Poster
4284

Minority Patients in the US Receiving Chimeric Antigen Receptor (CAR) T-cell Therapy: A SEER-based Simulation on Representation and Impact of Proximity to Authorized Treatment Centers (ATCs)

Mendel Goldfinger, MD[1,*], Zhen-Huan Hu, MPH[2,*], Aaron P. Rapoport, MD[3], Tanya Siddiqi, MD[4], Lazaros J. Lekakis, MD[5], Marcelo C. Pasquini, MD, MS[6], Shilpa Shahani, MD[2], Elande Baro, PhD[2], Jin Xie, PhD[2], Qinghua Song, PhD[2], Fang Sun, MD, PhD[2], Hairong Xu, MD, PhD[2], Frederick L. Locke, MD[7]

[1]Albert Einstein College of Medicine, Montefiore Medical Center, Bronx, NY, USA; [2]Kite, a Gilead Company, Santa Monica, CA, USA; [3]University of Maryland Greenebaum Comprehensive Cancer Center, Baltimore, MD, USA; [4]City of Hope National Medical Center, Duarte, CA, USA; [5]Sylvester Comprehensive Cancer Center, University of Miami Health System, Miami, FL, USA; [6]Center for International Blood and Marrow Transplant Research, Medical College of Wisconsin, Milwaukee, WI, USA; [7]Department of Blood and Marrow Transplant and Cellular Immunotherapy, H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL, USA

**Co-lead authors*

BACKGROUND

- Patients of minority race or ethnicity are often underrepresented in oncology clinical trials[1]
- Incidence rates for B-cell malignancies differ across races and ethnicities (**Figure 1**)[2,3]
- Survival rate is lower among Black or African American patients for most cancer types and among Hispanic patients for certain cancer types compared to White patients[4,5]

Figure 1. Age-adjusted Incidence Rates of B-cell Malignancies Analyzed by Race and Ethnicity[2,3]

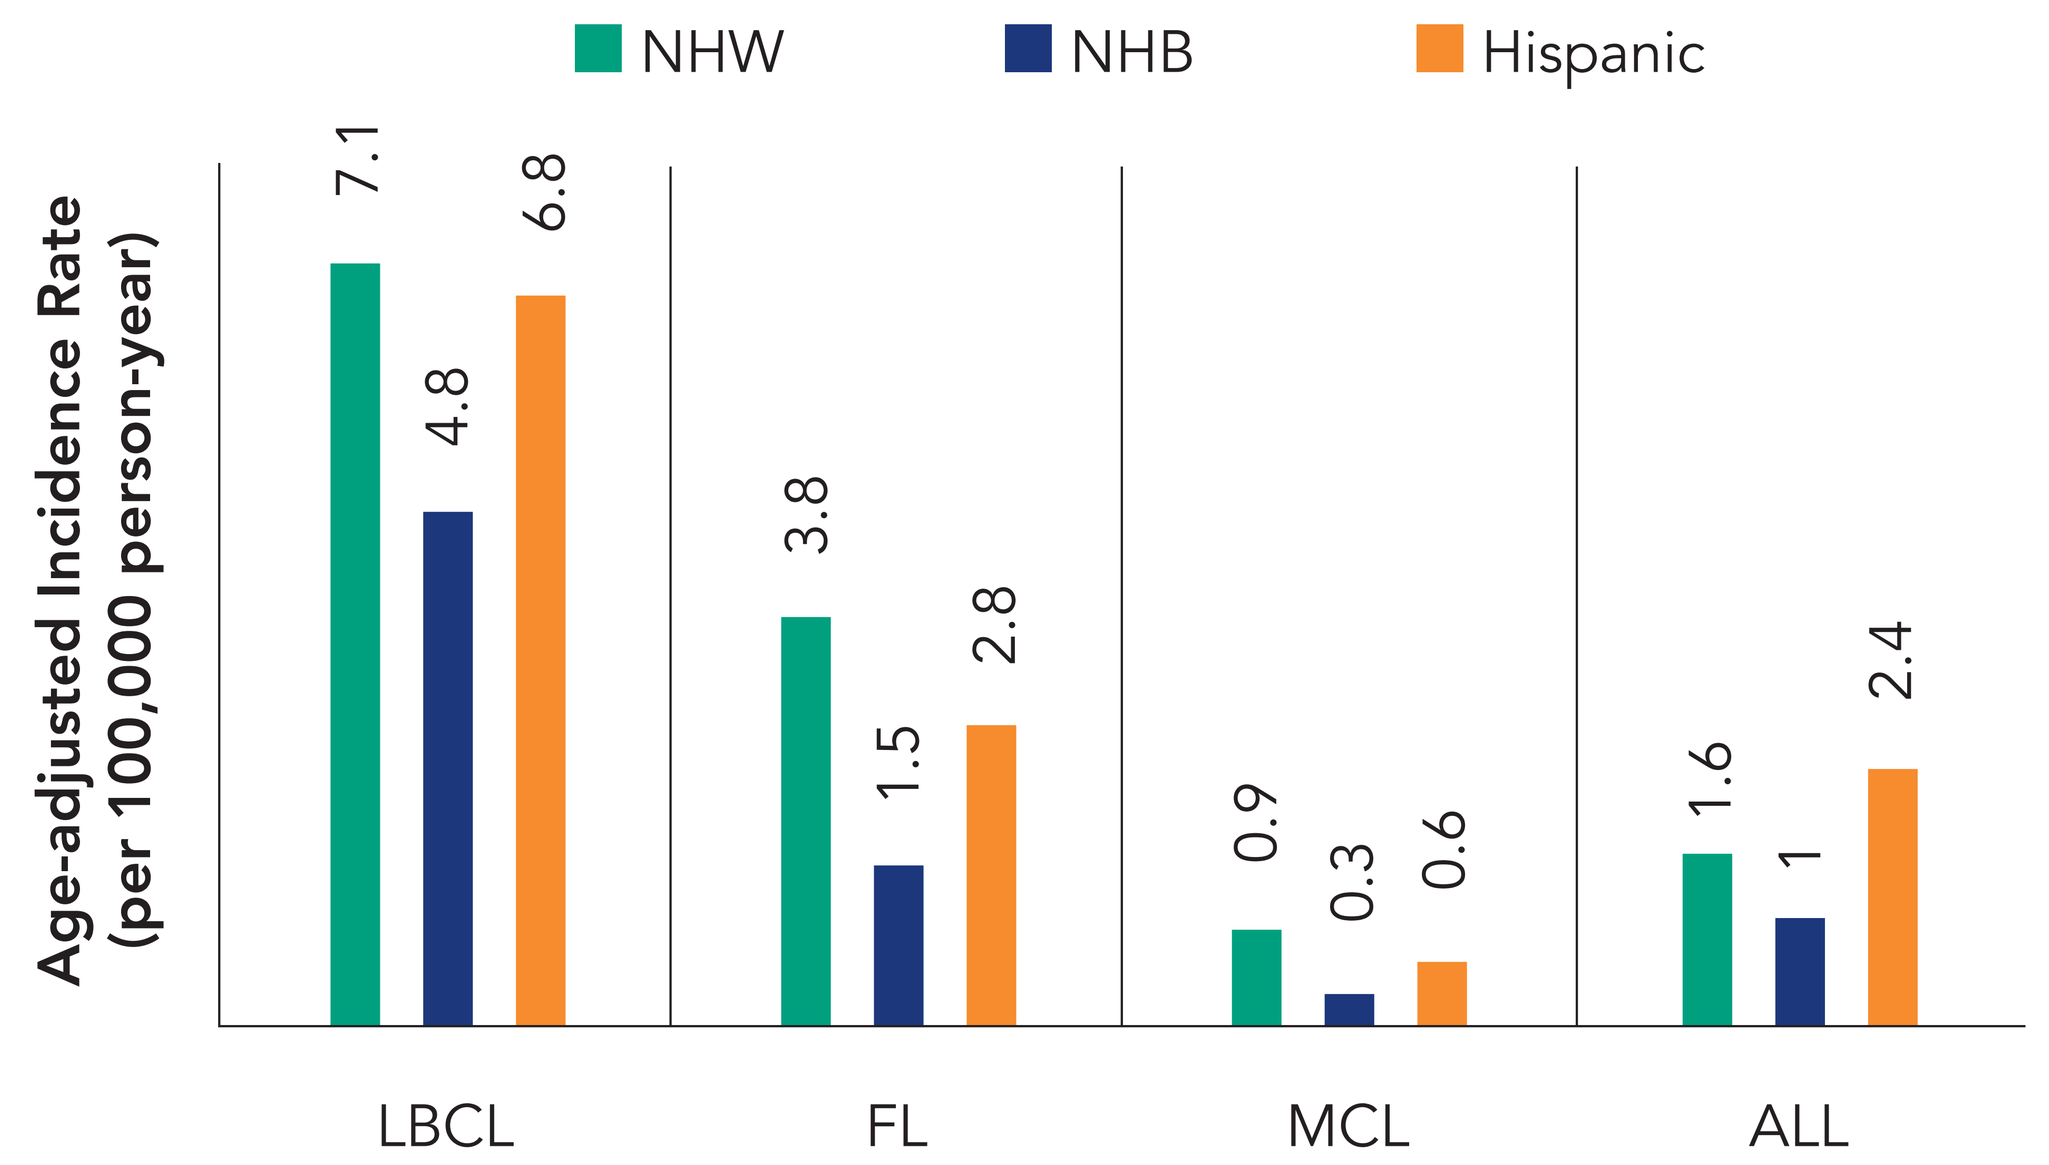
NHW
NHB
Hispanic
Age-adjusted Incidence Rate (per 100,000 person-year)
7.1
4.8
6.8
3.8
1.5
2.8
0.9
0.3
0.6
1.6
1
2.4
LBCL
FL
MCL
ALL

