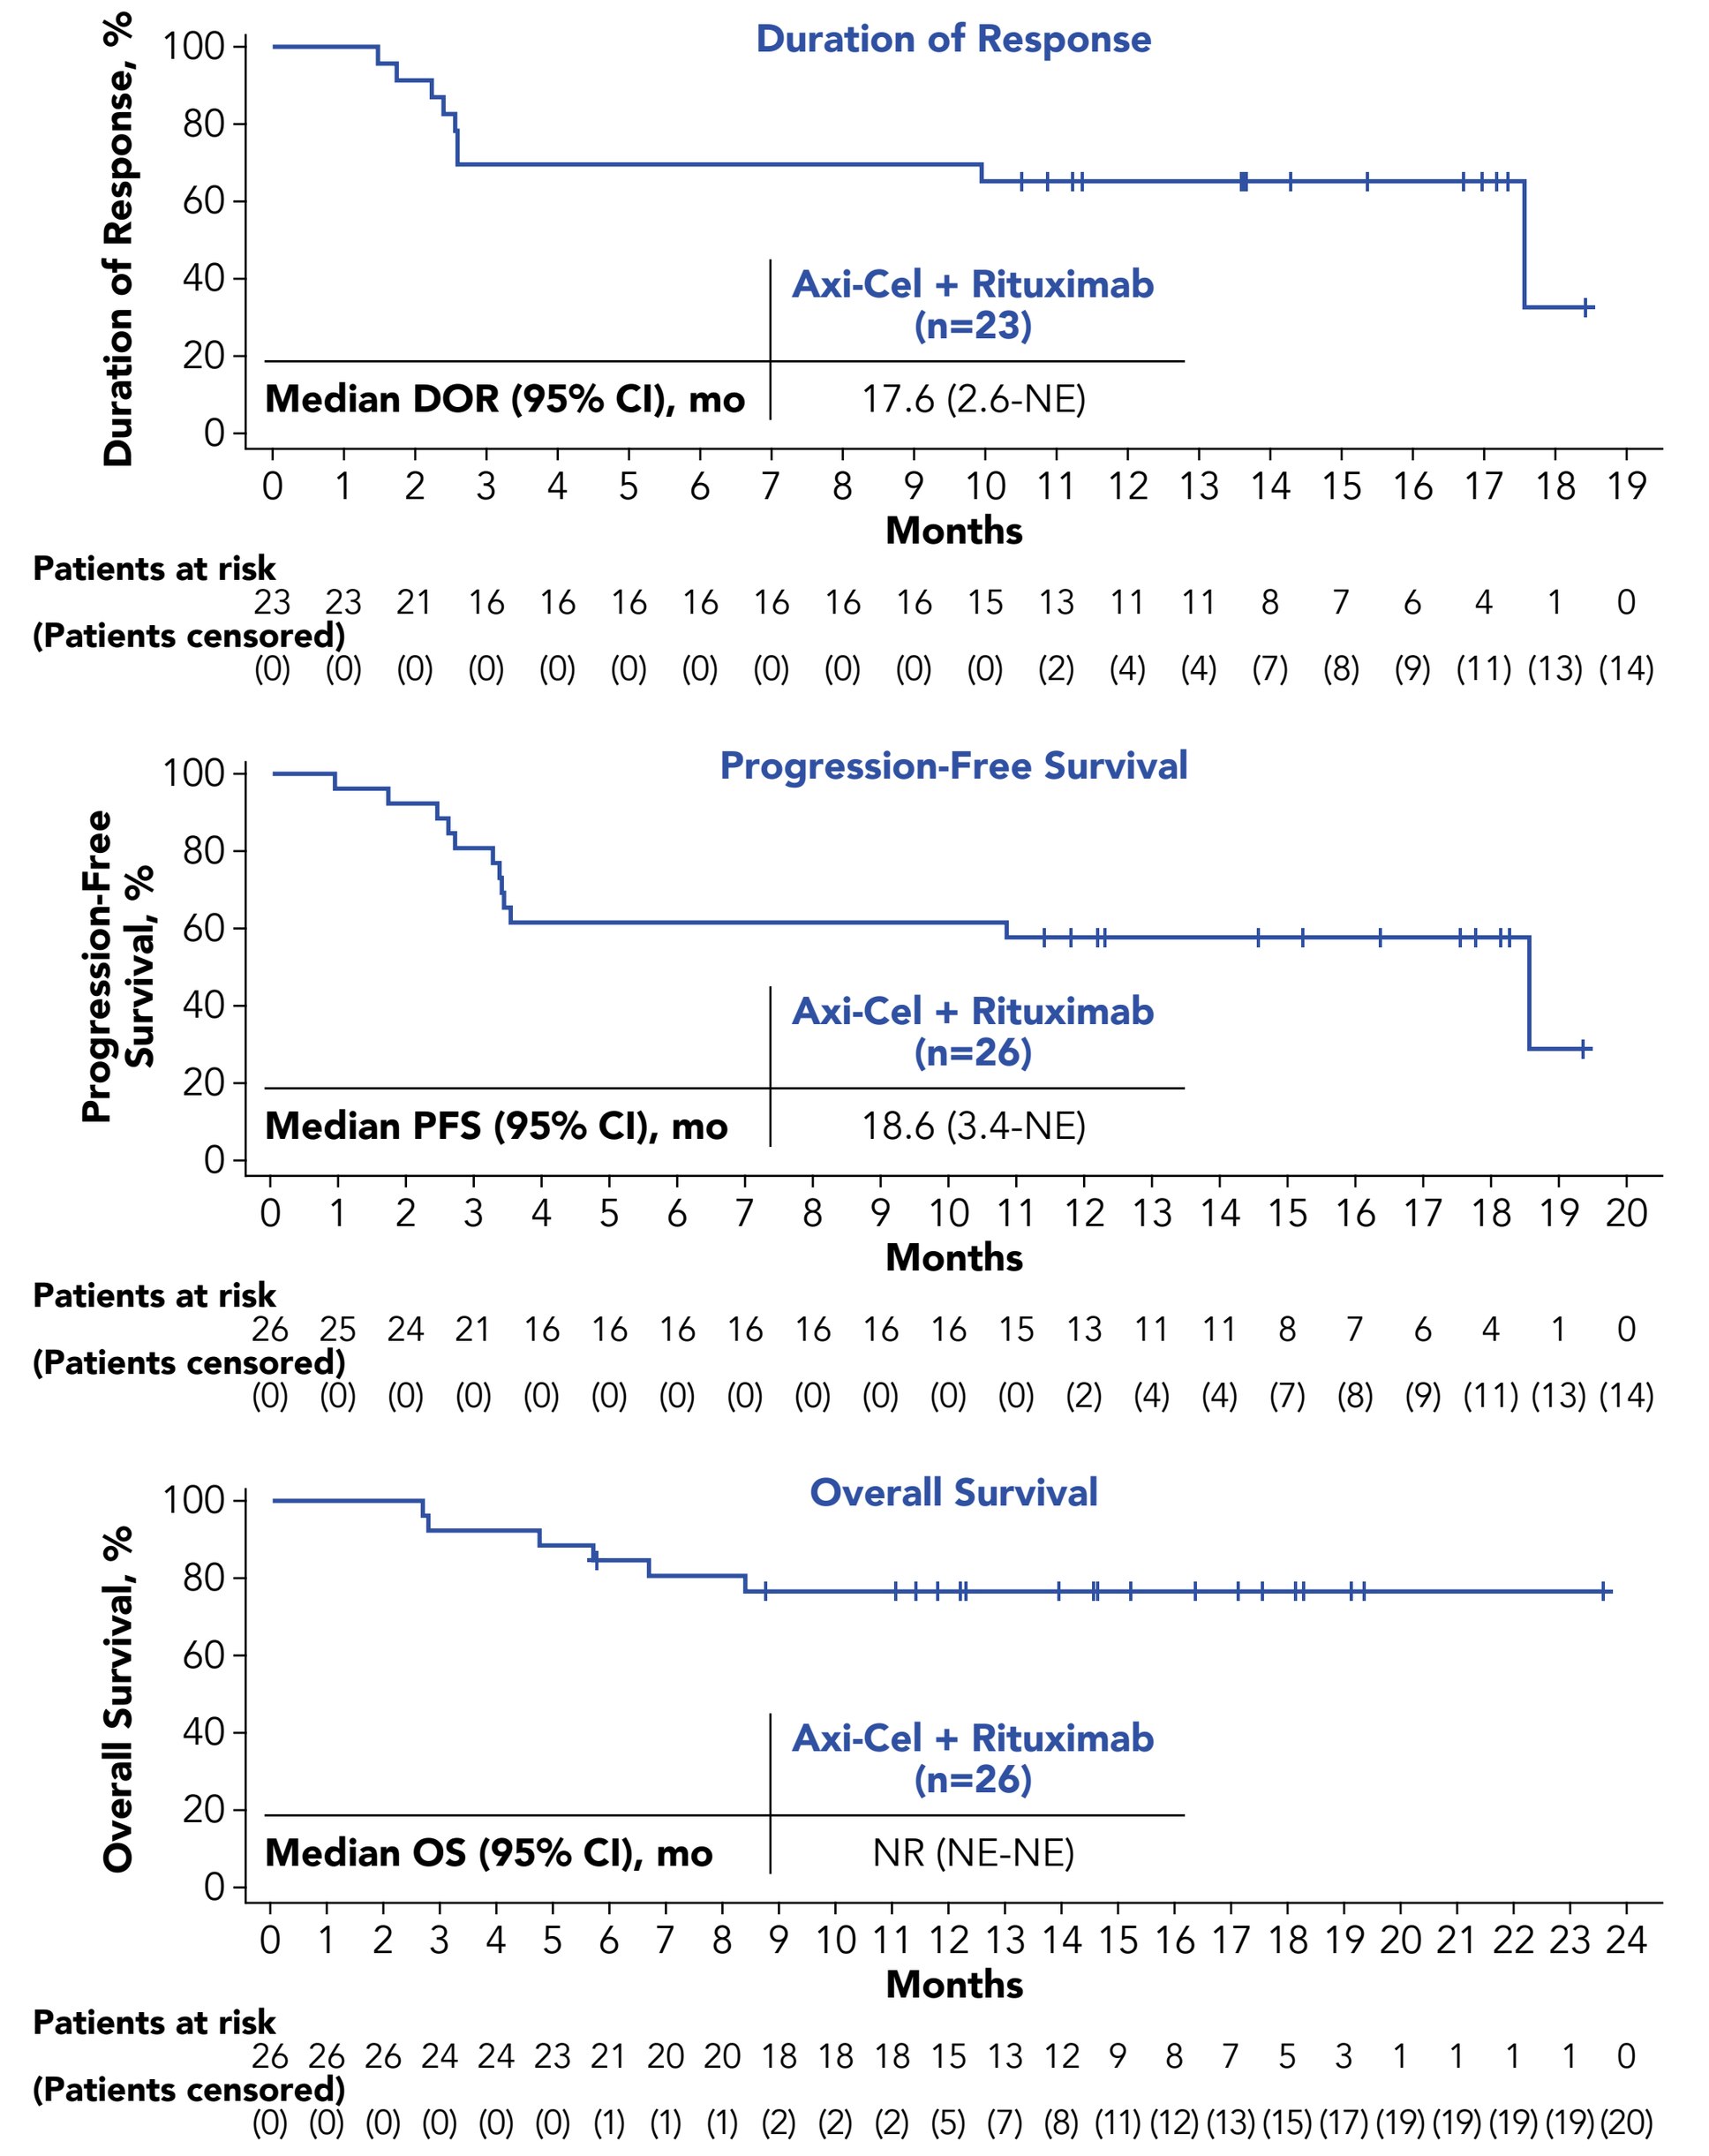
Duration of Response
Duration of Response, %
Axi-Cel + Rituximab (n=23)
Median DOR (95% CI), mo
17.6 (2.6-NE)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19
Months
Patients at risk
23 23 21 16 16 16 16 16 16 16 15 13 11 11 8 7 6 4 1 0
(Patients censored)
(0) (0) (0) (0) (0) (0) (0) (0) (0) (0) (0) (2) (4) (4) (7) (8) (9) (11) (13) (14)
Progression-Free Survival
Progression-Free Survival, %
Axi-Cel + Rituximab (n=26)
Median PFS (95% CI), mo
18.6 (3.4-NE)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20
Months
Patients at risk
26 25 24 21 16 16 16 16 16 16 16 15 13 11 11 8 7 6 4 1 0
(Patients censored)
(0) (0) (0) (0) (0) (0) (0) (0) (0) (0) (0) (0) (2) (4) (4) (7) (8) (9) (11) (13) (14)
Overall Survival
Overall Survival, %
Axi-Cel + Rituximab (n=26)
Median OS (95% CI), mo
NR (NE-NE)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
Months
Patients at risk
26 26 26 24 24 23 21 20 20 18 18 18 15 13 12 9 8 7 5 3 1 1 1 1 0
(Patients censored)
(0) (0) (0) (0) (0) (0) (1) (1) (1) (2) (2) (2) (5) (7) (8) (11) (12) (13) (15) (17) (19) (19) (19) (19) (20)

