Abstract #6531

Long-Term Survival Outcomes of Patients With Relapsed or Refractory B-Cell Acute Lymphoblastic Leukemia Treated With Brexucabtagene Autoleucel in ZUMA-3

Olalekan O. Oluwole, MD, MBBS, MPH[1]; Armin Ghobadi, MD[2]; Ryan D. Cassaday, MD[3]; Jae H. Park, MD[4]; Roch Houot, MD, PhD[5]; Aaron C. Logan, MD, PhD[6]; Nicolas Boissel, MD, PhD[7]; Thibaut Leguay, MD[8]; Michael R. Bishop, MD[9]; Max S. Topp, MD[10]; Kristen M. O'Dwyer, MD[11]; Maria R. Baer, MD[12]; Gary J. Schiller, MD[13]; Mehrdad Abedi, MD[14]; Monique C. Minnema, MD, PhD[15]; Patrick Stiff, MD[16]; Lang Zhou, PhD[17]; Rubina Siddiqi, PhD[17]; Rita Damico Khalid, DO[17]; and Bijal D. Shah, MD[18]

[1]Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [2]Washington University School of Medicine, St Louis, MO, USA; [3]University of Washington School of Medicine and Fred Hutchinson Cancer Center, Seattle, WA, USA; [4]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [5]CHU Rennes, Univ Rennes, Inserm & EFS, Rennes, France; [6]UCSF Medical Center, San Francisco, CA, USA; [7]Hôpital Saint-Louis, Paris, France; [8]Service d'hématologie clinique et thérapie cellulaire Hopital du Haut-Leveque CHU de Bordeaux, Bordeaux, France; [9]The University of Chicago Medicine, Chicago, IL, USA; [10]Medizinische Klinik und Poliklinik II, Universitätsklinikum Würzburg, Würzburg, Germany; [11]University of Rochester Medical Center, Rochester, NY, USA; [12]University of Maryland Marlene and Stewart Greenebaum Cancer Center, Baltimore, MD, USA; [13]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [14]University of California Davis Comprehensive Cancer Center, Sacramento, CA, USA; [15]UMC Utrecht Cancer Center, MS Hematologie, University Medical Center Utrecht, Utrecht, The Netherlands, on behalf of HOVON/LLPC; [16]Loyola University Chicago Stritch School of Medicine, Maywood, IL, USA; [17]Kite, a Gilead Company, Santa Monica, CA, USA; and [18]Moffitt Cancer Center, Tampa, FL, USA.

BACKGROUND

- Brexucabtagene autoleucel (brexu-cel) is an autologous anti-CD19 chimeric antigen receptor (CAR) T-cell therapy approved for patients aged ≥18 years with relapsed or refractory B-cell acute lymphocytic leukemia (R/R B-ALL) in the United States (US) and for patients aged ≥26 years with R/R B-ALL in the European Union (EU) based on results from the pivotal Phase 2, open-label, multicenter ZUMA-3 study[1,2]
 - In ZUMA-3, the median time from leukapheresis to brexu-cel manufacturing release was 13 days in the US and 14.5 days in the EU[3]
- With 41.6 months of median follow-up, the median overall survival (OS) among pooled Phase 1 and 2 ZUMA-3 patients treated at the pivotal dose of 1×10⁶ CAR T cells/kg (N=78) was 25.6 months (N=78; 95% CI, 16.2-47.0)[4]
- Additionally, survival benefit among ZUMA-3 patients was observed regardless of age, prior treatment, or subsequent allogeneic stem cell transplantation (alloSCT) status[4]
 - Patients with more prior therapies and certain prior therapies, such as blinatumomab, appeared to experience less benefit relative to the overall population, though unmatched baseline characteristics and small patient numbers may have confounded these results

OBJECTIVE

- To report 4-year survival, safety, and causes of mortality in ZUMA-3 patients including in key subgroups

METHODS

Figure 1. ZUMA-3 Study Design[3]

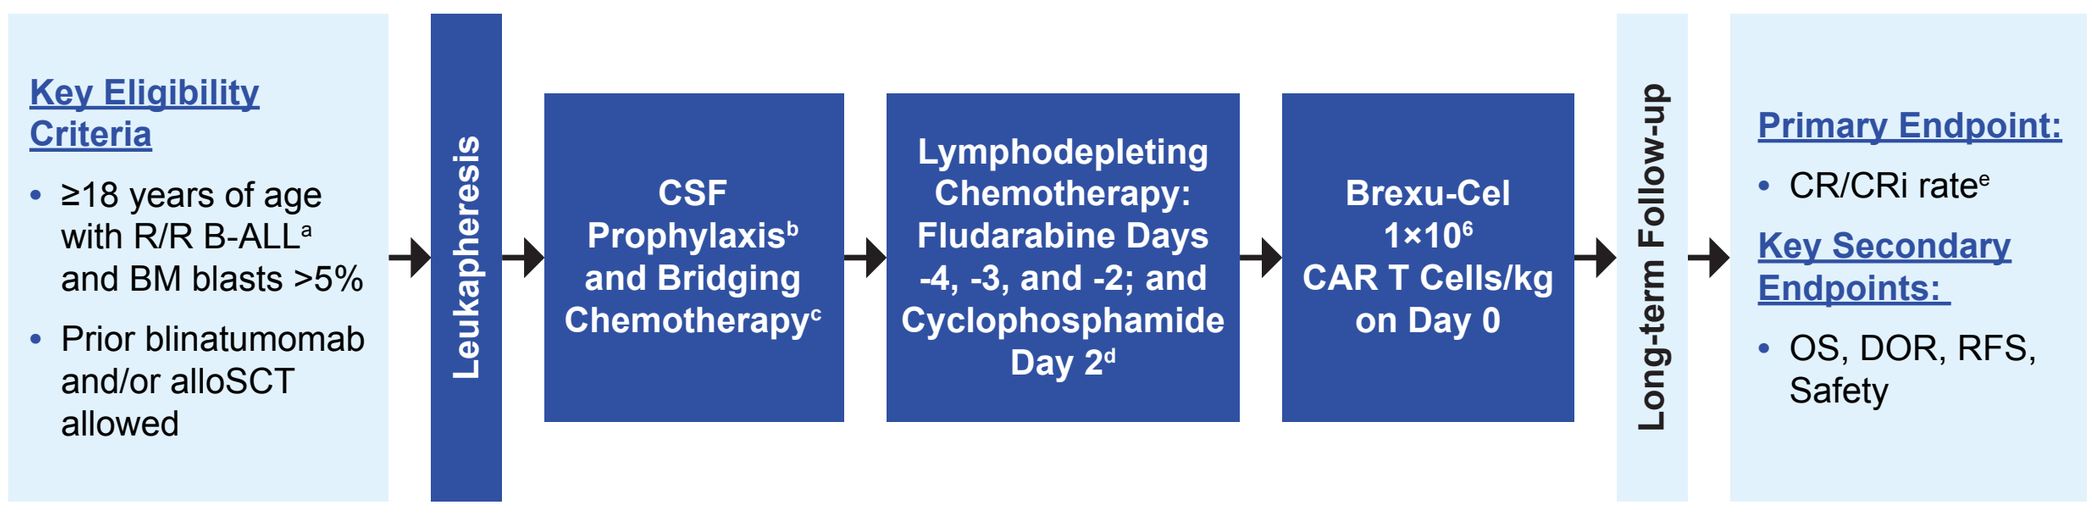
Key Eligibility Criteria
• ≥18 years of age with R/R B-ALL[a] and BM blasts >5%
• Prior blinatumomab and/or alloSCT allowed
Leukapheresis
CSF Prophylaxis[b] and Bridging Chemotherapy[c]
Lymphodepleting Chemotherapy: Fludarabine Days -4, -3, and -2; and Cyclophosphamide Day 2[d]
Brexu-Cel 1×10⁶ CAR T Cells/kg on Day 0
Long-term Follow-up
Primary Endpoint:
• CR/CRi rate[e]
Key Secondary Endpoints:
• OS, DOR, RFS, Safety

[a] R/R disease was defined as primary refractory, first relapse within 12 months, R/R after ≥2 prior lines of systemic therapy or relapsed after alloSCT. [b] All patients received CSF prophylaxis consisting of an intrathecal regimen according to institutional or national guidelines. [c] Bridging chemotherapy was recommended for all patients particularly those with >25% marrow blasts or >1000 blasts/µL of peripheral blood at screening, per physician's discretion. [d] Fludarabine 25 mg/m² IV on Days -4, -3, -2 and cyclophosphamide 900 mg/m² IV on Day -2. [e] Disease assessment was performed per independent central assessment through the month 24 visit or until disease progression. Disease assessment after the Month 24 visit for patients' whose disease had not progressed was performed per standard of care via investigator assessment.
alloSCT, allogeneic stem cell transplantation; BM, bone marrow; brexu-cel, brexucabtagene autoleucel; CAR, chimeric antigen receptor; CR, complete remission; CRi, complete remission with incomplete hematologic recovery; CSF, cerebrospinal fluid; DOR, duration of remission; IV, intravenously; OS, overall survival; RFS, relapse-free survival; R/R B-ALL, relapsed or refractory B-cell acute lymphocytic leukemia.