ALL, acute lymphoblastic leukemia; FL, follicular lymphoma; LBCL, large B-cell lymphoma; MCL, mantle cell lymphoma; NHB, non-Hispanic Black; NHW, non-Hispanic White.

METHODS

STUDY DESIGN

Objectives
- To estimate proportions of minority patients potentially eligible for either CAR T-cell trials or receiving standard-of-care CAR T-cell therapy in real-world settings
- To assess whether proximity to a CAR T-cell authorized treatment center (ATC) is a barrier for treatment

Data sources
- SEER Research Plus Data (17 registries, Nov 2021 submission)
- US County Population Data (1969–2020)[a]

Race/ethnicity categories
- Non-Hispanic White (NHW)
- Non-Hispanic Black (NHB)
- Non-Hispanic Other (NHO)
- Hispanic (all races)

Disease indications
- Large B-cell lymphoma (LBCL)
- Follicular lymphoma (FL)
- Mantle cell lymphoma (MCL)
- B-cell precursor acute lymphoblastic leukemia (B-ALL)

[a]Produced under a collaborative arrangement between the US Census Bureau and the National Center for Health Statistics with support from the National Cancer Institute.
CAR, chimeric antigen receptor; SEER, Surveillance, Epidemiology, and End Results.

Simulation Settings

	Clinical Trial Settings	Real-world Settings
≤50 miles of any ATC	• Populations based on counties within 50 miles of any **clinical trial ATC** • Sample sizes used in simulation were based on the corresponding **ZUMA trials**	• Populations based on counties within 50 miles of any **commercial ATC** • Sample sizes used in simulation were based on the corresponding **US PASS**[a]
Entire US	• Populations based on the entire US • Sample sizes used in simulation were based on the corresponding **ZUMA trials**	• Populations based on the entire US • Sample sizes used in simulation were based on the corresponding **US PASS**[a]

[a]For axicabtagene ciloleucel or brexucabtagene autoleucel, respectively.
ATC, authorized treatment center; PASS, post-authorization safety study.

SIMULATION METHOD

1. Estimate 5-year **limited duration prevalence** (LDP) prior to January 1, 2019 for adults aged ≥20 years based on SEER Research Plus Data by each combination of race/ethnicity, sex, and age
2. Multiply 5-year LDP with population counts based on US County Population Data in each combination of race/ethnicity, sex, and age to calculate **prevalence counts**
3. Draw random samples based on the prevalence counts to estimate the **proportions of patients by race/ethnicity and 95% CIs** for a given sample size

Repeat the process
- By disease indications
- For clinical trial and real-world settings
- For regions ≤50 miles of any ATCs and the entire US

ATC, authorized treatment center; LDP, limited duration prevalence; SEER, Surveillance, Epidemiology, and End Results.

RESULTS

- As of January 1, 2019, 176.9 of 245.8 million people aged ≥20 years (72%) lived within 50 miles of a commercial ATC (**Figure 2**)
- Proportions of minority race or ethnicity in those areas are comparable to proportions of minorities in the overall US population

Figure 2. Populations Within 50 Miles of Any Commercial ATC[a]