Axi-cel, axicabtagene ciloleucel; DOR, duration of response; mo, month; NE, not evaluable; NR, not reached; OS overall survival; PFS, progression-free survival.

- The estimated duration of response rate at 12 months was 65% (95% CI, 42-81)
- The estimated progression-free survival (PFS) rate at 12 months was 58% (95% CI, 37-74)
- The estimated 12-month overall survival rate was 77% (95% CI, 55-89), and 6 patients (23%) died of progressive disease

Figure 7. Peak CAR T-Cell and Rituximab Levels in Blood

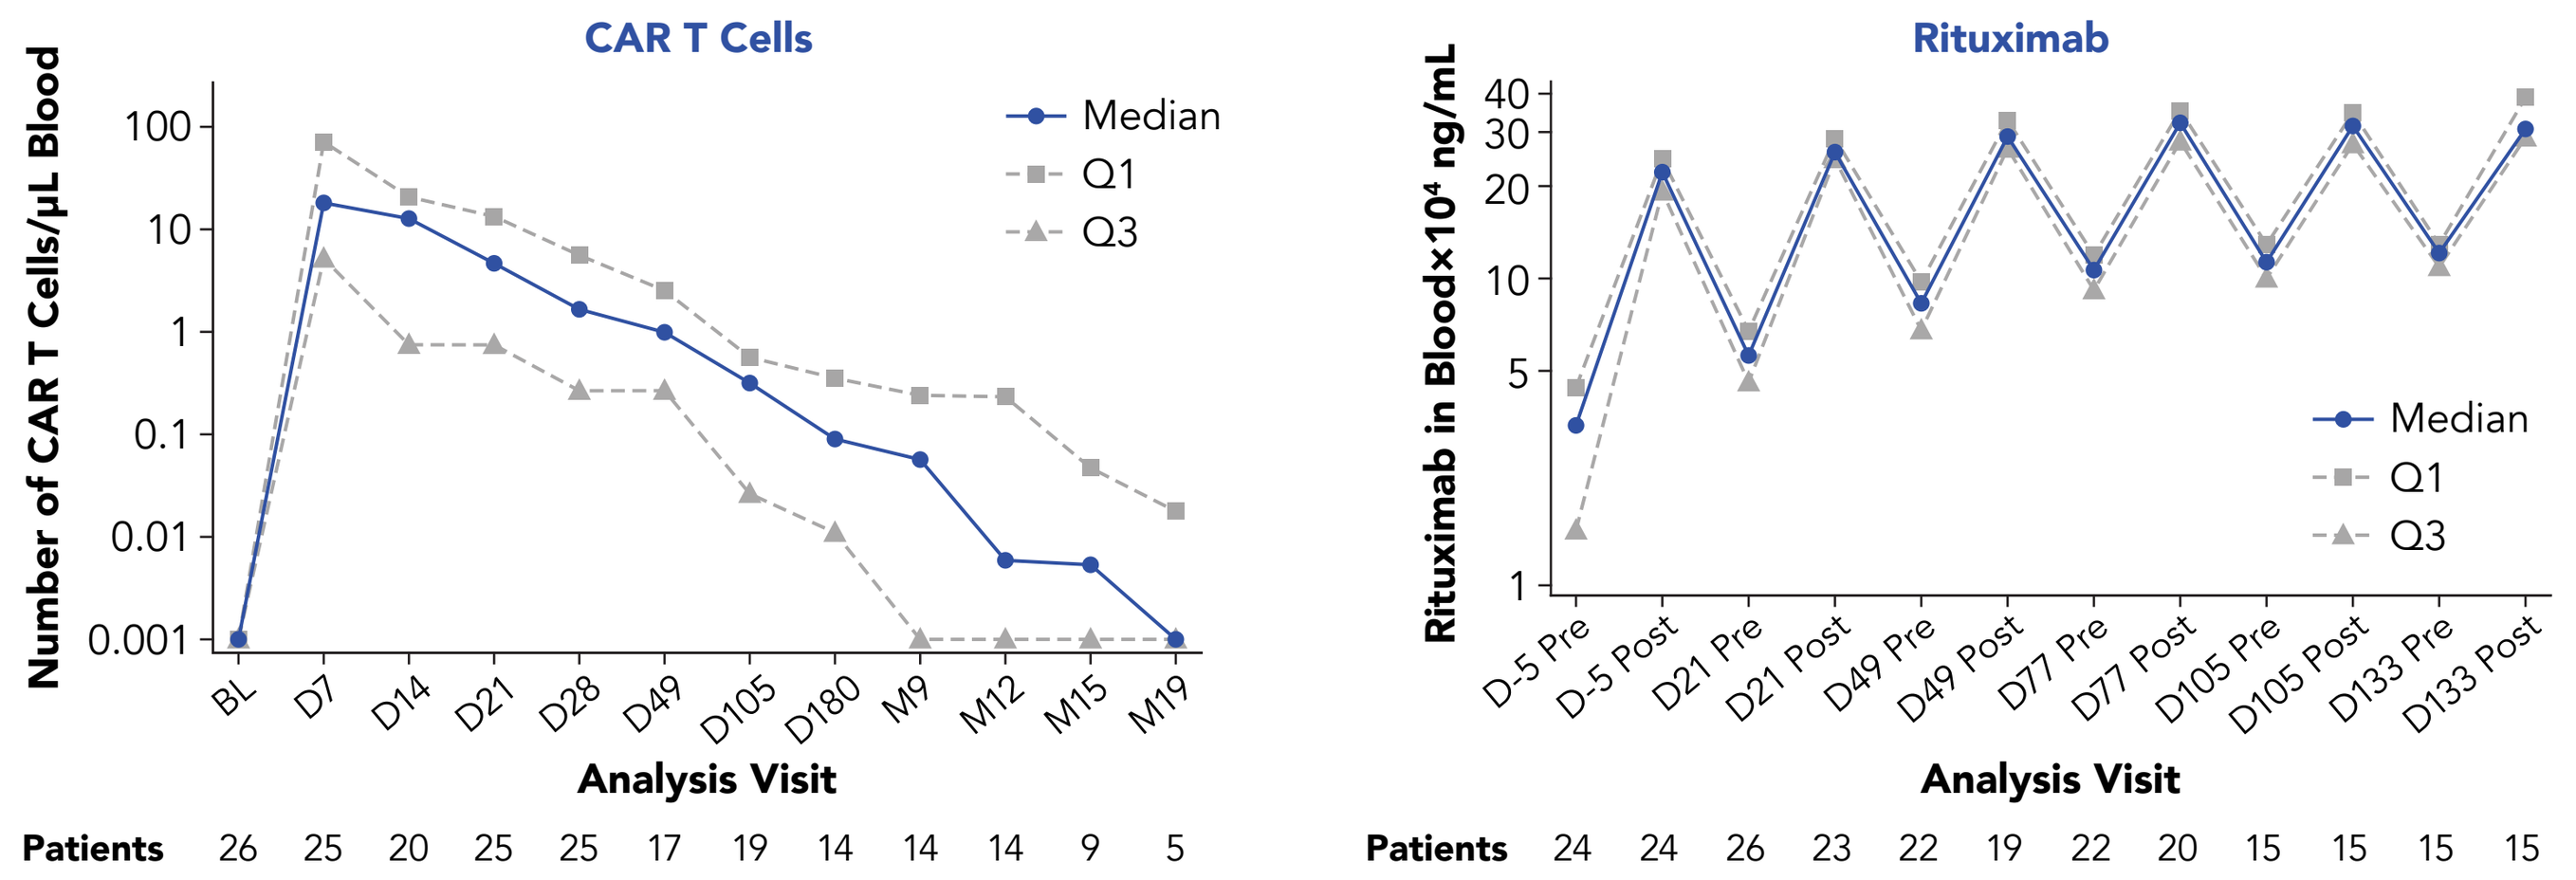
CAR T Cells
Number of CAR T Cells/μL Blood
Median
Q1
Q3
BL D7 D14 D21 D28 D49 D105 D180 M9 M12 M15 M19
Analysis Visit
Patients 26 25 20 25 25 17 19 14 14 14 9 5
Rituximab
Rituximab in Blood×10^4 ng/mL
D-5 Pre D-5 Post D21 Pre D21 Post D49 Pre D49 Post D77 Pre D77 Post D105 Pre D105 Post D133 Pre D133 Post
Analysis Visit
Patients 24 24 26 23 22 19 22 20 15 15 15 15

Parameter, median (range)	ZUMA-14 (n=26)[a]	ZUMA-1 C1+2 (n=98)[a]
Peak, cells/μL	40.3 (13.6-95.9)	38.3 (14.7-83.0)
AUC_{0-28}, cells/μL×days	376.8 (136.3-895.3)	453.4 (148.7-920.3)
Time to peak, days	8 (8-8)	8 (8-15)

[a] Includes patients with evaluable data for CAR T-cell therapy.
AUC_{0-28}, area under curve (day 0 to day 28); BL, baseline; C, cohort; CAR, chimeric antigen receptor; D, day; M, month; Q, quartile.