- A detailed study design for ZUMA-3 was previously reported (**Figure 1**)[3]
- Efficacy and safety outcomes reported herein for ZUMA-3 include all Phase 1 and 2 patients enrolled at the pivotal dose of brexu-cel (N=99), pooled Phase 1 and 2 patients treated at the pivotal dose of brexu-cel (N=78), and Phase 2 treated patients (N=55)
- ZUMA-3 subgroup analyses are exploratory in nature with descriptive statistics reported herein
- Independent central assessment of response was not performed after 24-month assessments; as such, investigator-assessed data are reported for the analysis of patients in ongoing remission
- Time-to-event outcomes, including OS, non-progressive disease (PD)-related mortality, and PD-related mortality, were analyzed using the Kaplan-Meier method
- OS was calculated from time of infusion to death by any cause
- Subsequent alloSCT was allowed at investigator's discretion

RESULTS

- The median follow-up time for Phase 1 and 2 treated patients (N=78), as of the data cutoff date of July 23, 2023, was 53.6 months (range, 44.7-82.3)
- Baseline characteristics for ZUMA-3 patients and key subgroups were previously reported[5]
 - Baseline characteristics were largely similar between key patient subgroups, except patients with prior blinatumomab had a greater median number of prior therapies and higher baseline median bone marrow blast percentages than patients without prior blinatumomab, and patients with ≥2 prior therapies had a greater proportion of patients aged ≥65 years and more patients with prior blinatumomab treatment than patients with 1 prior therapy[5]

Figure 2. Response Status by Investigator Assessment in ZUMA-3 (N=78)

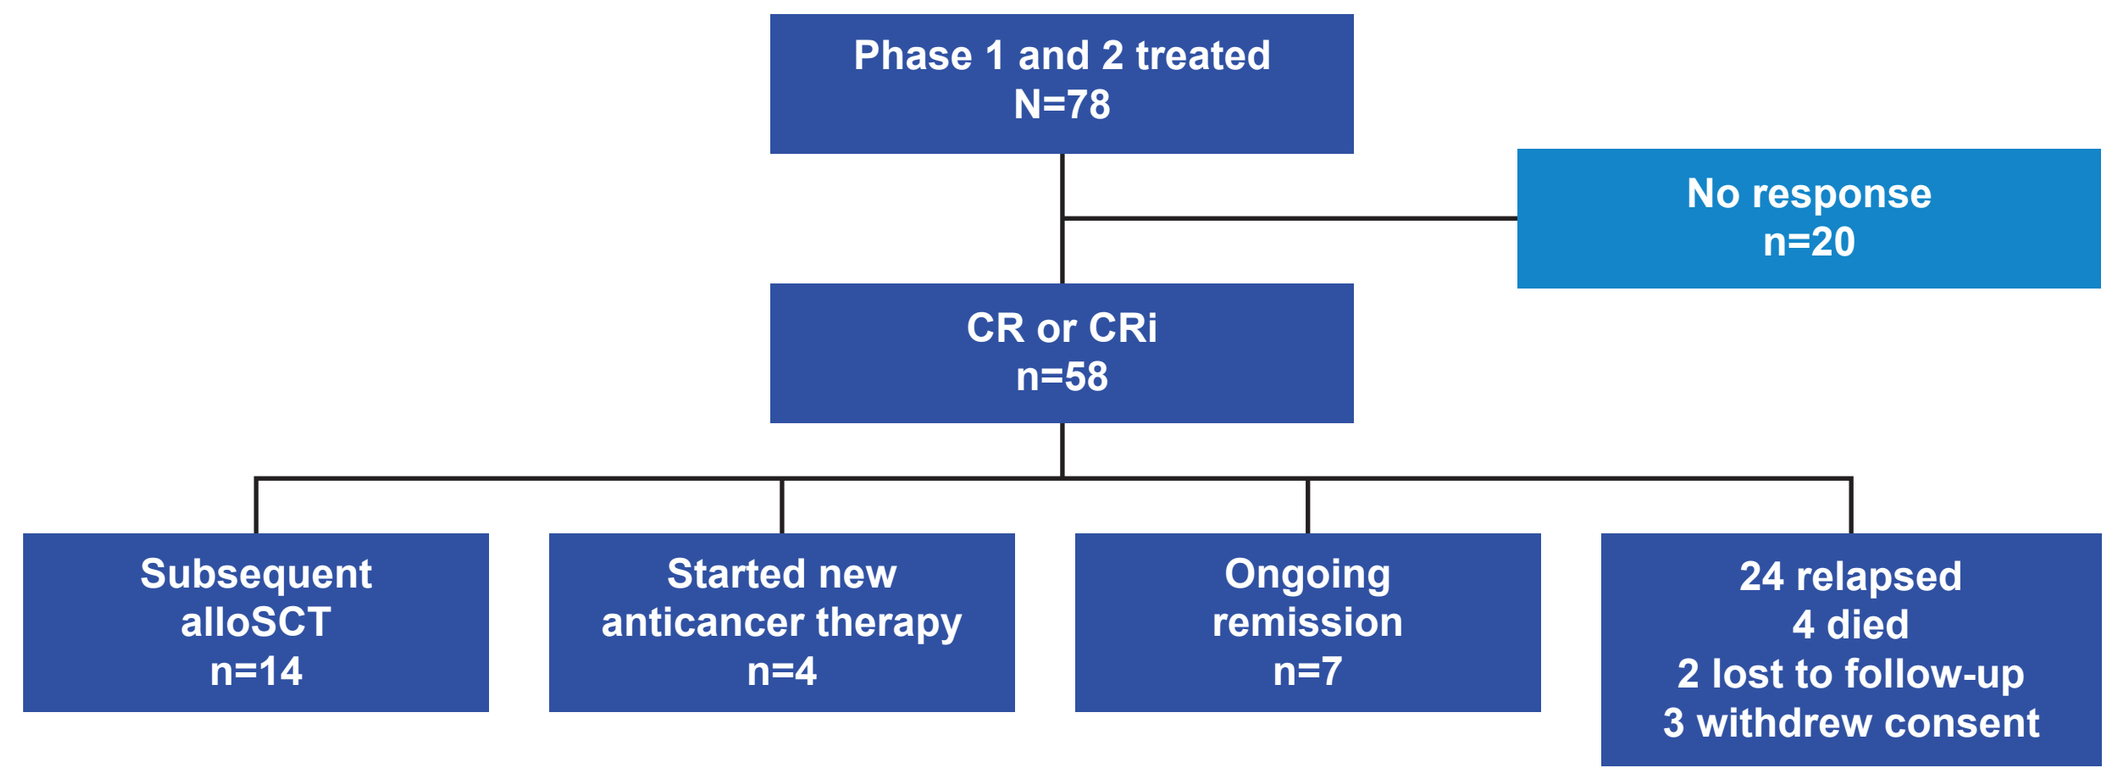
Phase 1 and 2 treated
N=78
No response
n=20
CR or CRi
n=58
Subsequent alloSCT
n=14
Started new anticancer therapy
n=4
Ongoing remission
n=7
24 relapsed
4 died
2 lost to follow-up
3 withdrew consent

alloSCT, allogeneic stem cell transplantation; CR, complete remission; CRi, complete remission with incomplete hematologic recovery.

- As of data cutoff, 7 patients were in ongoing remission per investigator assessment, 24 relapsed, 14 proceeded to subsequent alloSCT, 4 died, 4 started new anticancer therapy, 3 withdrew consent, and 2 were lost to follow-up (**Figure 2**)