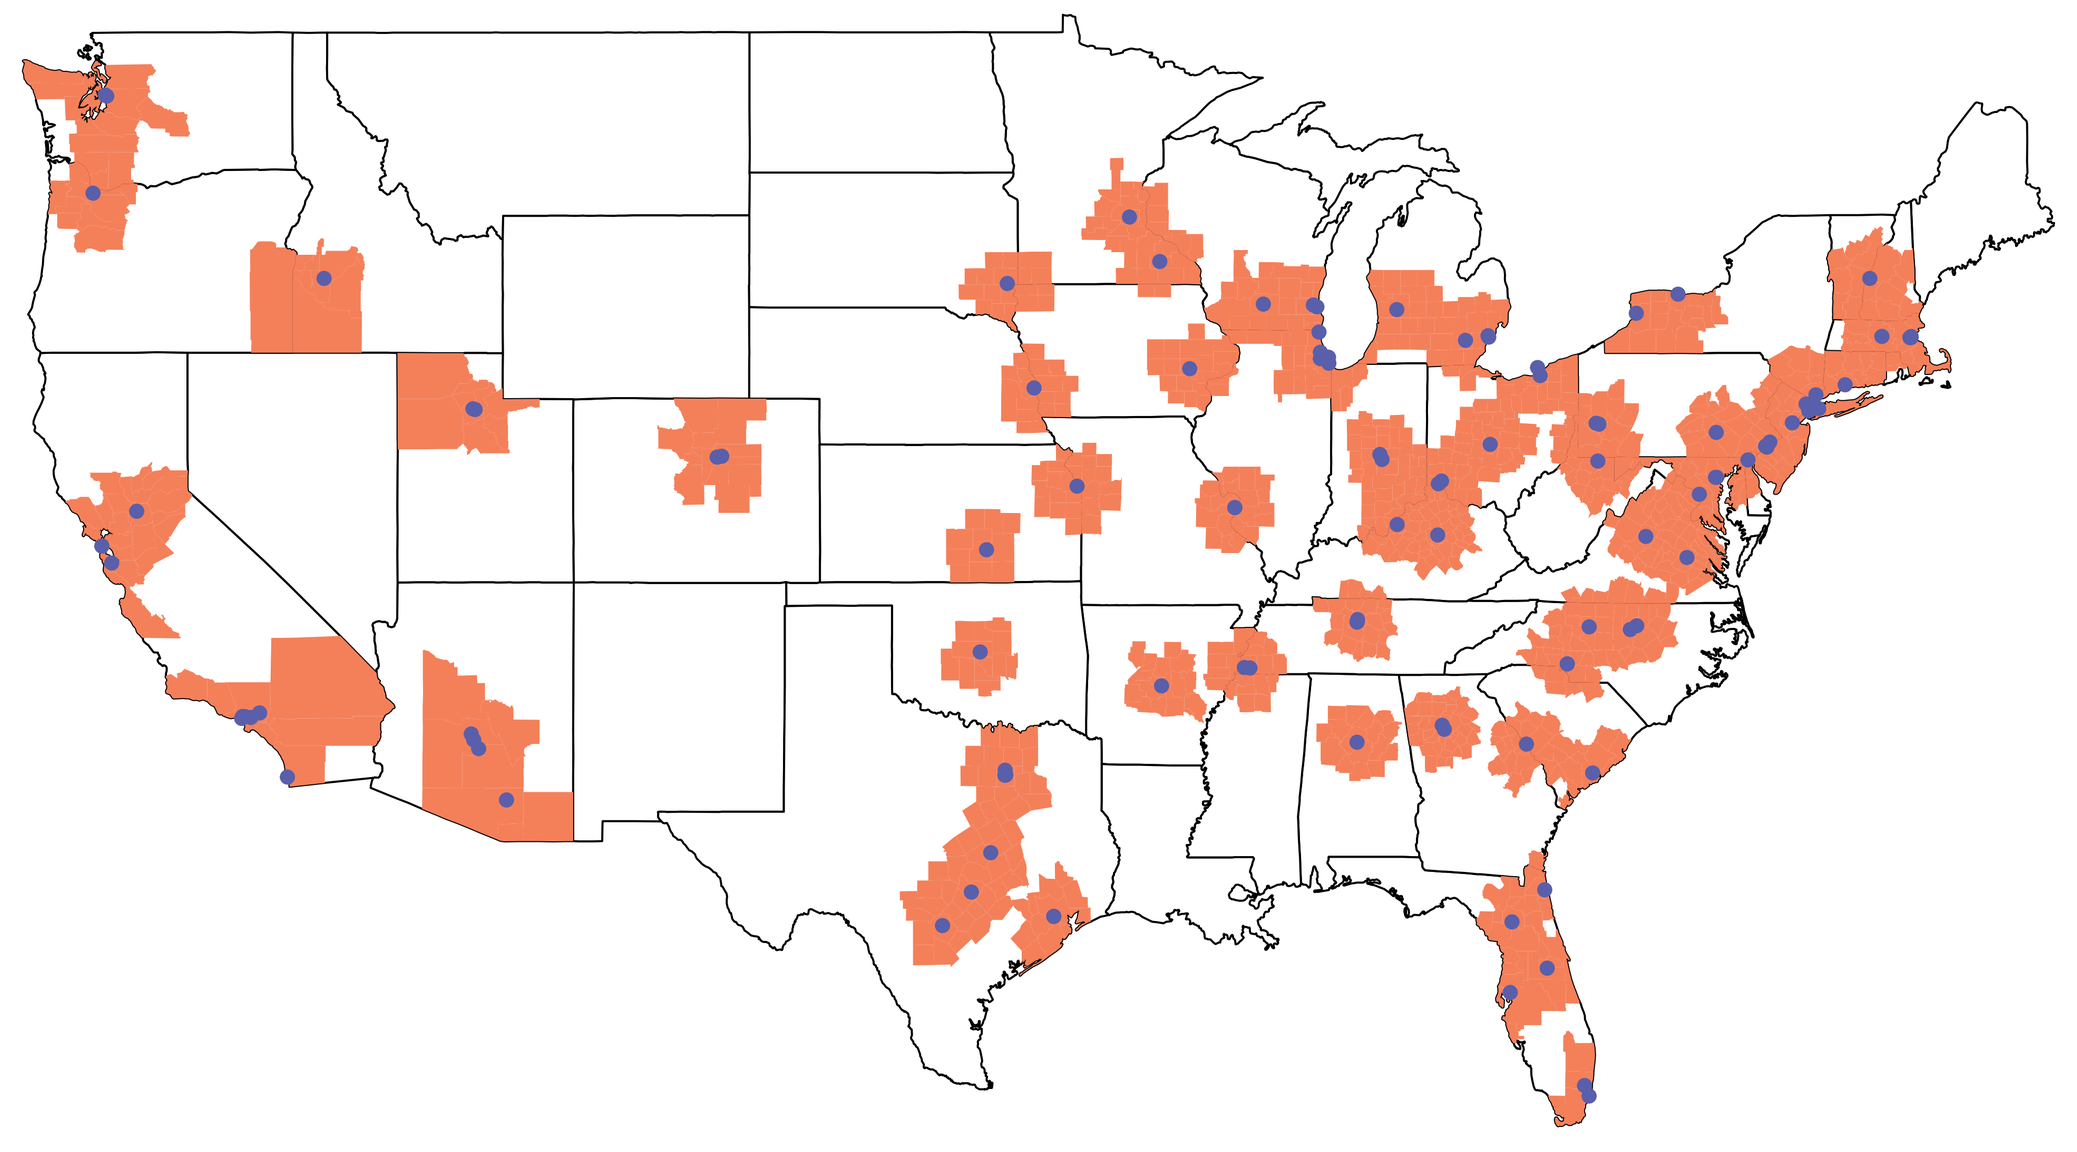

[a]Commercial ATCs for axicabtagene ciloleucel or brexucabtagene autoleucel.

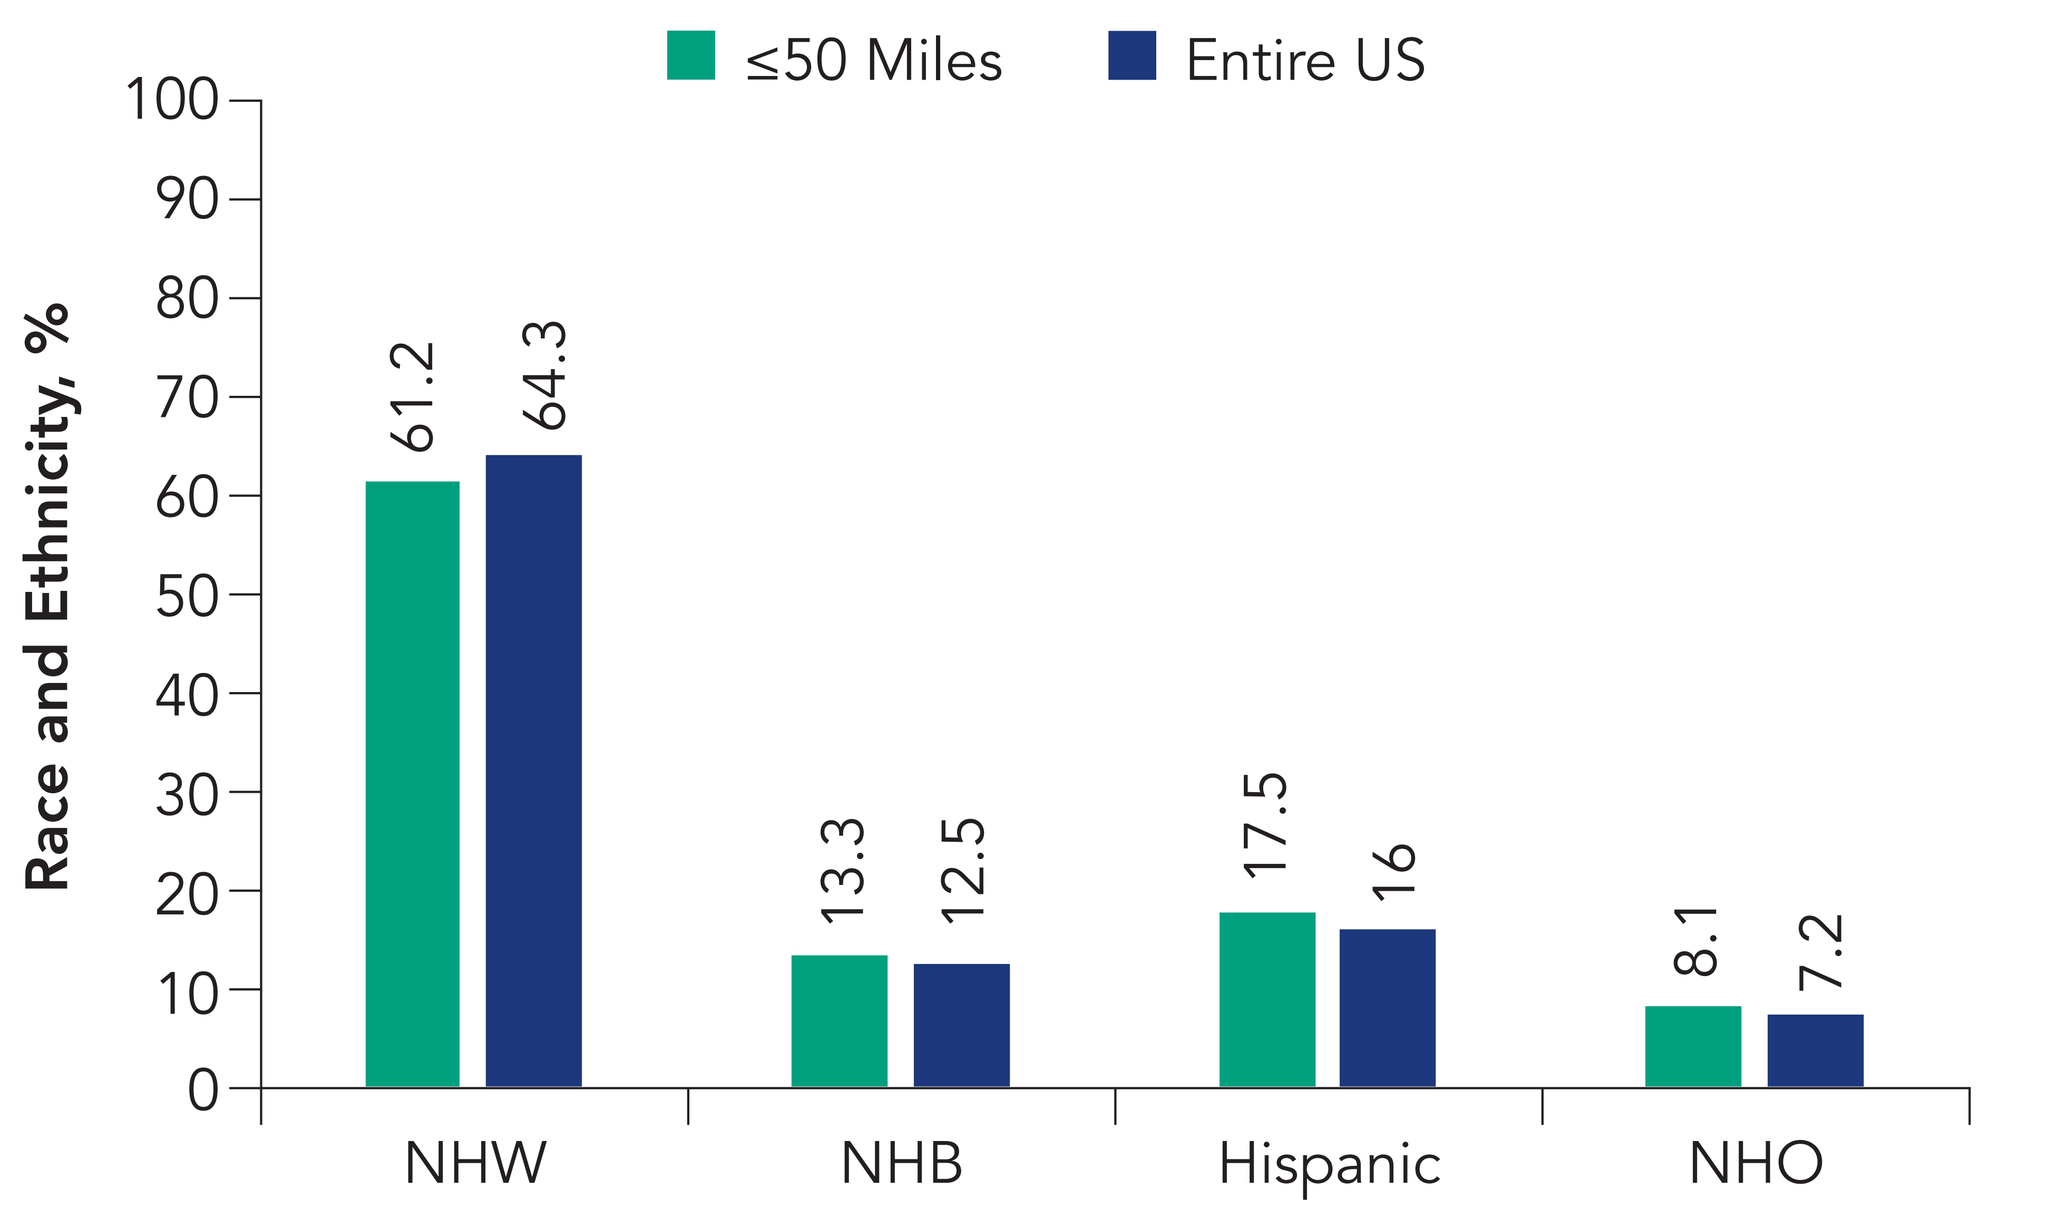
Overall Race and Ethnicity Populations in the US
≤50 Miles
Entire US
Race and Ethnicity, %
61.2
64.3
13.3
12.5
17.5
16
8.1
7.2
NHW
NHB
Hispanic
NHO