- Peak CAR T-cell levels in ZUMA-14 were comparable to ZUMA-1 (**Figure 7**)
- Peak and area under the curve rituximab levels were elevated in responders versus non-responders (data not shown)

Figure 8. Key Cytokines Are Induced Following Axi-Cel and Rituximab Infusion in Responders Relative to Non-Responders

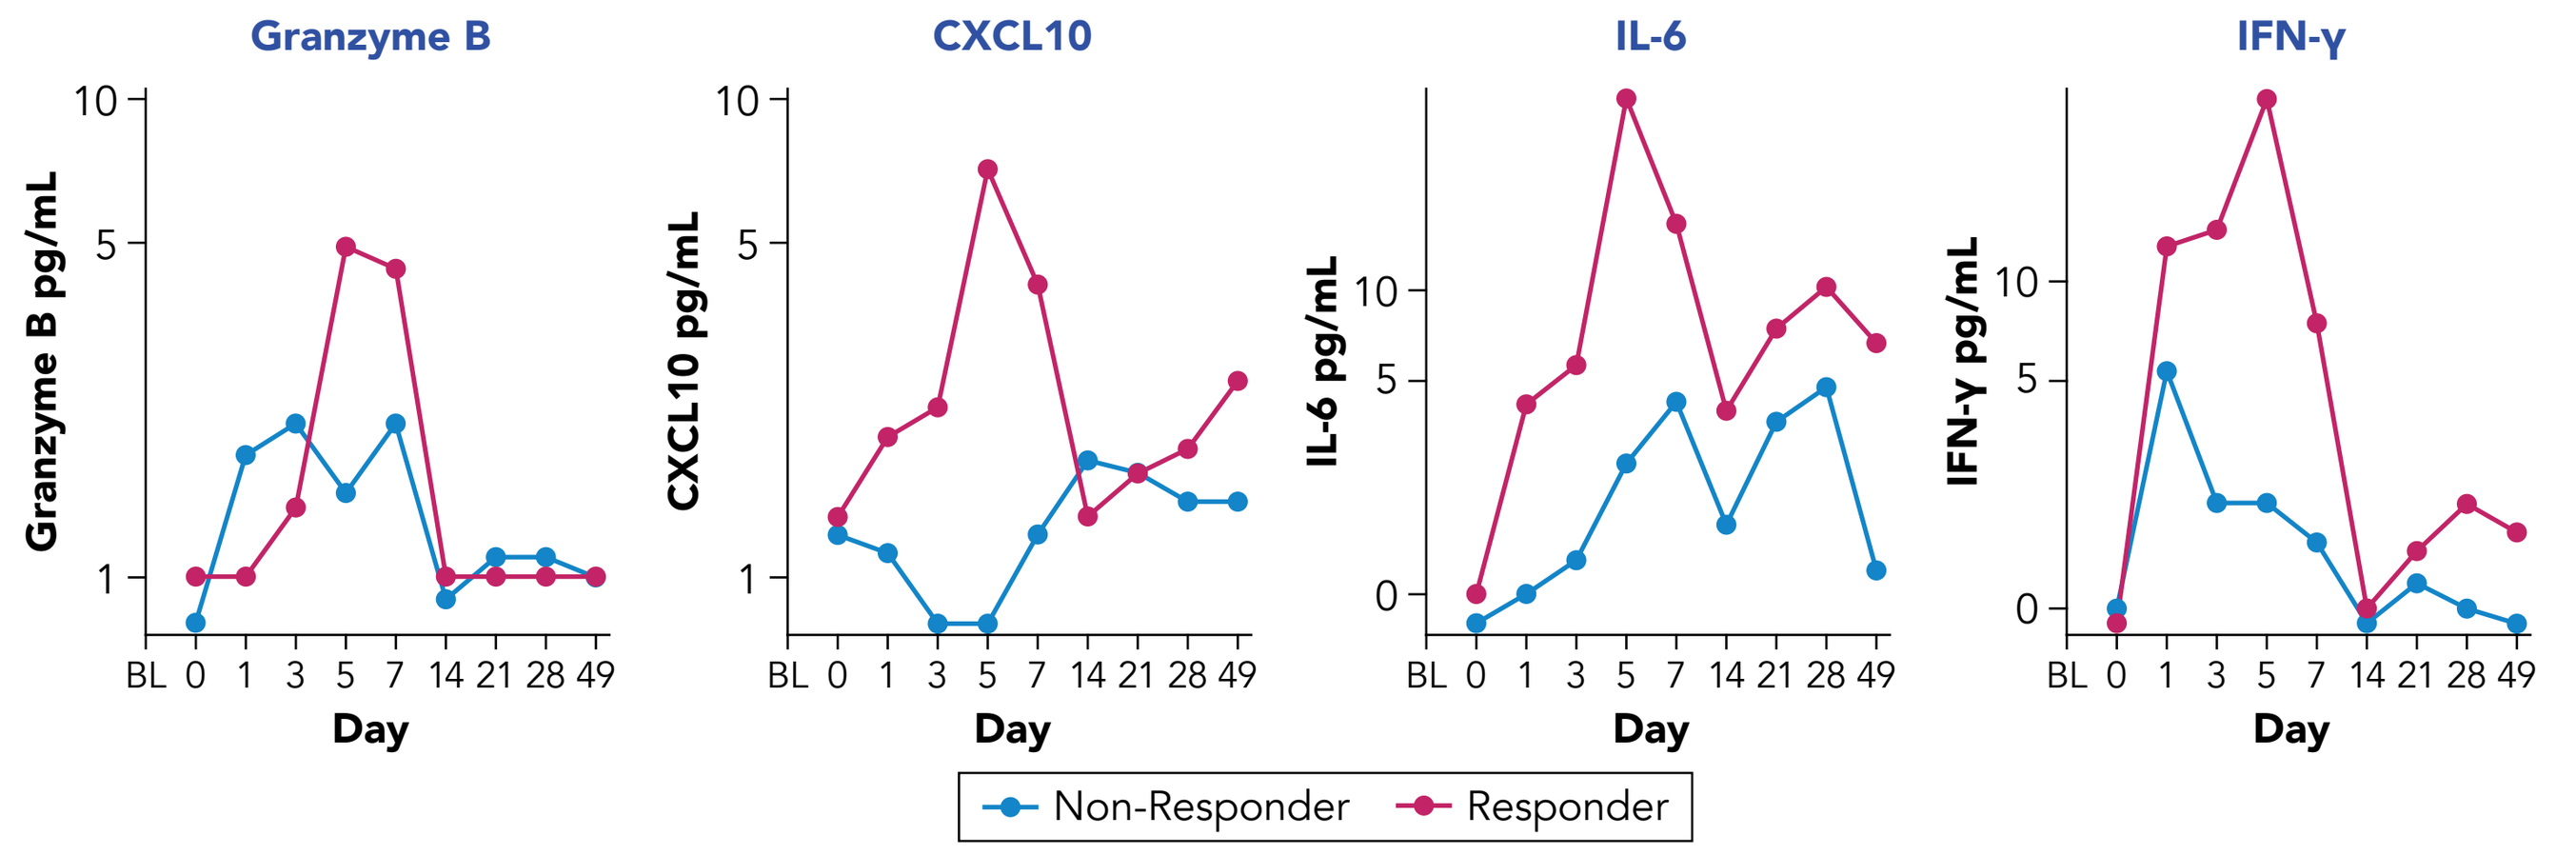
Granzyme B
Granzyme B pg/mL
CXCL10
CXCL10 pg/mL
IL-6
IL-6 pg/mL
IFN-γ
IFN-γ pg/mL
BL 0 1 3 5 7 14 21 28 49
Day
Non-Responder
Responder