RESULTS

Figure 3. Overall Survival Update in ZUMA-3

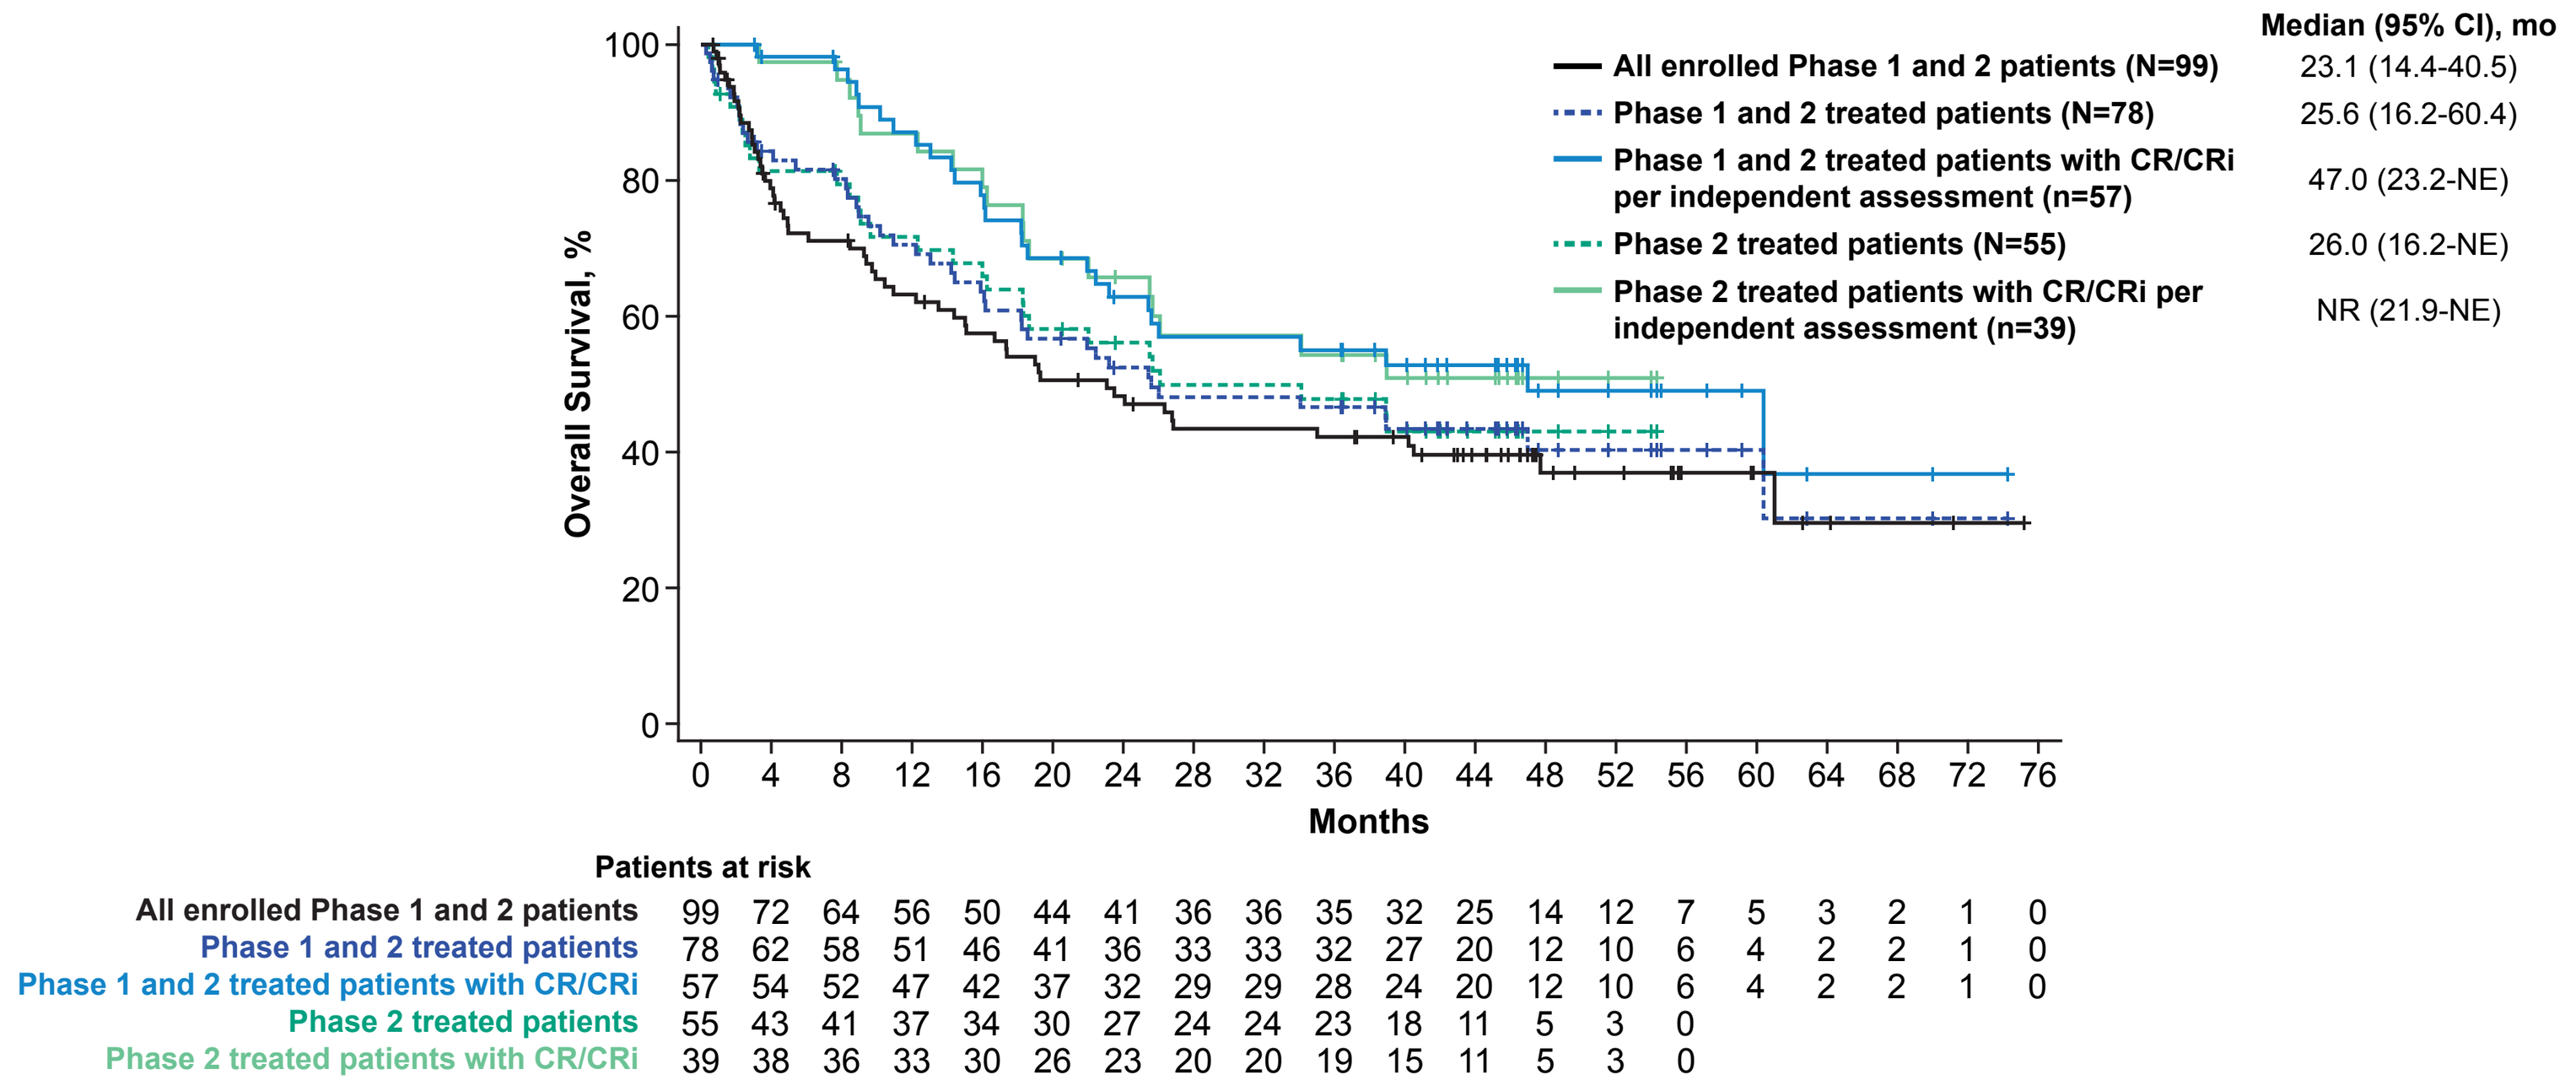
Overall Survival, %
Months
Median (95% CI), mo
All enrolled Phase 1 and 2 patients (N=99) 23.1 (14.4-40.5)
Phase 1 and 2 treated patients (N=78) 25.6 (16.2-60.4)
Phase 1 and 2 treated patients with CR/CRi per independent assessment (n=57) 47.0 (23.2-NE)
Phase 2 treated patients (N=55) 26.0 (16.2-NE)
Phase 2 treated patients with CR/CRi per independent assessment (n=39) NR (21.9-NE)
Patients at risk
All enrolled Phase 1 and 2 patients 99 72 64 56 50 44 41 36 36 35 32 25 14 12 7 5 3 2 1 0
Phase 1 and 2 treated patients 78 62 58 51 46 41 36 33 33 32 27 20 12 10 6 4 2 2 1 0
Phase 1 and 2 treated patients with CR/CRi 57 54 52 47 42 37 32 29 29 28 24 20 12 10 6 4 2 2 1 0
Phase 2 treated patients 55 43 41 37 34 30 27 24 24 23 18 11 5 3 0
Phase 2 treated patients with CR/CRi 39 38 36 33 30 26 23 20 20 19 15 11 5 3 0

alloSCT allogeneic stem cell transplantation; brexu-cel brexucabtagene autoleucel; CR, complete remission; CRi, complete remission with incomplete hematologic recovery; NE, not estimable; NR, not reached.

- The median follow-up time for OS, calculated using the reverse Kaplan-Meier method, was 43.5 months (95% CI, 41.2-46.3) for Phase 2 treated patients (N=55), 46.3 months (95% CI, 42.4-51.5) for Phase 1 and 2 treated patients (N=78), and 46.6 months (95% CI, 43.8-48.4) for all enrolled Phase 1 and 2 patients (N=99)
- Median OS among Phase 2 treated patients (N=55) was 26.0 months (95% CI, 16.2-not estimable [NE]), and was similar among Phase 1 and 2 treated patients (N=78; 25.6 months [95% CI, 16.2-60.4]), as well as all enrolled Phase 1 and 2 patients (N=99; 23.1 months [95% CI, 14.4-40.5]; **Figure 3**)
- ZUMA-3 Phase 1 and 2 responders per independent assessment (CR/CRi; n=57) had a median OS of nearly 4 years (47.0 months [95% CI, 23.2-NE]) and was not reached in Phase 2 responders per independent assessment (95% CI, 21.9-NE; **Figure 3**)

Figure 4. Overall Survival Update in ZUMA-3 by Key Subgroups (N=78)