ATC, authorized treatment center; NHB, non-Hispanic Black; NHO, non-Hispanic Other; NHW, non-Hispanic White.

- In clinical trial settings, the estimated proportions of NHB patients ≤50 miles of an ATC were numerically but not statistically significantly higher (except in B-ALL) compared with the actual enrollment. The estimated proportions of Hispanic patients were statistically higher in FL and B-ALL (**Figure 3**)
- In real-world settings, the estimated proportions of NHB patients were comparable to those in trial settings (<1% difference) while the estimated proportions of Hispanic patients were numerically lower compared with trial settings

Figure 3. Estimated Proportions of Minority Patients

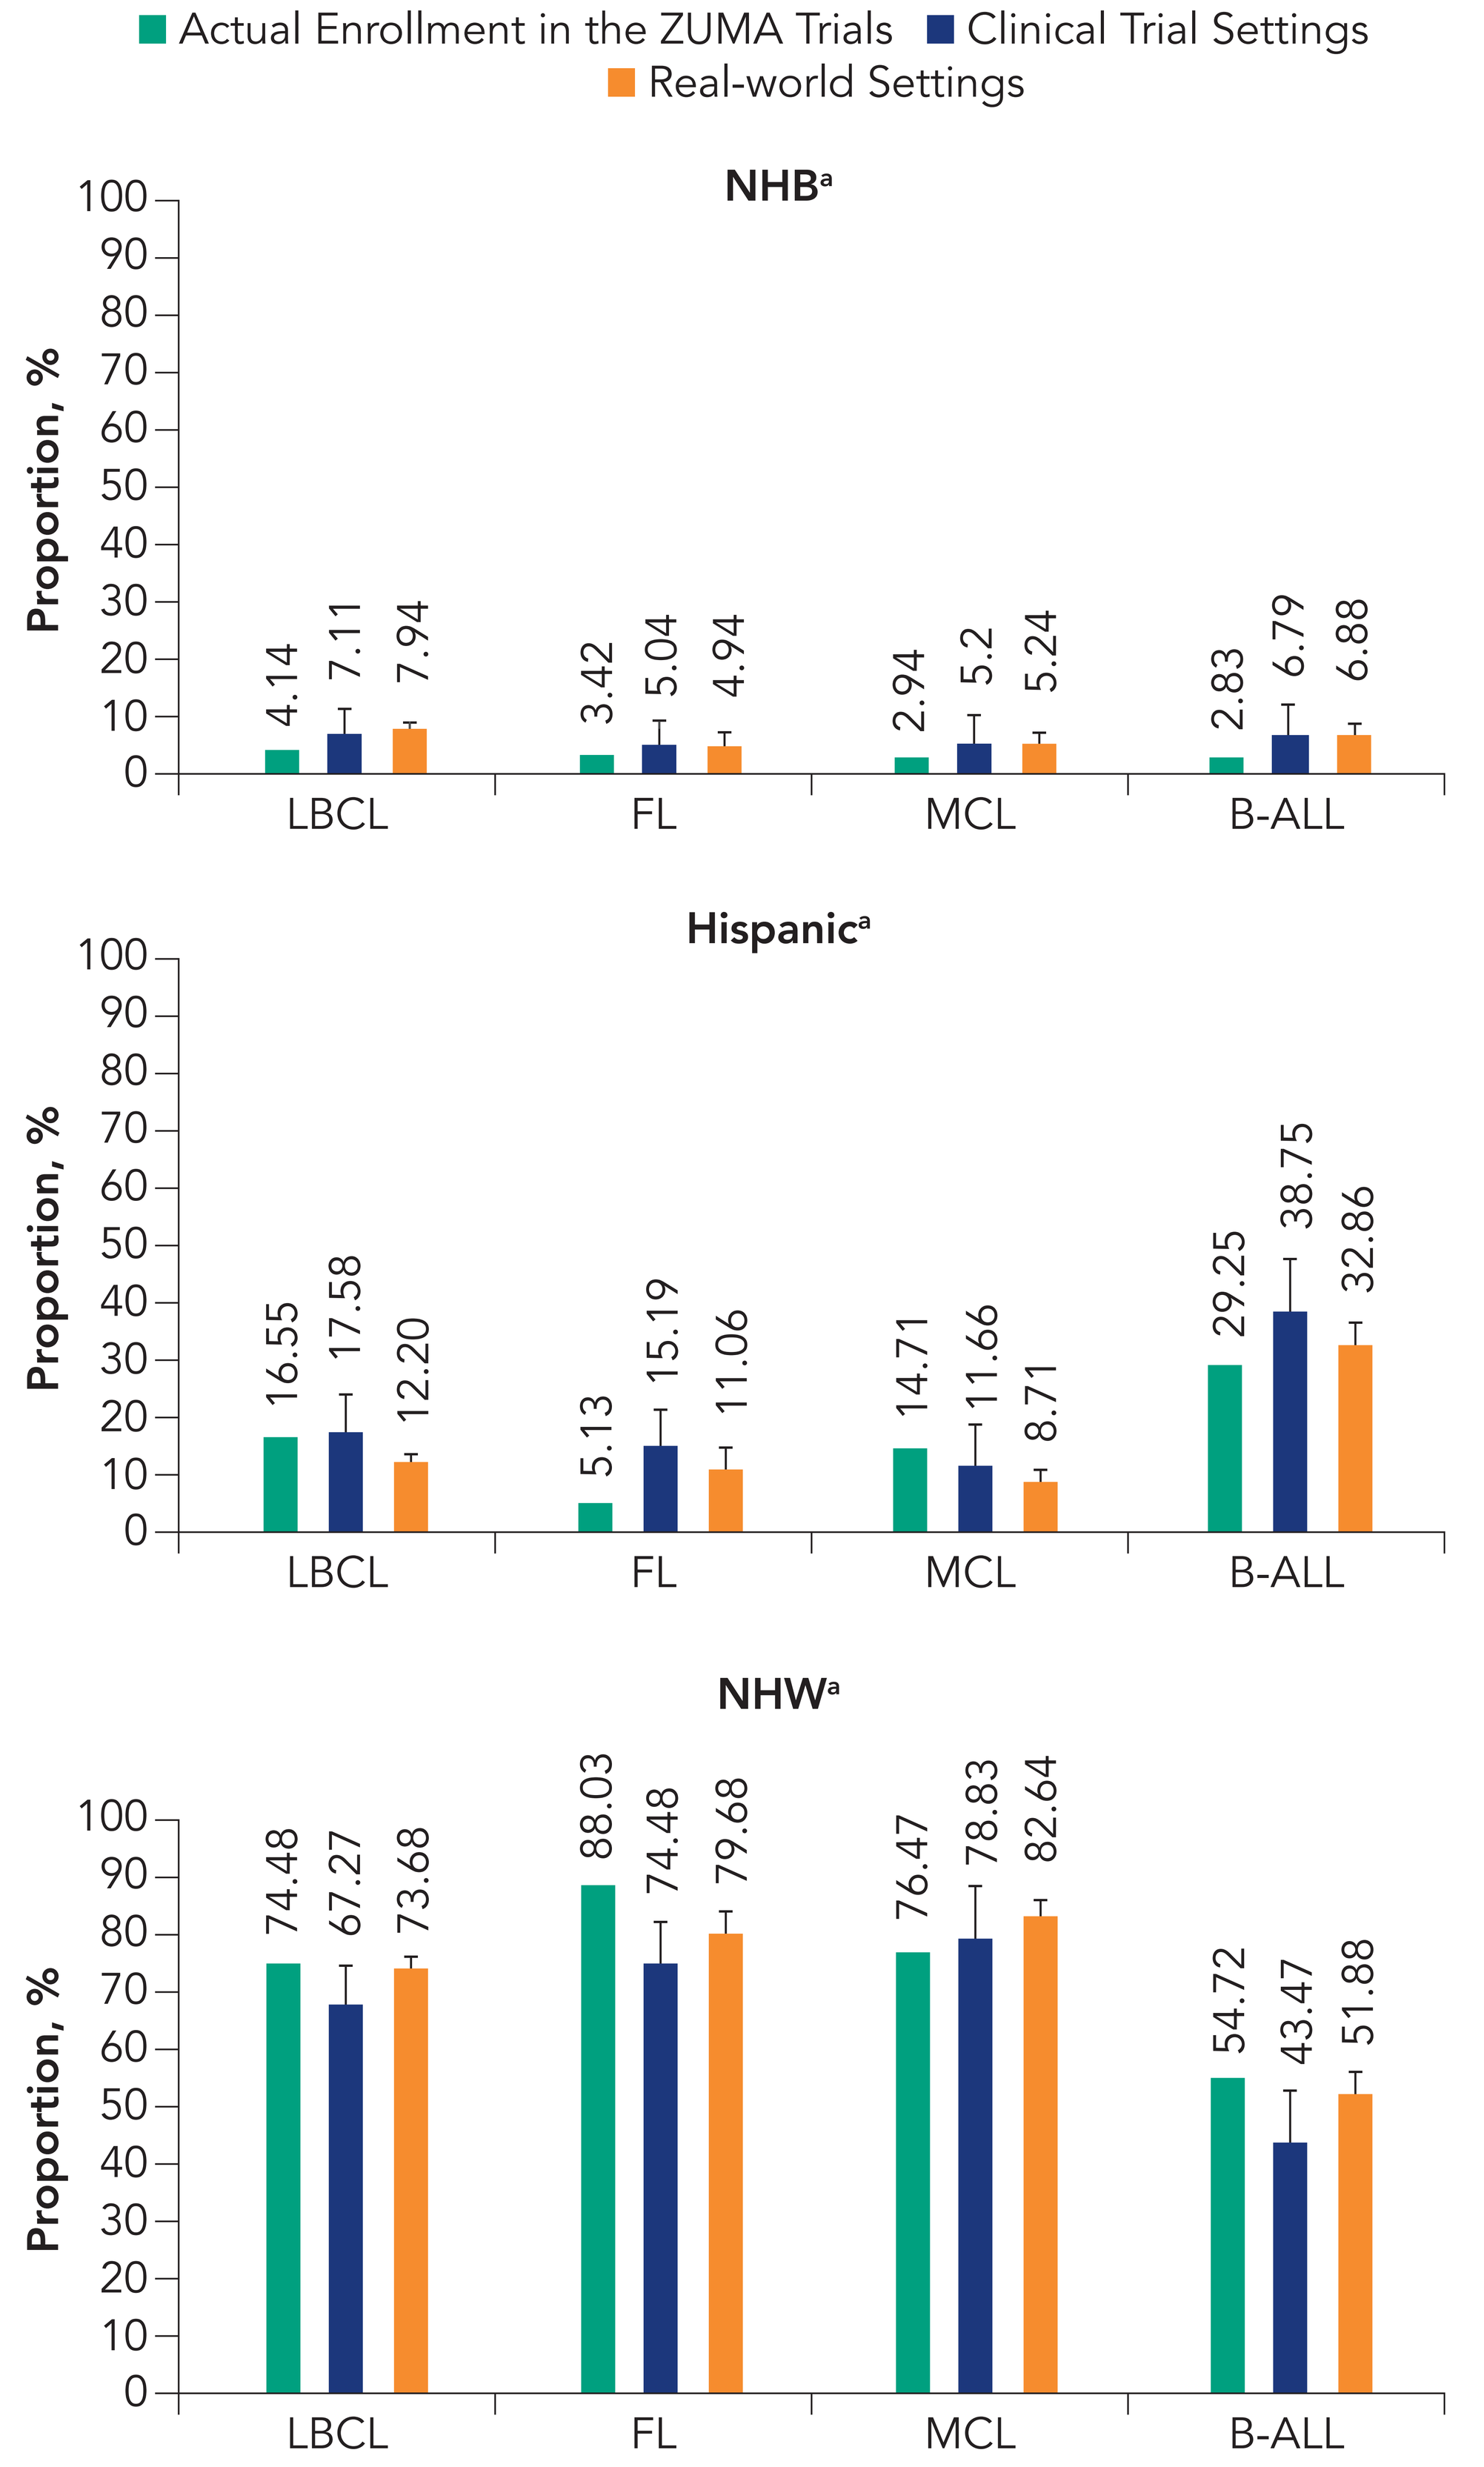
Actual Enrollment in the ZUMA Trials
Clinical Trial Settings
Real-world Settings
NHB[a]
Proportion, %
4.14 7.11 7.94
3.42 5.04 4.94
2.94 5.2 5.24
2.83 6.79 6.88
Hispanic[a]
16.55 17.58 12.20
5.13 15.19 11.06
14.71 11.66 8.71