BL, baseline; CXCL, chemokine CXC ligand; IFN, interferon; IL, interleukin.

- Immune-modulating cytokines, including interleukin (IL)-6, chemokine CXC ligand (CXCL)10, and interferon (IFN)-γ, were induced in patients following axi-cel and subsequent rituximab infusion on Days 21 to 28 (**Figure 8**)
- In addition, granzyme B, IL-6, CXCL10, and IFN-γ were more prominently elevated in responders versus non-responders

Figure 9. Key Serum Cytokines Elevated Post Axi-Cel Infusion in Association With Safety Outcomes (CRS and NEs)

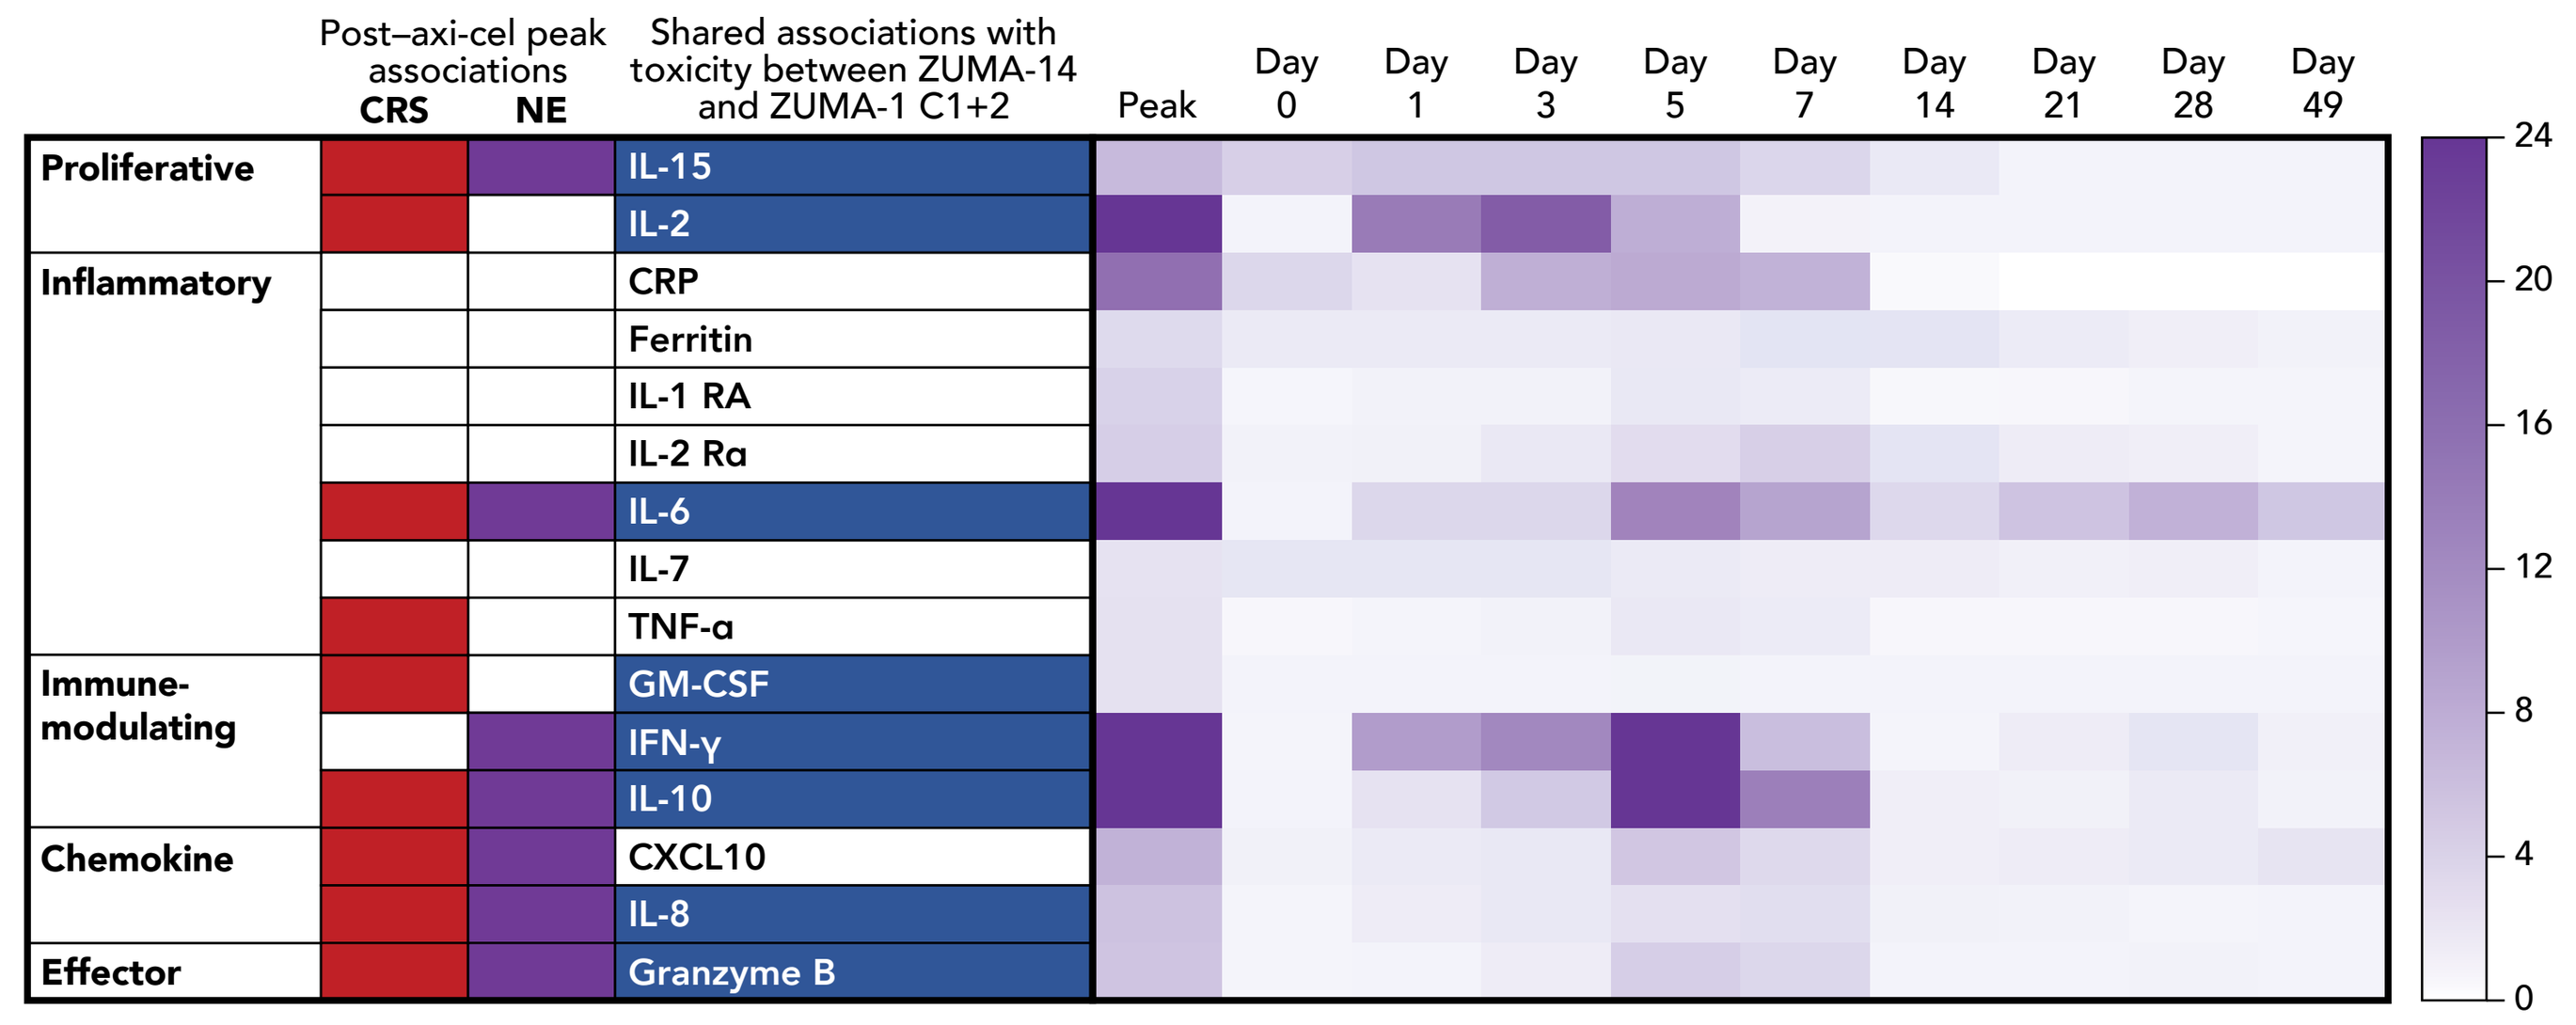
Post–axi-cel peak associations
CRS
NE
Shared associations with toxicity between ZUMA-14 and ZUMA-1 C1+2
Peak
Day 0
Day 1
Day 3
Day 5
Day 7
Day 14
Day 21
Day 28
Day 49
Proliferative
IL-15
IL-2
Inflammatory
CRP
Ferritin
IL-1 RA
IL-2 Rα
IL-6
IL-7
TNF-α
Immune-modulating
GM-CSF
IFN-γ
IL-10
Chemokine
CXCL10
IL-8
Effector
Granzyme B
24
20
16
12
8
4
0

Heatmap shows serum analytes that are elevated post axi-cel infusion in >50% patients with fold change >2 from peak to baseline. The median of fold change from baseline at each post-baseline time point and peak were plotted. *P* values were calculated from Wilcoxon rank sum test (only cytokines with *P*<.05 are shown).
Axi-cel, axicabtagene ciloleucel; C, cohort; CRP, C-reactive protein; CRS, cytokine release syndrome; CXCL, chemokine CXC ligand; GM-CSF, granulocyte-macrophage colony-stimulating factor; IFN, interferon; IL, interleukin; NE, neurologic event; RA, receptor antagonist; TNF, tumor necrosis factor.