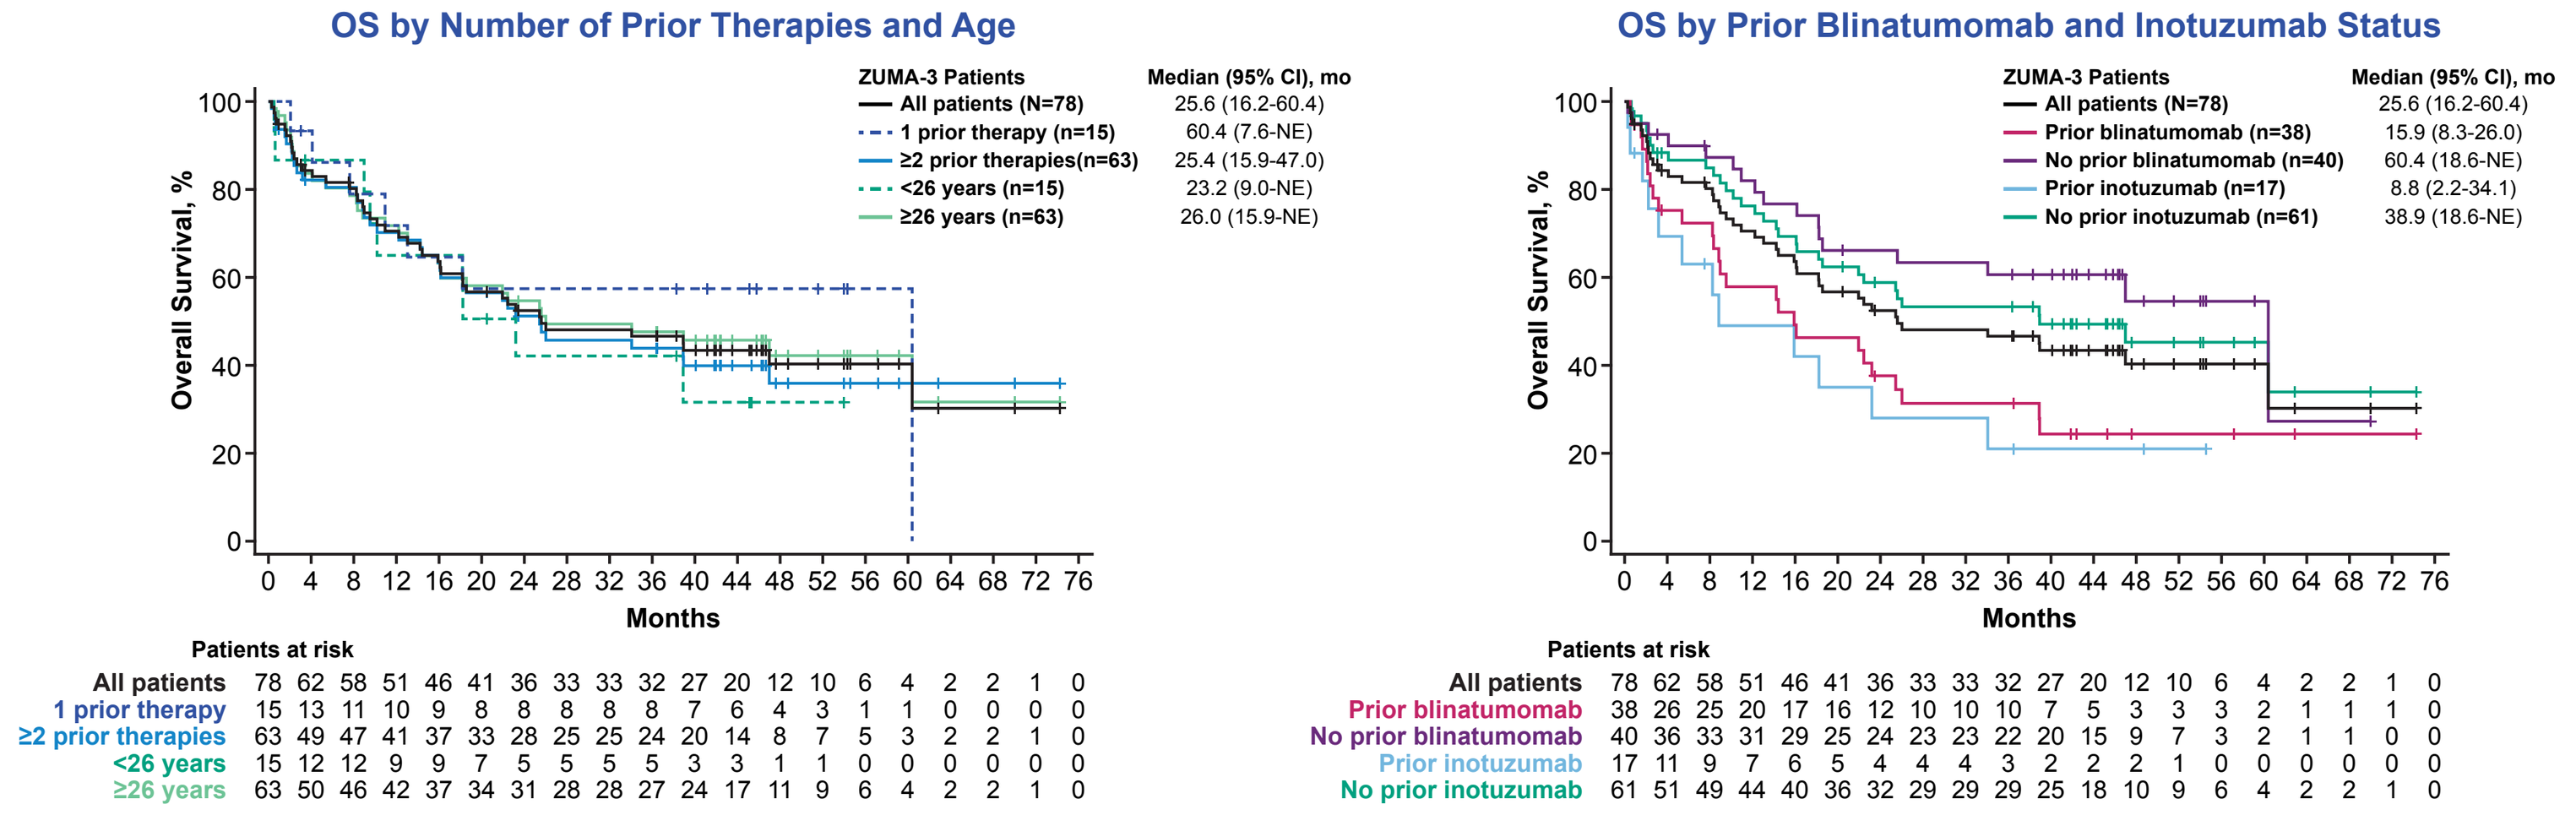
OS by Number of Prior Therapies and Age
ZUMA-3 Patients Median (95% CI), mo
All patients (N=78) 25.6 (16.2-60.4)
1 prior therapy (n=15) 60.4 (7.6-NE)
≥2 prior therapies(n=63) 25.4 (15.9-47.0)
<26 years (n=15) 23.2 (9.0-NE)
≥26 years (n=63) 26.0 (15.9-NE)
Overall Survival, %
Months
Patients at risk
All patients 78 62 58 51 46 41 36 33 33 32 27 20 12 10 6 4 2 2 1 0
1 prior therapy 15 13 11 10 9 8 8 8 8 8 7 6 4 3 1 1 0 0 0 0
≥2 prior therapies 63 49 47 41 37 33 28 25 25 24 20 14 8 7 5 3 2 2 1 0
<26 years 15 12 12 9 9 7 5 5 5 5 3 3 1 1 0 0 0 0 0 0
≥26 years 63 50 46 42 37 34 31 28 28 27 24 17 11 9 6 4 2 2 1 0
OS by Prior Blinatumomab and Inotuzumab Status
ZUMA-3 Patients Median (95% CI), mo
All patients (N=78) 25.6 (16.2-60.4)
Prior blinatumomab (n=38) 15.9 (8.3-26.0)
No prior blinatumomab (n=40) 60.4 (18.6-NE)
Prior inotuzumab (n=17) 8.8 (2.2-34.1)
No prior inotuzumab (n=61) 38.9 (18.6-NE)
Patients at risk
All patients 78 62 58 51 46 41 36 33 33 32 27 20 12 10 6 4 2 2 1 0
Prior blinatumomab 38 26 25 20 17 16 12 10 10 10 7 5 3 3 3 2 1 1 1 0
No prior blinatumomab 40 36 33 31 29 25 24 23 23 22 20 15 9 7 3 2 1 1 0 0
Prior inotuzumab 17 11 9 7 6 5 4 4 4 3 2 2 2 1 0 0 0 0 0 0
No prior inotuzumab 61 51 49 44 40 36 32 29 29 29 25 18 10 9 6 4 2 2 1 0

Brexu-cel, brexucabtagene autoleucel; NE, not estimable; OS, overall survival.

- Median OS for both age groups, <26 and ≥26 years, was similar to the overall population, while patients with only 1 prior therapy or no prior blinatumomab had numerically longer median OS than the overall population (**Figure 4**)
- In patients with ≥2 prior therapies (n=63), those with prior blinatumomab (n=37) had a median OS of 16.1 months (95% CI, 8.8-26.0) and those without prior blinatumomab (n=26) had a median OS of 47.0 months (95% CI, 18.6-NE; data not shown)

Figure 5. Overall Survival in ZUMA-3 Responders Per Independent Assessment by Subsequent AlloSCT Status