29.25 38.75 32.86
NHW[a]
74.48 67.27 73.68
88.03 74.48 79.68
76.47 78.83 82.64
54.72 43.47 51.88
LBCL
FL
MCL
B-ALL

[a]Based on simulations of regions ≤50 miles of an ATC. Actual clinical trial enrollments were based on US patients in ZUMA-1 Phase 1 and Phase 2 Cohorts 1, 2 and 6 for LBCL, ZUMA-5 for FL, ZUMA-2 Cohort 1 for MCL, and ZUMA-3 Phase 1 and 2 for B-ALL. 95% confidence intervals (error bars) for the estimates were derived from 3,000 random samplings of the prevalence counts.
B-ALL, B-cell precursor acute lymphoblastic leukemia; FL, follicular lymphoma; LBCL, large B-cell lymphoma; MCL, mantle cell lymphoma; NHB, non-Hispanic Black; NHW, non-Hispanic White.

REFERENCES

1. Awidi M, et al. *JCO Oncol Pract.* 2021;17(5):265–271.
2. Teras LR, et al. *CA Cancer J Clin.* 2016;66(6):443–459.
3. Feng Q, et al. *Am J Epidemiol.* 2021;190(4):519–527.
4. DeSantis CE, et al. *CA Cancer J Clin.* 2019;69(3):211–233.
5. Miller KD, et al. *CA Cancer J Clin.* 2021;71(6):466–487.