- Peak inflammatory and effector cytokines significantly associated with both Grade ≥2 CRS and NEs in ZUMA-14, including granzyme B, IL-10, IL-6, and IL-8 (**Figure 9**), were also associated with Grade ≥3 CRS and NEs in ZUMA-1 Cohorts 1+2[4]
- IFN-γ was also associated with higher-grade NEs in ZUMA-1 Cohorts 1+2[5]

CONCLUSIONS

- Results from ZUMA-14 demonstrated that axi-cel in combination with rituximab elicited a high CR rate and durable PFS in patients with refractory LBCL
 - ORR was 88%, the CR rate was 73%, and the median PFS was 18.6 months
- The safety profile of axi-cel in combination with rituximab was manageable, with no new safety signals detected
 - No patients experienced a Grade ≥3 CRS event, whereas Grade ≥3 NEs occurred in 15% of patients
- Peak CAR T-cell levels and pharmacodynamic findings in ZUMA-14 appear to be consistent with those observed in ZUMA-1
 - Peak inflammatory and effector cytokines significantly associated with both CRS and NEs in the present study were also induced in ZUMA-1[4]
- Overall, axi-cel in combination with rituximab in patients with R/R LBCL showed encouraging activity with a manageable safety profile

REFERENCES

1. YESCARTA® (axicabtagene ciloleucel) [Prescribing information]. Kite Pharma, Inc; 2021.
2. Locke FL, et al. *Lancet Oncol.* 2019;20:31-42.
3. Mihara K, et al. *Br J Haematol.* 2010;151(1):37-46.
4. Neelapu SS, et al. *N Engl J Med.* 2017;377(26):2531-2544.
5. Locke FL, et al. *Blood Adv.* 2020;4(19):4898-4911.

ACKNOWLEDGMENTS

- The patients, families, friends, and caregivers
- The study investigators, coordinators, and health care staff at each study site
- Medical writing support was provided by Jennifer Yang, PhD, of Nexus Global Group Science, funded by Kite, a Gilead Company
- This study was funded by Kite, a Gilead Company
- © 2022 American Society of Clinical Oncology, Inc. Reused with permission. This abstract was accepted and previously presented at the 2022 ASCO Annual Meeting. All rights reserved.

DISCLOSURES

PS: consulting or advisory role with Roche-Genentech, Hutchison Medipharma, ADC Therapeutics, TG Therapeutics; research funding from AstraZeneca-Acerta, ALX Oncology. **LL:** consultancy and travel support with AbbVie, AstraZeneca, Merck, TG Therapeutics, Janssen, Epizyme, Kite, a Gilead Company, Celgene, Pharmacyclics, Bristol Myers Squibb, ADC Therapeutics, BeiGene, Seattle Genetics; speakers' bureau participation and travel support for Kite, BeiGene, Pharmacyclics, Janssen, AstraZeneca, Seattle Genetics, TG Therapeutics, Epizyme, Karyopharm, Celgene, Bristol Myers Squibb; Participation on a Data Safety Advisory Board with TG therapeutics, AbbVie, Pharmacyclics, Janssen, AstraZeneca, Seattle Genetics, ADC Therapeutics. **PShiraz:** research funding from Kite, a Gilead Company, and Orca Bio. **EB:** honoraria from Kite, a Gilead Company; consultancy or advisory role for Kite, Gilead, and Roche; speakers' bureau participation for Kite, and AstraZeneca; and research funding from MustangBio, Merck, Amgen, and AstraZeneca. **OOO:** consultancy or advisory role for Kite, a Gilead Company, Janssen, Pfizer, Novartis, and Curio Science; honoraria from Kite; and research funding from Kite. **MU:** advisory board role for Gilead and Stemline. **AR:** honoraria from Cigna, Amgen, and Takeda; consultancy or advisory role for Amgen and Takeda; and speakers' bureau participation for Takeda. **JS:** employment with Kite, a Gilead Company. **RS:** employment with Kite, a Gilead Company and Atara; leadership role with Kite and Atara; stock or other ownership in Kite and Atara; and patents, royalties, and other intellectual property from Kite. **JK:** employment with Kite, a Gilead Company; stock or other ownership in Kite. **PM:** employment with Kite, a Gilead Company. **JD:** employment with Kite, a Gilead Company; stock or other ownership in Gilead Sciences; consultancy or advisory role for GliaCure/Tufts; and patents, royalties, or other intellectual property from: Patent US8598141 (Dec 03, 2013). **MS:** employment with Kite, a Gilead Company; stock or other ownership in Gilead Sciences; honoraria from Kite; travel support from Kite and other relationships with Kite. **HX:** employment with Kite, a Gilead Company. **KP:** research funding for Aptevo Therapeutics, AstraZeneca, Bristol Myers Squibb, Celgene, Curis Inc., Epizyme, Genentech/Roche, Kite, a Gilead Company, MEI Pharma, Millennium/Takeda, Sunesis Pharmaceuticals, Trillium Therapeutics, Velos Bio, Xencor; consultancy for AstraZeneca, BeiGene, Bristol Myers Squibb, Celgene, Epizyme, Genentech/ Roche, Juno Pharmaceuticals, Kite, Loxo Oncology, MEI Pharma, MorphoSys, Pharmacyclics/ Janssen, TG Therapeutics; speakers bureau for AstraZeneca, Bristol Myers Squibb, Celgene, Genentech/Roche, Kite, Pharmacyclics/Janssen, TG Therapeutics.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from the author of this poster.

Presented at the 2022 European Hematology Association Annual Meeting