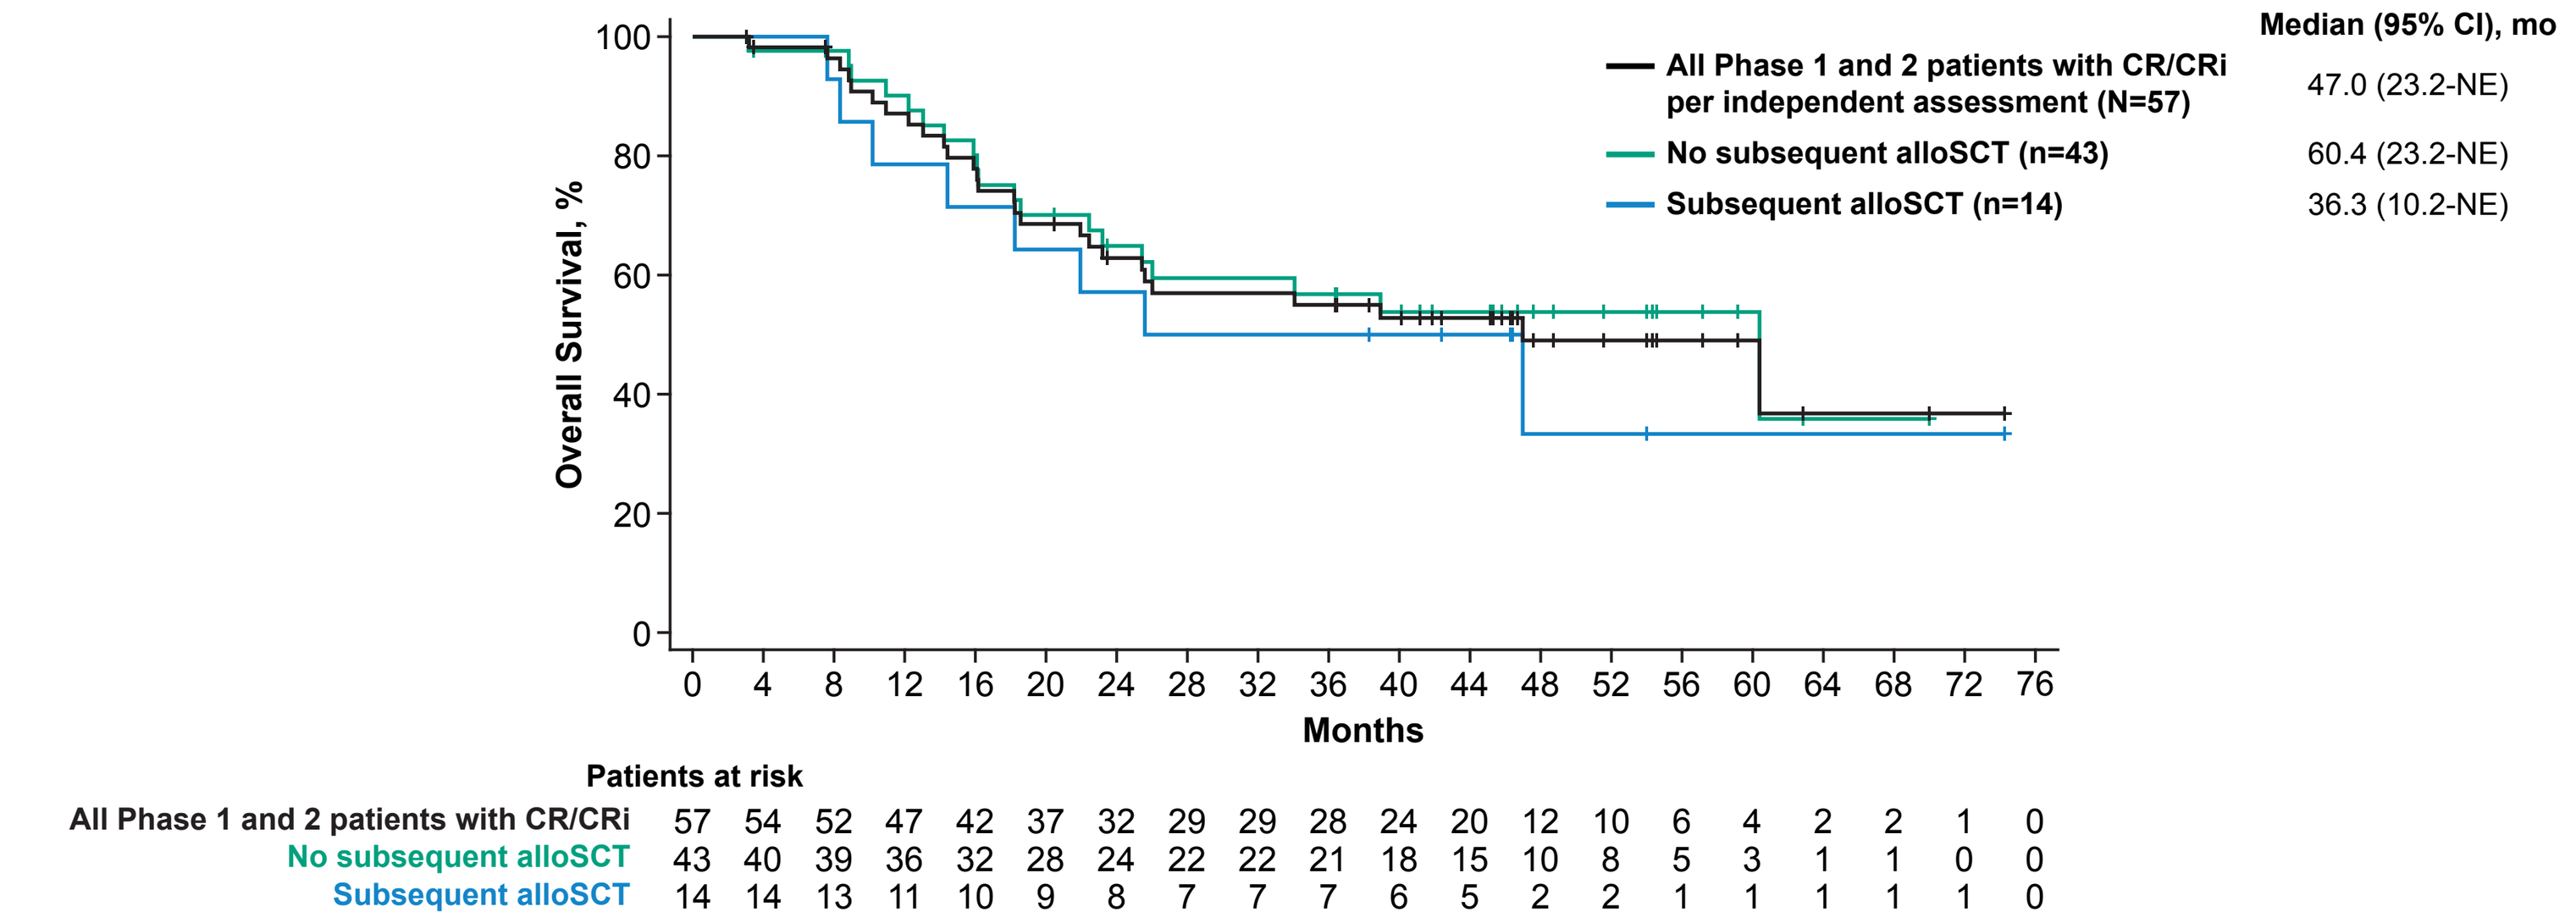
Median (95% CI), mo
All Phase 1 and 2 patients with CR/CRi per independent assessment (N=57) 47.0 (23.2-NE)
No subsequent alloSCT (n=43) 60.4 (23.2-NE)
Subsequent alloSCT (n=14) 36.3 (10.2-NE)
Overall Survival, %
Months
Patients at risk
All Phase 1 and 2 patients with CR/CRi 57 54 52 47 42 37 32 29 29 28 24 20 12 10 6 4 2 2 1 0
No subsequent alloSCT 43 40 39 36 32 28 24 22 22 21 18 15 10 8 5 3 1 1 0 0
Subsequent alloSCT 14 14 13 11 10 9 8 7 7 7 6 5 2 2 1 1 1 1 1 0

alloSCT, allogeneic stem cell transplantation; brexu-cel, brexucabtagene autoleucel; CR, complete remission; CRi, complete remission with incomplete hematologic recovery; NE, not estimable.

- Of the 57 responders per independent assessment, 14 patients proceeded to subsequent alloSCT while in CR/CRi
- Median OS was numerically longer among responders per independent assessment who did not proceed to subsequent alloSCT (n=43; 60.4 months [95% CI, 23.2-NE]) than those who did (n=14; 36.3 months [95% CI, 10.2-NE]; **Figure 5**)

Figure 6. Overall Survival Rates at 48 Months in ZUMA-3 by Key Subgroups (N=78)

	n[a]	OS rate	LCI	UCI
Overall (N=78)	12	0.40	0.28	0.52
Sex				
Male (n=42)	5	0.43	0.27	0.59
Female (n=36)	7	0.39	0.22	0.55
Age				
18-25 (n=15)	1	0.32	0.09	0.58
>25 (n=63)	11	0.42	0.29	0.55
% blasts in BM at baseline				
<5 (n=8)	2	0.33	0.06	0.66
5-25 (n=14)	2	0.51	0.21	0.75
25-50 (n=12)	1	0.49	0.19	0.73
50-75 (n=14)	2	0.46	0.18	0.70
75-100 (n=30)	5	0.34	0.17	0.52
Prior lines of therapy				
1 (n=15)	4	0.57	0.29	0.78
>1 (n=63)	8	0.36	0.23	0.49
Prior alloSCT				
Yes (n=29)	5	0.43	0.25	0.60
No (n=49)	7	0.39	0.23	0.54
Prior blinatumomab				
Yes (n=38)	3	0.24	0.11	0.40
No (n=40)	9	0.55	0.35	0.70
Prior blinatumomab as last line of therapy				
Yes (n=17)	1	0.20	0.05	0.42
No (n=61)	11	0.46	0.32	0.60
Prior inotuzumab				
Yes (n=17)	2	0.21	0.05	0.44
No (n=61)	10	0.45	0.31	0.59
Prior refractory				
Yes (n=24)	3	0.41	0.21	0.60
No (n=54)	9	0.40	0.26	0.55