ACKNOWLEDGMENTS

- The authors thank Hailin Wang, MPH, for his support with quality control of data
- Editorial assistance was provided by Fishawack Health, funded by Kite, a Gilead Company

- Proportions of NHB patients ≤50 miles of an ATC across all disease types were comparable with the estimates of the entire US (<1% difference) regardless of trial or real-world settings (**Figures 4** and **5**)
- For Hispanic patients, the estimated proportions ≤50 miles of an ATC were numerically higher compared with the estimates of the entire US in the trial settings; but the estimates were more comparable in real-world settings

Figure 4. Impact of Proximity to ATCs in Clinical Trial Settings

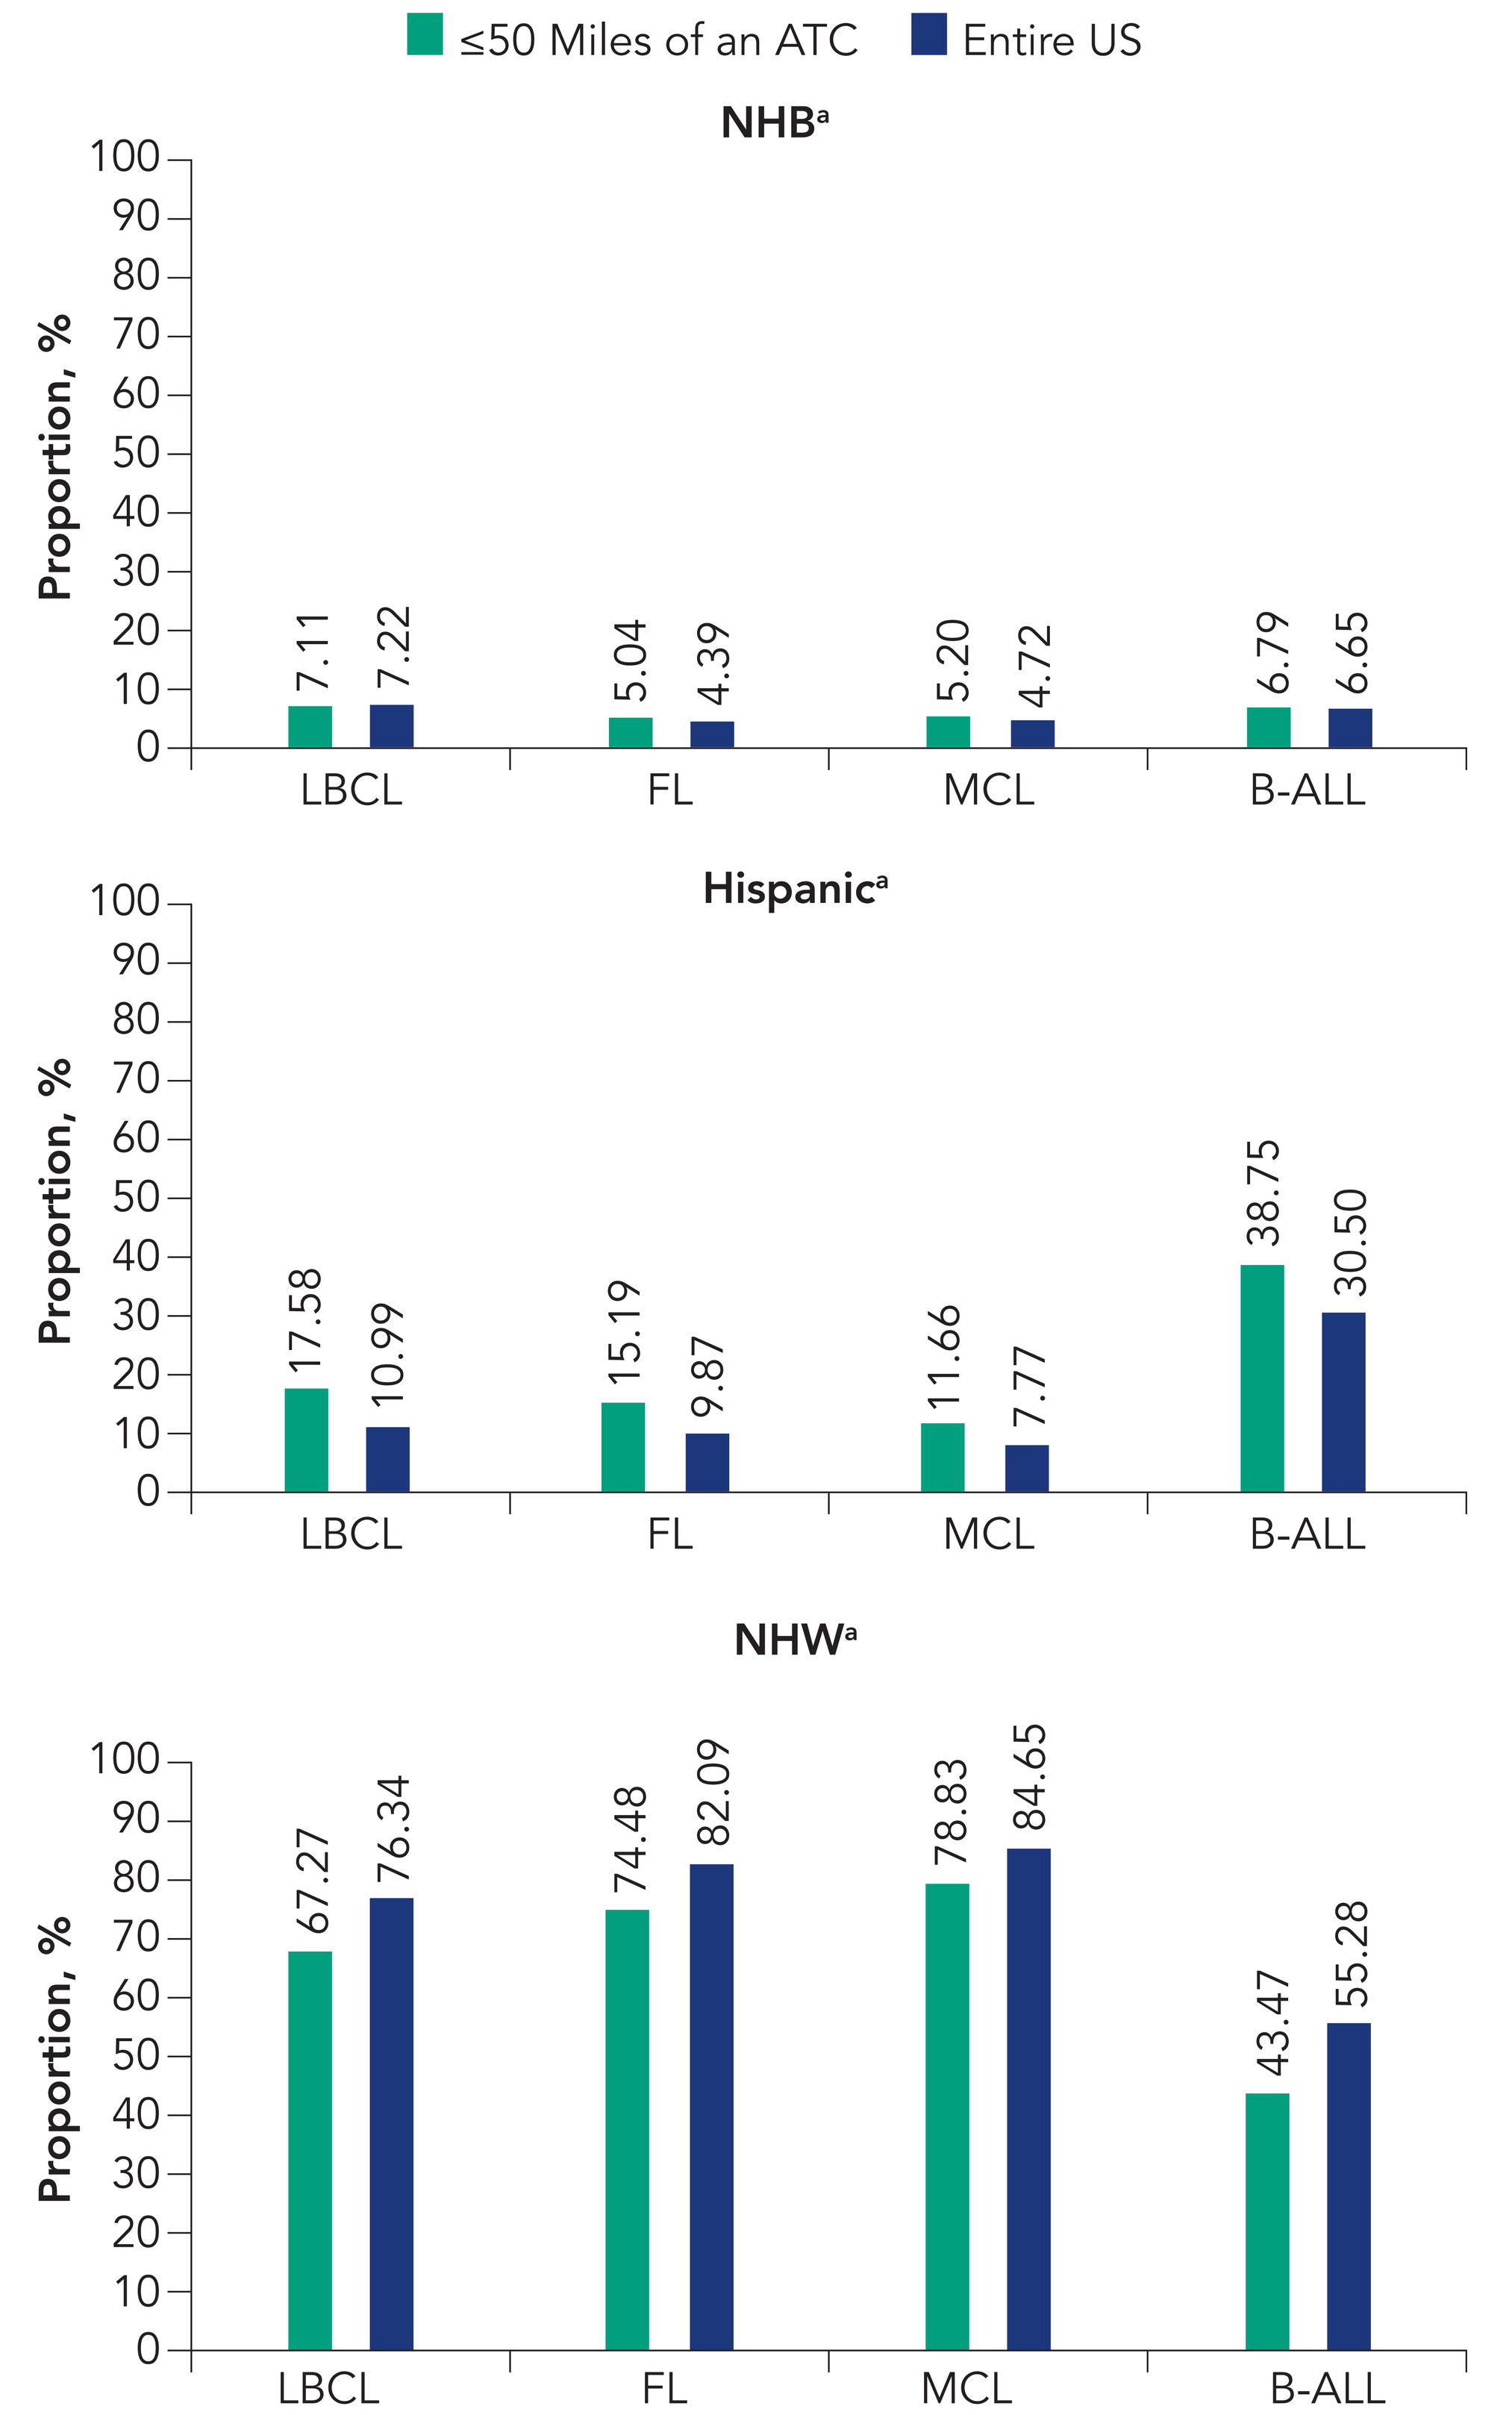
≤50 Miles of an ATC
Entire US
NHB[a]
Proportion, %
7.11 7.22
5.04 4.39
5.20 4.72
6.79 6.65
Hispanic[a]
17.58 10.99
15.19 9.87
11.66 7.77
38.75 30.50
NHW[a]
67.27 76.34
74.48 82.09
78.83 84.65
43.47 55.28
LBCL
FL
MCL
B-ALL