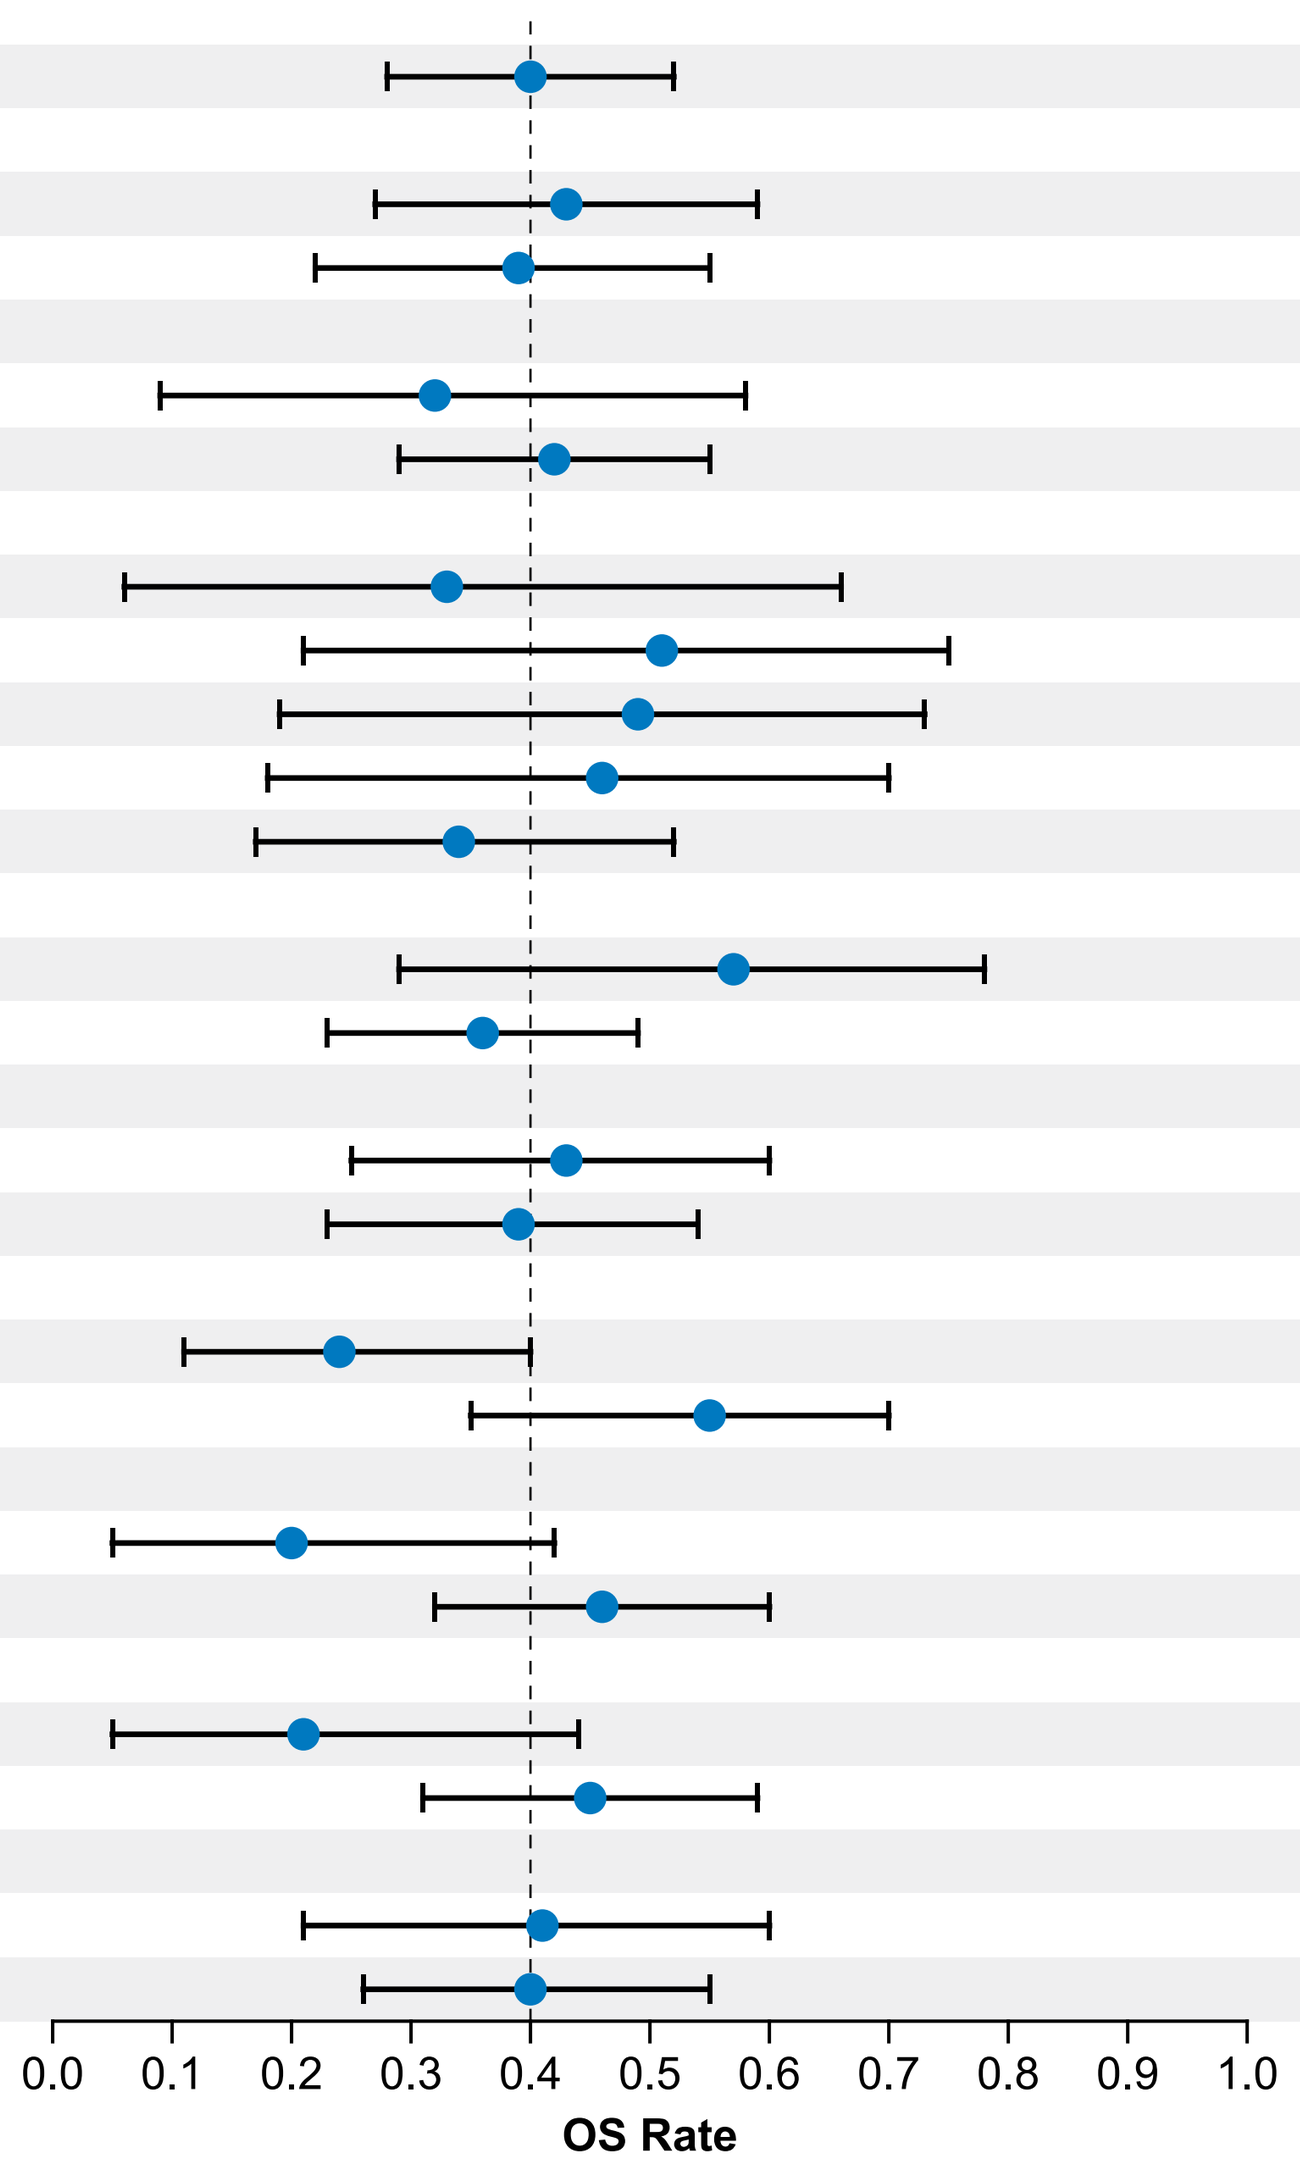
0.0 0.1 0.2 0.3 0.4 0.5 0.6 0.7 0.8 0.9 1.0
OS Rate

[a] Number of patients at risk at Month 48.
alloSCT, allogeneic stem cell transplantation; LCI, lower confidence interval; OS, overall survival; UCI, upper confidence interval.

- The 48-month OS rate among all treated Phase 1 and 2 patients (N=78) was 40% (95% CI, 28-52; **Figure 6**)
 - Rates were similar among most subgroups, though they appeared numerically lower among patients with prior blinatumomab (n=38; 24% [95% CI, 11-40]) or prior inotuzumab (n=17; 21% [95% CI, 5-44]) and higher among patients without prior blinatumomab (n=40; 55% [95% CI, 35-70] or 1 prior therapy (n=15; 57% [95% CI, 29-78])
 - Small numbers of patients in certain subgroups and unbalanced patient characteristics may have confounded these results

Figure 7. Cumulative Incidence of Cause-Specific Mortality Among Phase 1 and 2 Treated Patients (N=78)

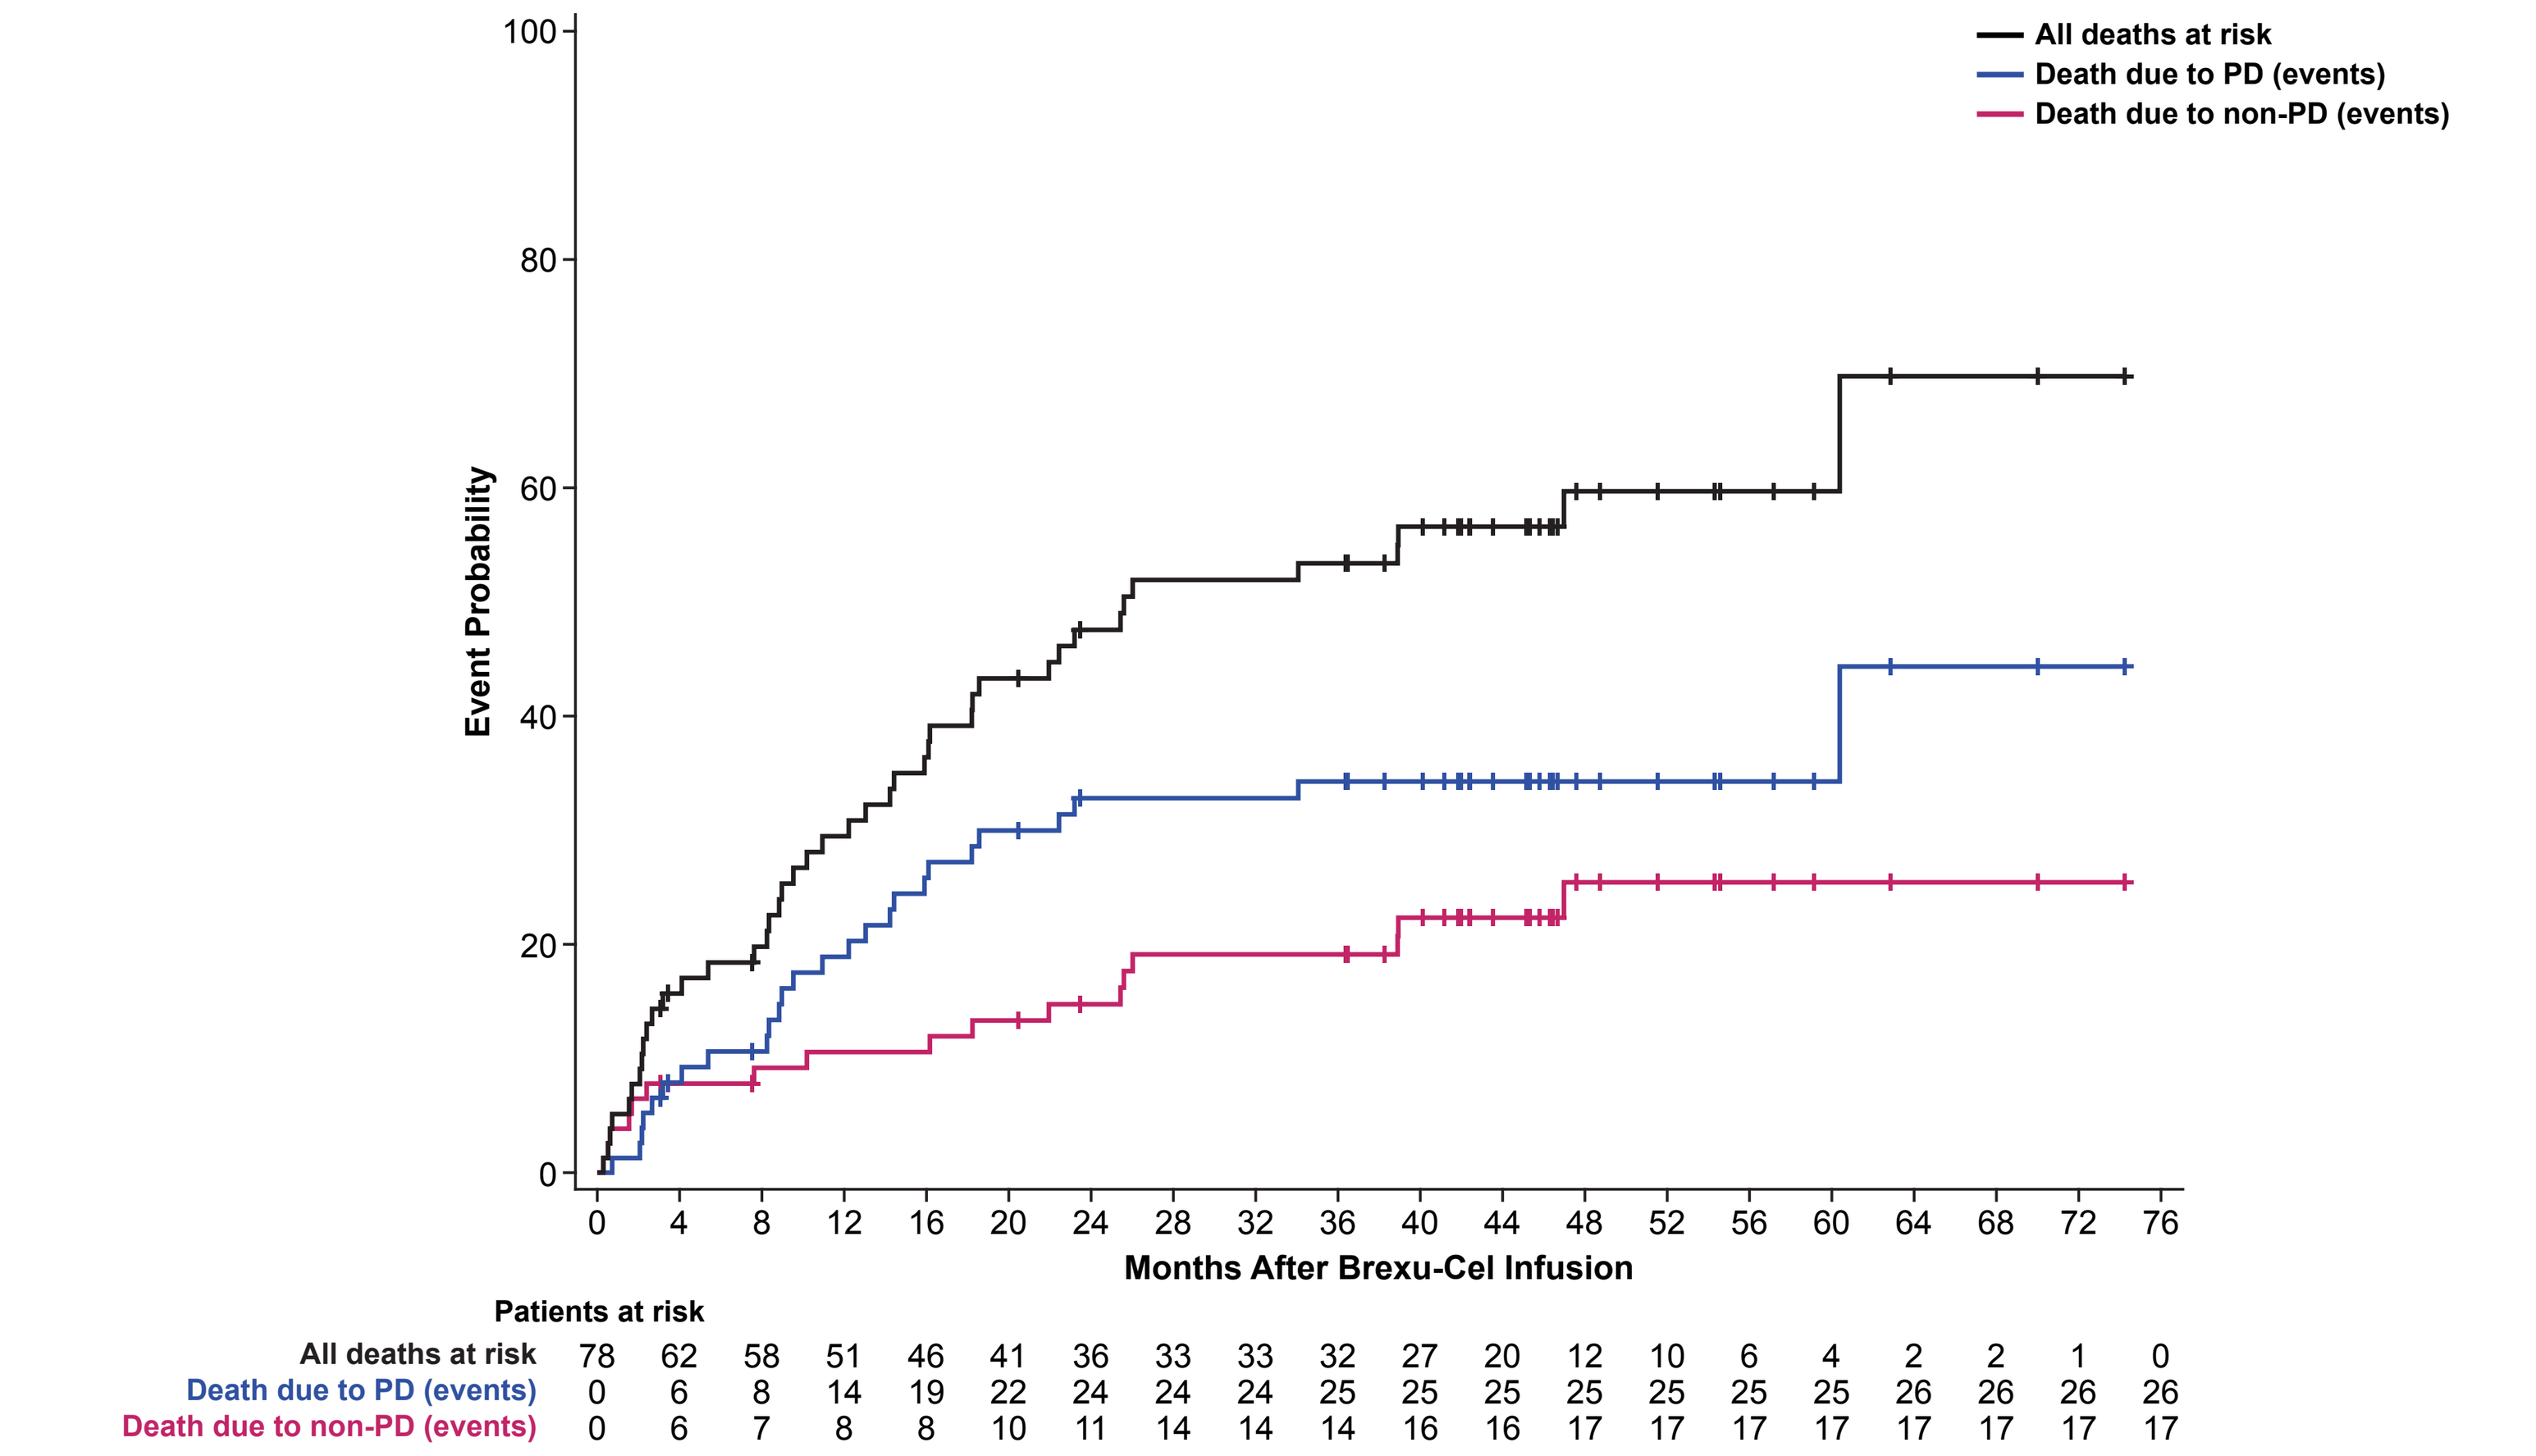
All deaths at risk
Death due to PD (events)
Death due to non-PD (events)
Event Probability
Months After Brexu-Cel Infusion
Patients at risk
All deaths at risk 78 62 58 51 46 41 36 33 33 32 27 20 12 10 6 4 2 2 1 0
Death due to PD (events) 0 6 8 14 19 22 24 24 24 25 25 25 25 25 25 25 26 26 26 26
Death due to non-PD (events) 0 6 7 8 8 10 11 14 14 14 16 16 17 17 17 17 17 17 17 17

Brexu-cel, brexucabtagene autoleucel; PD, progressive disease.