Figure 5. Impact of Proximity to ATCs in Real-world Settings

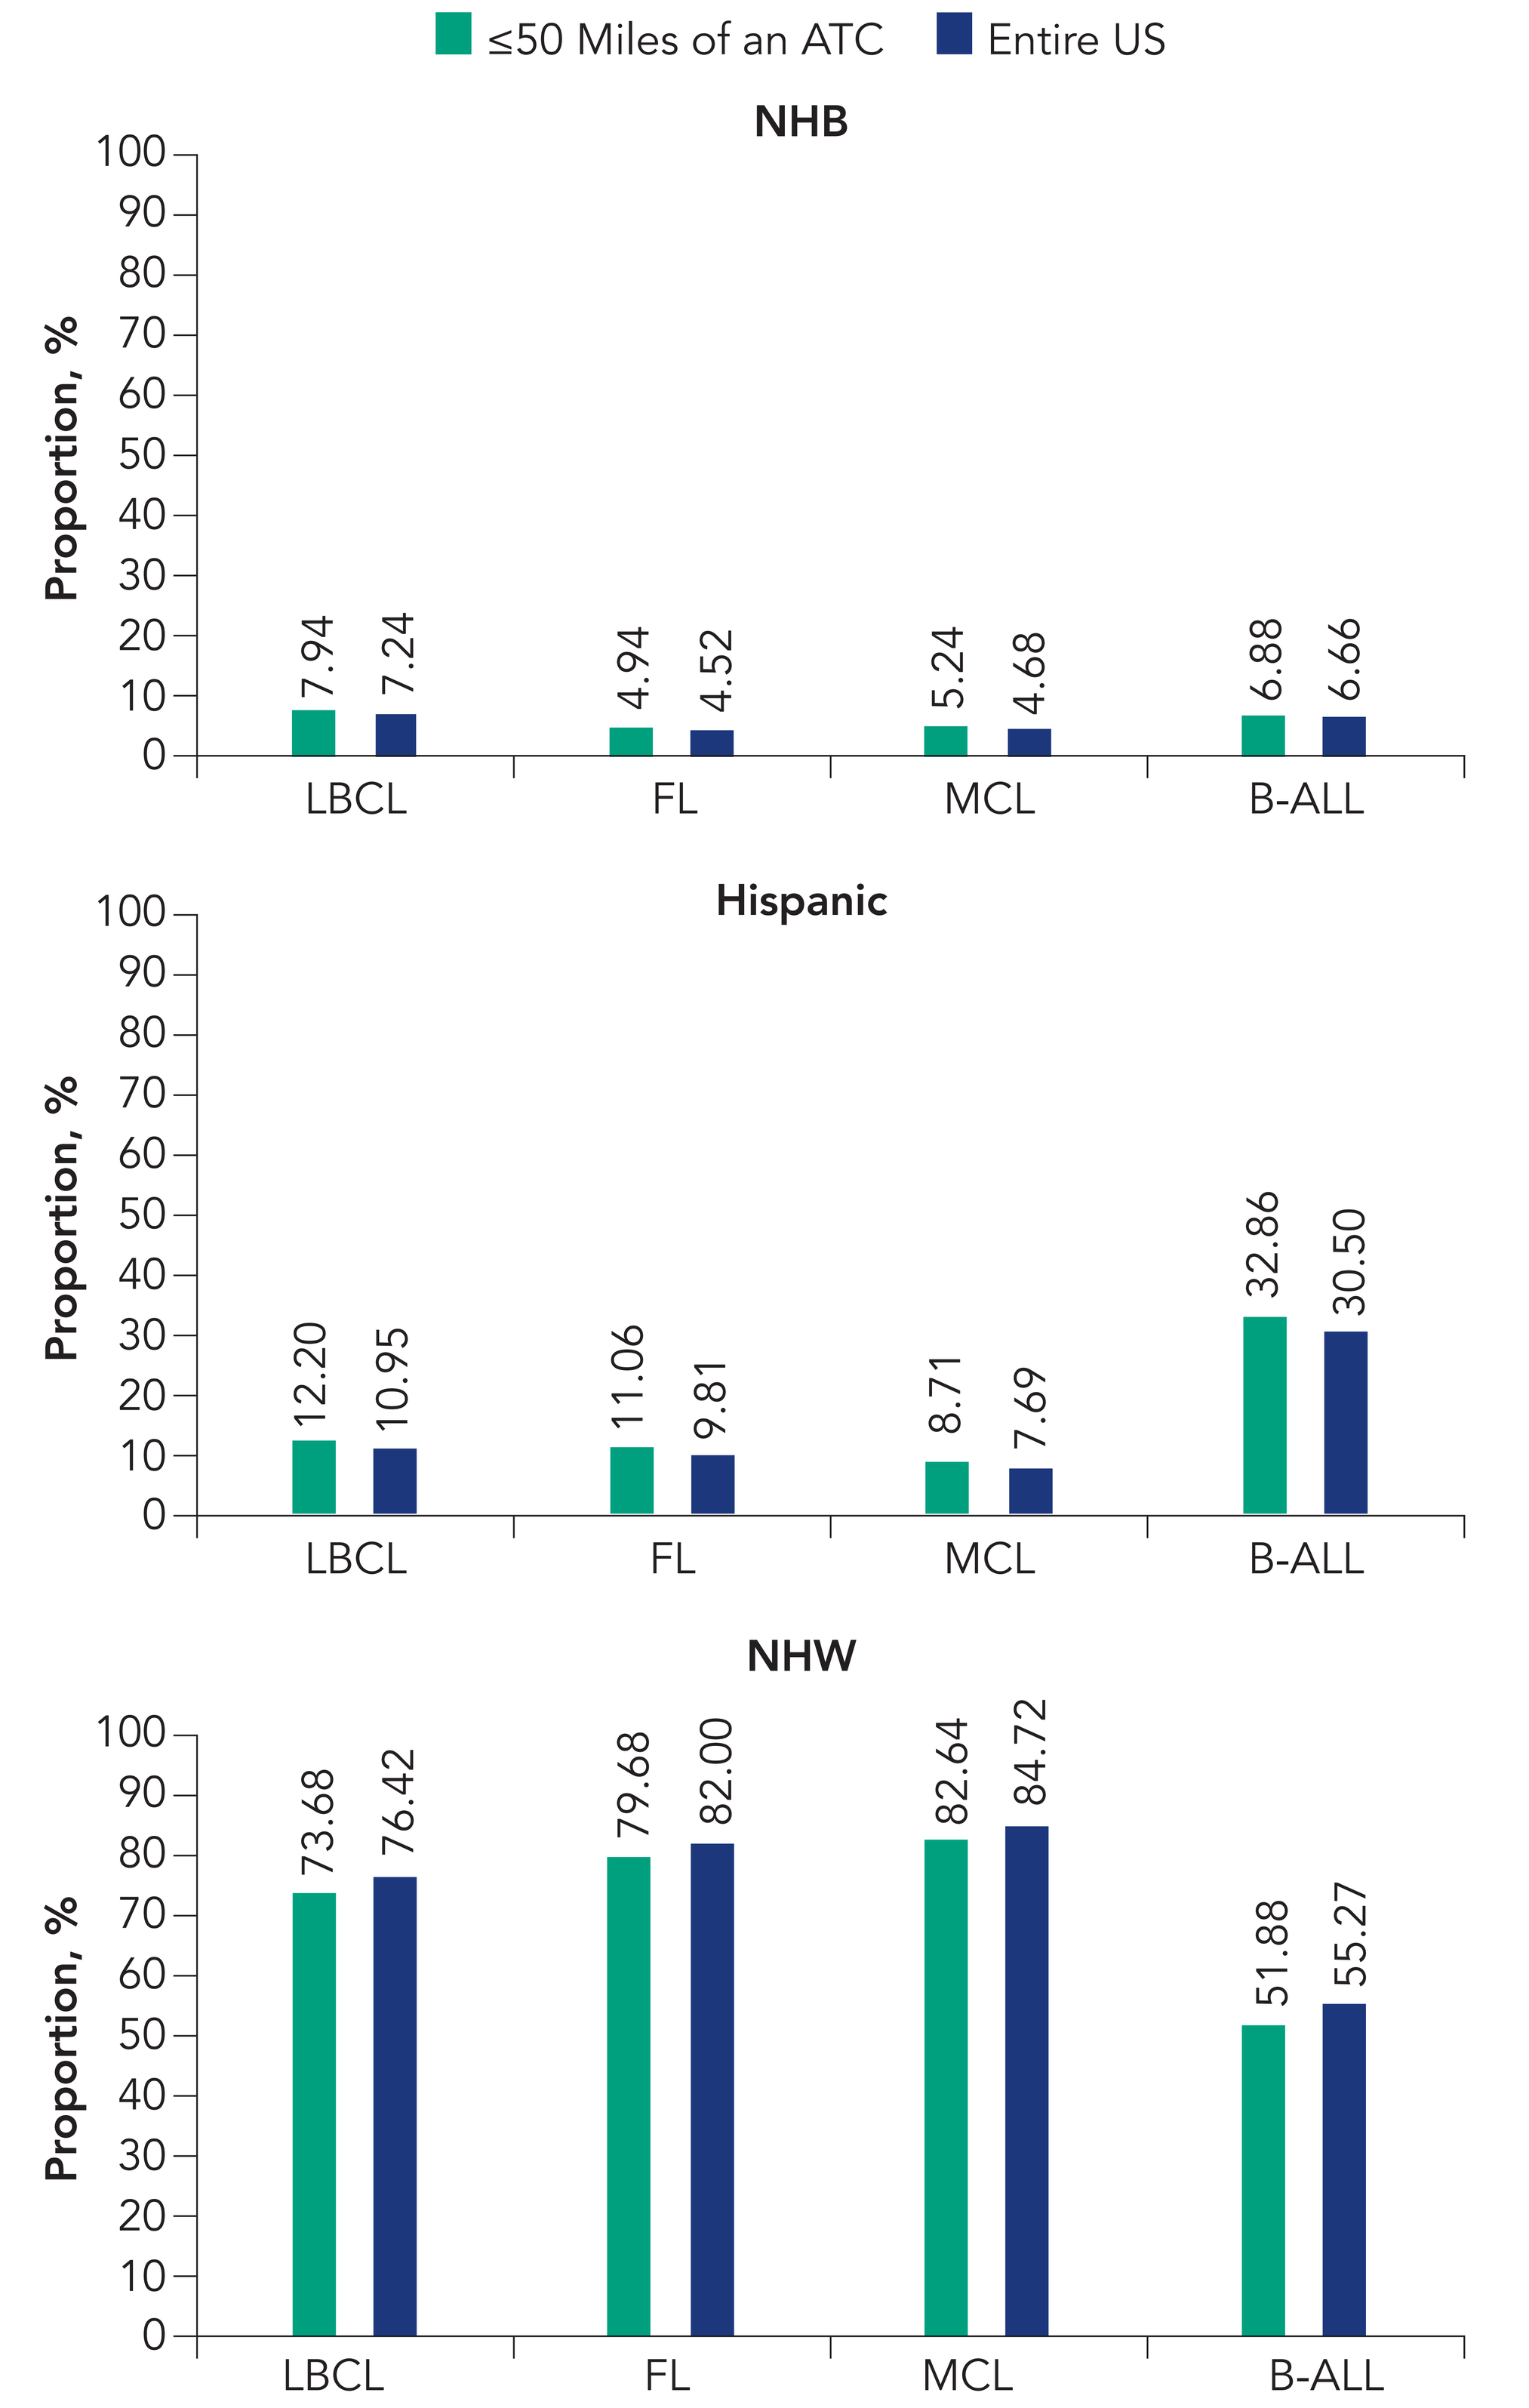
≤50 Miles of an ATC
Entire US
NHB
Proportion, %
7.94 7.24
4.94 4.52
5.24 4.68
6.88 6.66
Hispanic
12.20 10.95
11.06 9.81
8.71 7.69
32.86 30.50
NHW
73.68 76.42
79.68 82.00
82.64 84.72
51.88 55.27
LBCL
FL
MCL
B-ALL

[a]Actual clinical trial enrollments were based on US patients in ZUMA-1 Phase 1 and Phase 2 Cohorts 1, 2 and 6 for LBCL, ZUMA-5 for FL, ZUMA-2 Cohort 1 for MCL, and ZUMA-3 Phase 1 and 2 for B-ALL.
ATC, authorized treatment center; B-ALL, B-cell precursor acute lymphoblastic leukemia; FL, follicular lymphoma; LBCL, large B-cell lymphoma; MCL, mantle cell lymphoma; NHB, non-Hispanic Black; NHW, non-Hispanic White.

CONCLUSIONS

- This is the first simulation study to estimate the proportions of minority patients by considering disease prevalence and proximity to CAR T-cell ATCs
- The possibility of patient underrepresentation is raised as the actual enrolled proportions of minority patients with certain B-cell malignancies were numerically lower than simulation-based estimates, even though most findings were not statistically significant
- The estimated proportions of minority patients were comparable to or even higher than that of NHW patients in regions close to ATCs, indicating that proximity to ATCs may not be the main effect modifier for trial participation and accessibility of care for minority patients
- Further investigation into other factors influencing diversity in clinical trials and accessibility of state-of-the-art cancer care in real-world settings is warranted

DISCLOSURES

Full author disclosures are available through the Quick Response (QR) code.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from the author of this poster.

Presented at the 2022 American Society of Hematology Annual Meeting