- At data cutoff, 43 of 78 patients had died, 26 due to PD and 17 due to other causes (**Figure 7**)
- The 48-month cumulative incidence rate of death due to PD was 34% (95% CI, 24-45) and the 48-month cumulative incidence rate of death due to other causes was 25% (95% CI, 15-37; **Figure 7**)

Table 1. Causes of Non-PD Mortality

Causes of Non-PD Mortality[a]	Non-PD mortality (n=17)	Time from infusion to death, days	Related to brexu-cel
Grade 5 AE during protocol-specific reporting period, n (%)	9 (12)	–	–
Brain herniation		8	Yes
Pneumonia		15	No
Septic shock		18	Yes
Pneumonia fungal		46	No
Sepsis		50	No
Sepsis		72	No
Herpes simplex viremia[b]		309	No
Respiratory failure		491	No
GVHD		773	No
Other reason for death or occurred outside AE reporting period,[c] n (%)	8 (10)	–	–
Hemorrhagic shock due to GI bleed DIC B-ALL[b,d]		231	No
Multi-organ failure due to infection and GVHD, occurring post-alloSCT[b]		554	No
Cardiopulmonary arrest occurring post-alloSCT[b]		667	No
Hypoxia[b]		778	No
COVID-19		791	No
Intracranial hemorrhage		1183	No
Missing		1184	No
Pulmonary GVHD[b]		1429	No

[a] Reasons for death were provided by investigators. [b] Death occurred after patient received subsequent alloSCT. [c] Primary reason for death was not reported as an AE; however, some deaths occurred outside the AE reporting period. [d] Death was inaccurately described as B-ALL as patient was in CR at time of death.
AE, adverse event; alloSCT, allogeneic stem cell transplantation; B-ALL, B-cell acute lymphocytic leukemia; brexu-cel, brexucabtagene autoleucel; CR, complete remission; DIC, disseminated intravascular coagulation; GI, gastrointestinal; GVHD, graft-versus-host disease; PD, progressive disease.

- Six of 17 deaths not due to PD (35%) occurred in patients who had received subsequent alloSCT (**Table 1**)

Table 2. Treatment-Emergent Infections in ZUMA-3 by Key Subgroups (N=78)

	Age Category		Prior Blinatumomab		No. of Prior Therapies	
	<26 years (n=15)	≥26 years (n=63)	Yes (n=38)	No (n=40)	1 (n=15)	≥2 (n=63)
Any treatment-emergent infection, n (%)	6 (40)	25 (40)	19 (50)	12 (30)	3 (20)	28 (44)
Worst infection experienced was Grade ≥3, n (%)	6 (40)	17 (27)	15 (39)	8 (20)	3 (20)	20 (32)

- No adverse events or deaths occurred since the prior analysis[6]
- Infections of any grade since study start appeared higher in patients with prior blinatumomab while appearing lower in patients with 1 prior therapy and patients without prior blinatumomab (**Table 2**)
 - Grade ≥3 infections appeared higher in patients aged <26 years and in patients with prior blinatumomab

CONCLUSIONS

- After >4 years of follow-up, patients in ZUMA-3 continued to experience OS benefit regardless of age, prior therapy, or subsequent alloSCT status
 - Patients with prior blinatumomab had a numerically lower 48-month OS rate
 - Responders per independent assessment who did not proceed to subsequent alloSCT had a numerically longer median OS than those who did (60.4 and 36.3 months, respectively)
 - Small subgroups and unbalanced patient characteristics limit interpretation of these results
- The non-PD mortality rate was 25% (n=17) after 4 years of median follow-up with only 2 of the 17 non-PD-related deaths deemed related to brexu-cel
- No new safety signals or deaths occurred among Phase 1 and 2 treated patients since the 3-year analysis
- Grade ≥3 infections occurred at a numerically higher rate in younger patients, those with prior blinatumomab, and those with ≥2 prior therapies
- Further studies are needed to fully assess the impact of age, prior therapies, and subsequent alloSCT on outcomes after brexu-cel

REFERENCES

1. TECARTUS® (brexucabtagene autoleucel) [Prescribing information]. Kite Pharma, Inc; 2024.
2. TECARTUS® (brexucabtagene autoleucel) [summary of product characteristics]. Amsterdam, The Netherlands: Kite Pharma EU B.V.; 2024.
3. Shah BD, et al. *Lancet*. 2021;398(10299):491-502.
4. Shah BD, et al. *Journal of Clinical Oncology*. 2023;41(16_suppl):7023.
5. Shah BD, et al. *J Immunother Cancer*. 2023;11:e007118.
6. Shah BD, et al. EU CAR T 2023. Abstract 34.

ACKNOWLEDGMENTS

- The patients, families, friends, and caregivers
- The study investigators, coordinators, and health care staff at each study site
- Medical writing support was provided by Edward Sheetz, PhD, of Nexus Global Group Science, funded by Kite, a Gilead Company
- This study was funded by Kite, a Gilead Company

DISCLOSURES

OOO: honoraria from Gilead Sciences and Pfizer; consulting/advisory role for AbbVie, ADC Therapeutics, Cargo, Caribou Biosciences, Epizyme, Gilead Sciences, Kite, a Gilead Company, Nektar, Novartis, Pfizer, and TGR; speakers' bureau participation for ADC Therapeutics; and research funding from Allogene, Daiichi Sankyo, Kite, and Pfizer.
Full author disclosures are available through the Quick Response (QR) code

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Presented at the 2024 American Society of Clinical Oncology Annual Congress

FULL AUTHOR DISCLOSURES

OOO: honoraria from Gilead Sciences and Pfizer; consulting/advisory role for AbbVie, ADC Therapeutics, Cargo, Caribou Biosciences, Epizyme, Gilead Sciences, Kite, a Gilead Company, Nektar, Novartis, Pfizer, and TGR; speakers' bureau participation for ADC Therapeutics; and research funding from Allogene, Daiichi Sankyo, Kite, and Pfizer. **AG:** honoraria from Kite, a Gilead Company; consulting/advisory role for Amgen, Atara, Bristol Myers Squibb, CRISPR Therapeutics, Kite, and Wugen Inc.; research funding from Amgen, Genentech, and Kite. **RDC:** employment with and stock or other ownership in Seagen (immediate family member); honoraria from Amgen, Jazz, Kite, a Gilead Company, and Pfizer; consultancy/advisory role for Amgen, Autolus, and Kite; research funding from Amgen, Incyte, Kite, Merck, Pfizer, Servier, and Vanda; travel support from Kite; and other relationship with Autolus (independent review committee), and PeproMene Bio (DSMB). **JHP:** consulting/advisory role for AstraZeneca, Kite, a Gilead Company, and Novartis; and research funding from Amgen, Genentech, and Juno. **RH:** honoraria from ADC Therapeutics, Bristol Myers Squibb, Celgene, Gilead Sciences, Janssen, Kite, a Gilead Company, MSD, and Novartis; and consulting/advisory role for Kite and Gilead Sciences. **ACL:** consulting/advisory role for AbbVie, Actinium, Bristol Myers Squibb, Pfizer, and Takeda; research funding from Amgen, Astellas, Autolus, Kadmon, Kite, a Gilead Company, Pharmacyclics, and Talaris; and other relationship with **DSMB:** Servier. **NB:** honoraria and consulting/advisory role for Amgen, Kite, a Gilead Company, and Novartis; and research funding and expert testimony for Amgen. **TL:** honoraria from Amgen, Incyte, Kite, a Gilead Company, and Servier; and consulting/advisory role for Amgen, Incyte, and Servier. **MRB:** honoraria from ADC Therapeutics, Bristol Myers Squibb, Incyte, and Sanofi; consulting/advisory role for Bristol Myers Squibb, Kite, a Gilead Company, Novartis, and Sana Bio; and speakers' bureau participation for ADC Therapeutics, Bristol Myers Squibb, Incyte, and Sanofi. **MST:** consulting/advisory role for AstraZeneca, Bristol Myers Squibb, Genmab, Kite, a Gilead Company, and Roche; research funding from Kite, Regeneron, Roche, and Takeda; and travel support from Janssen and Kite. **KMO:** no relevant financial relationships to disclose. **MRB:** research funding from AbbVie, Ascentage, Kite, a Gilead Company, Kura, and Takeda. **GJS:** honoraria and research funding from and speakers' bureau participation for Kite, a Gilead Company. **MA:** honoraria from and consulting/advisory role for Celgene; speakers' bureau participation for AbbVie, Bristol Myers Squibb, Celgene, and Kite, a Gilead Company. **MCM:** consulting or advisory role for CDR-life, GSK, and Janssen/Cilag (institution); speakers' bureau participation for BMS, Jansen/Cilag, Pfizer, and Siemens; research funding from BeiGene (institution); and travel, accommodations, and expenses from Bristol Myers Squibb. **PS:** honoraria from and consulting or advisory role for MorphoSys and CRISPR Therapeutics; and research funding from Amgen, Gamida Cell, Pfizer, Karyopharm, Gilead Sciences, Incyte, Seagen, and Cellectar. **LZ:** employment with Kite, a Gilead Company; and stock or other ownership in AbbVie RSU. **RS:** employment with Amgen and Kite, a Gilead Company; stock or other ownership in Amgen and Gilead Sciences; and travel support from Kite/Gilead Sciences. **RDK:** employment with, stock or other ownership in, and travel support from Kite, a Gilead Company. **BDS:** consulting/advisory role for Adaptive Biotechnologies, Amgen, Autolus, BeiGene, Bristol Myers Squibb, Century Therapeutics, Deciphera, Jazz, Kite, a Gilead Company, Lilly, Novartis, Precision Biosciences, Pfizer, and Takeda; research funding from Jazz Pharmaceuticals, Kite, and Servier; and other relationship with PeproMene Bio.

Presented at the 2024 American Society of Clinical Oncology Annual Congress